Abstract: 2716

Development of TNF superfamily agonists: 4-1BB ligand fusion shows anti-tumor activity *in vivo*

Eric Tam, Galina Craig, Cormac Cosgrove, Alexander Koshkaryev, Ross Fulton, Daryl Drummond, and Andreas Raue

Merrimack Pharmaceuticals, Inc., Cambridge MA, USA

Summary

Members of the tumor necrosis factor (TNF) superfamily of costimulatory receptors have emerged as promising targets for modulation of tumor infiltrating lymphocytes. Among these, 4-1BB (CD137) has a prominent role in promoting survival and expansion of cytotoxic CD8 T cells as well as activation of CD4 T cells, NK cells, and antigen-presenting cells. The clinical relevance of 4-1BB agonism is currently being evaluated for two antibodies. However, there is evidence that for the trimeric TNF receptor family, the homotrimeric ligands are superior activators compared to bivalent agonistic antibodies. Here we describe a format termed STIMULI (stabilized immunoligand), which consists of three protomers of a TNF family ligand linked together as a single polypeptide chain and fused to the C-terminus of an antibody Fc region. We converted 4-1BBL, OX40L, and CD40L into STIMULI format as a fusion to the Fc of human IgG1. Compared to clinically relevant agonistic antibodies the human STIMULI molecules showed greater signaling activity, increased immune cell activation, and cytokine release. We also generated a murine surrogate of the 4-1BBL fusion (STIM-41BBL). While murine STIM-41BBL showed greater activation and cytokine release than an agonistic antibody in isolated T cells, both molecules were comparable in an OT-1 T cell proliferation and a NK cell activation assay. We went on to test the murine STIM-41BBL in the MC38 tumor model where it was observed to be synergistic with anti-PD1. In summary, while the human STIMULI molecules look promising, further *in vivo* work is underway using the murine STIM-41BBL to understand the advantages and limitations of the STIMULI format compared to agonist antibodies.

STIMULI format – single chain TNF superfamily ligand fusions to Fc or IgG

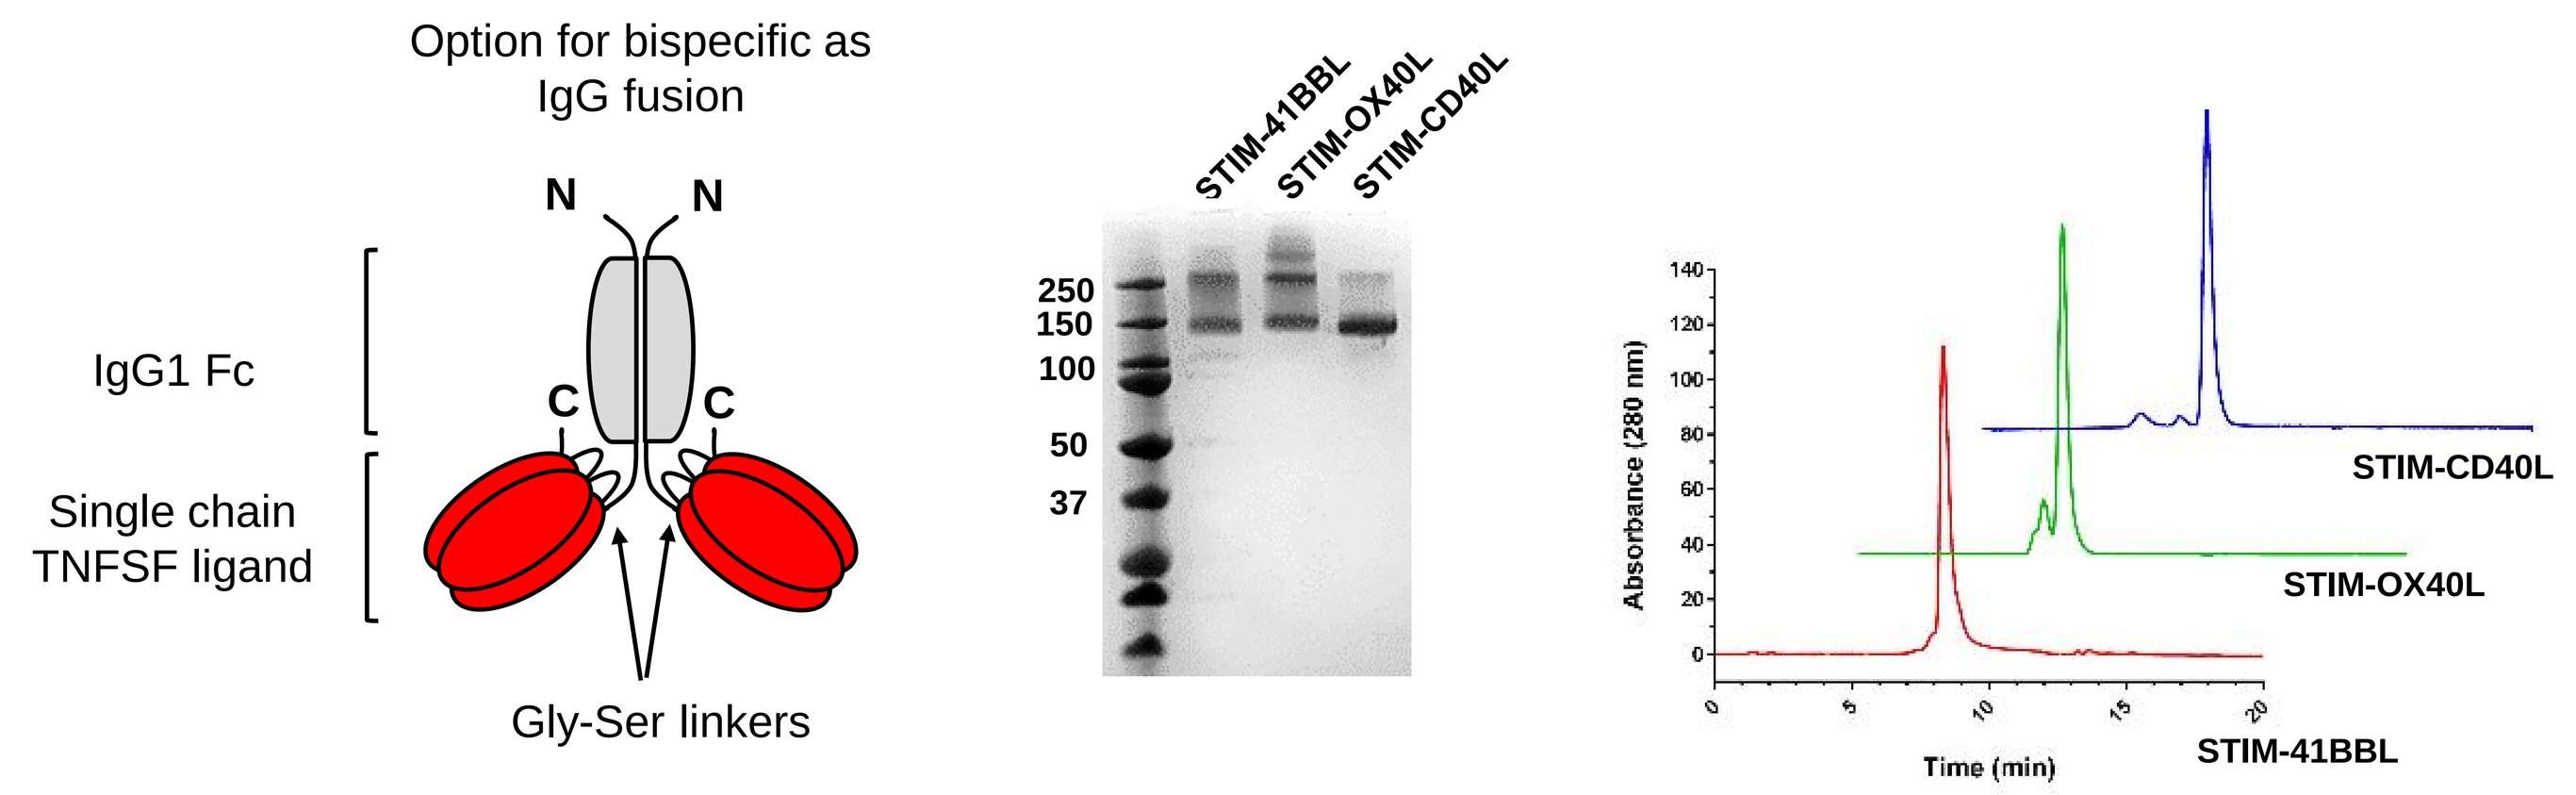
Option for bispecific as IgG fusion
IgG1 Fc
Single chain TNFSF ligand
Gly-Ser linkers
STIM-41BBL
STIM-OX40L
STIM-CD40L
STIM-CD40L
STIM-OX40L
STIM-41BBL
Time (min)
Absorbance (280 nm)

Single chain versions of TNF superfamily ligand homotrimers were generated by connecting the three protomers using glycine serine linkers. Fusion to the C-terminus of human IgG1 Fc allows the option of developing a bispecific with the N-terminus using an IgG. Proteins were expressed in CHO cells, purified using Protein A, and were observed to be the expected M.W. by SDS-PAGE and SEC.

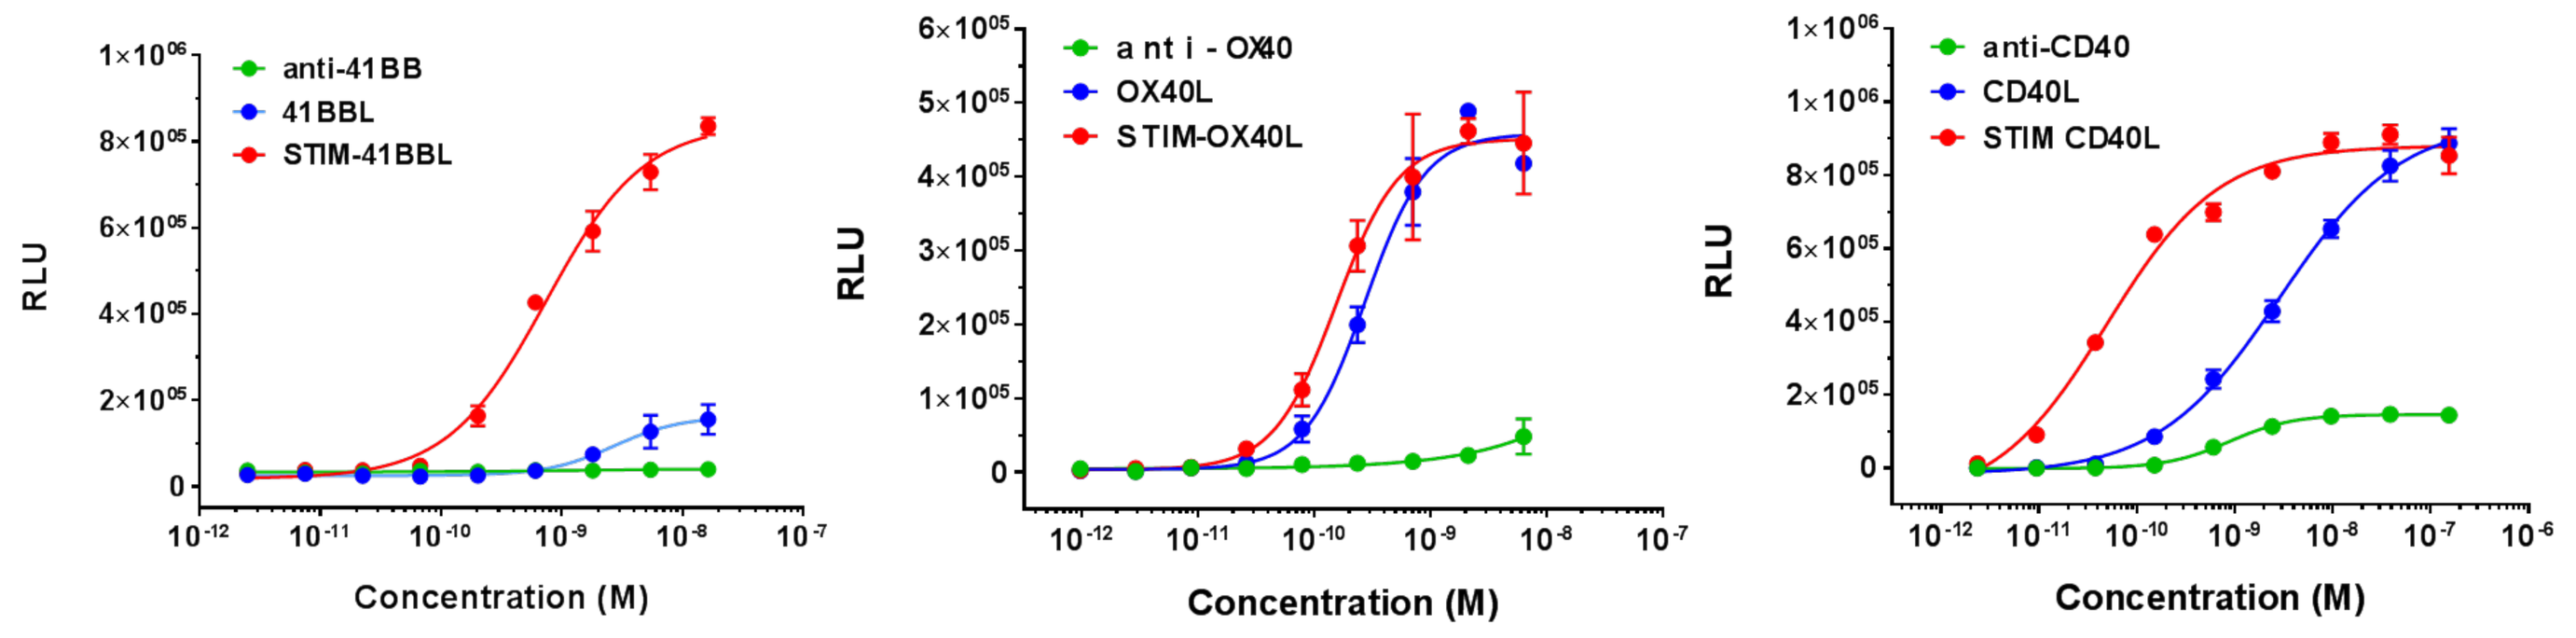
anti-41BB
41BBL
STIM-41BBL
anti-OX40
OX40L
STIM-OX40L
anti-CD40
CD40L
STIM CD40L
RLU
Concentration (M)

STIMULI versions of 4-1BBL, OX40L, and CD40L were compared with their corresponding recombinant ligands and agonistic antibodies (anti-41BBL: utomilumab; anti-OX40L: MEDI6469; anti-CD40: CP-870,893) using NF-κB-luc2P Jurkat reporter cell line engineered to express either 4-1BB, OX40, or CD40 (Promega).

Human STIMULI molecules promote immune cell activation and proliferation

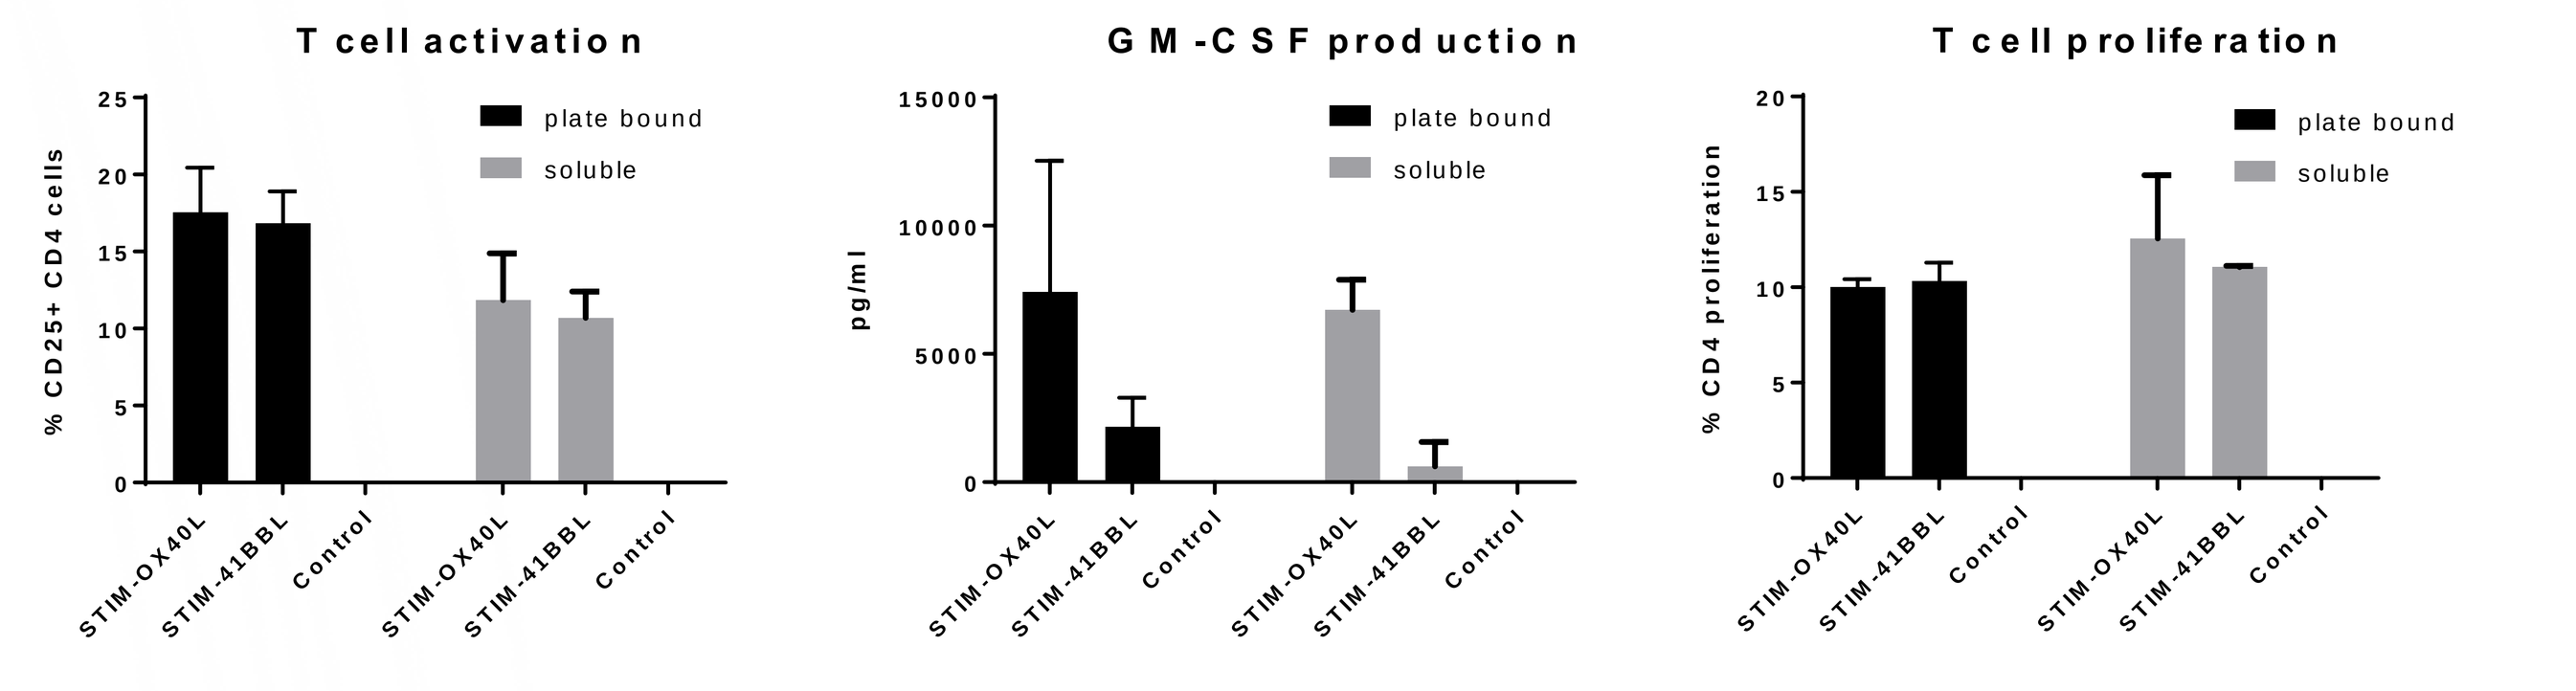
T cell activation
GM-CSF production
T cell proliferation
plate bound
soluble
% CD25+ CD4 cells
pg/ml
% CD4 proliferation

CD4+ T cells purified from whole blood of healthy donors were stained with cell trace violet and incubated with plate-bound anti-CD3 (2 μg/mL) and either plate-bound or soluble STIM-41BBL (12.5 nM), STIM-OX40L (12.5 nM) or isotype control. After 72 hrs of incubation, the percentage of CD25+ CD4 cells and cell divisions were observed using flow cytometry and GM-CSF levels were measured by Luminex.

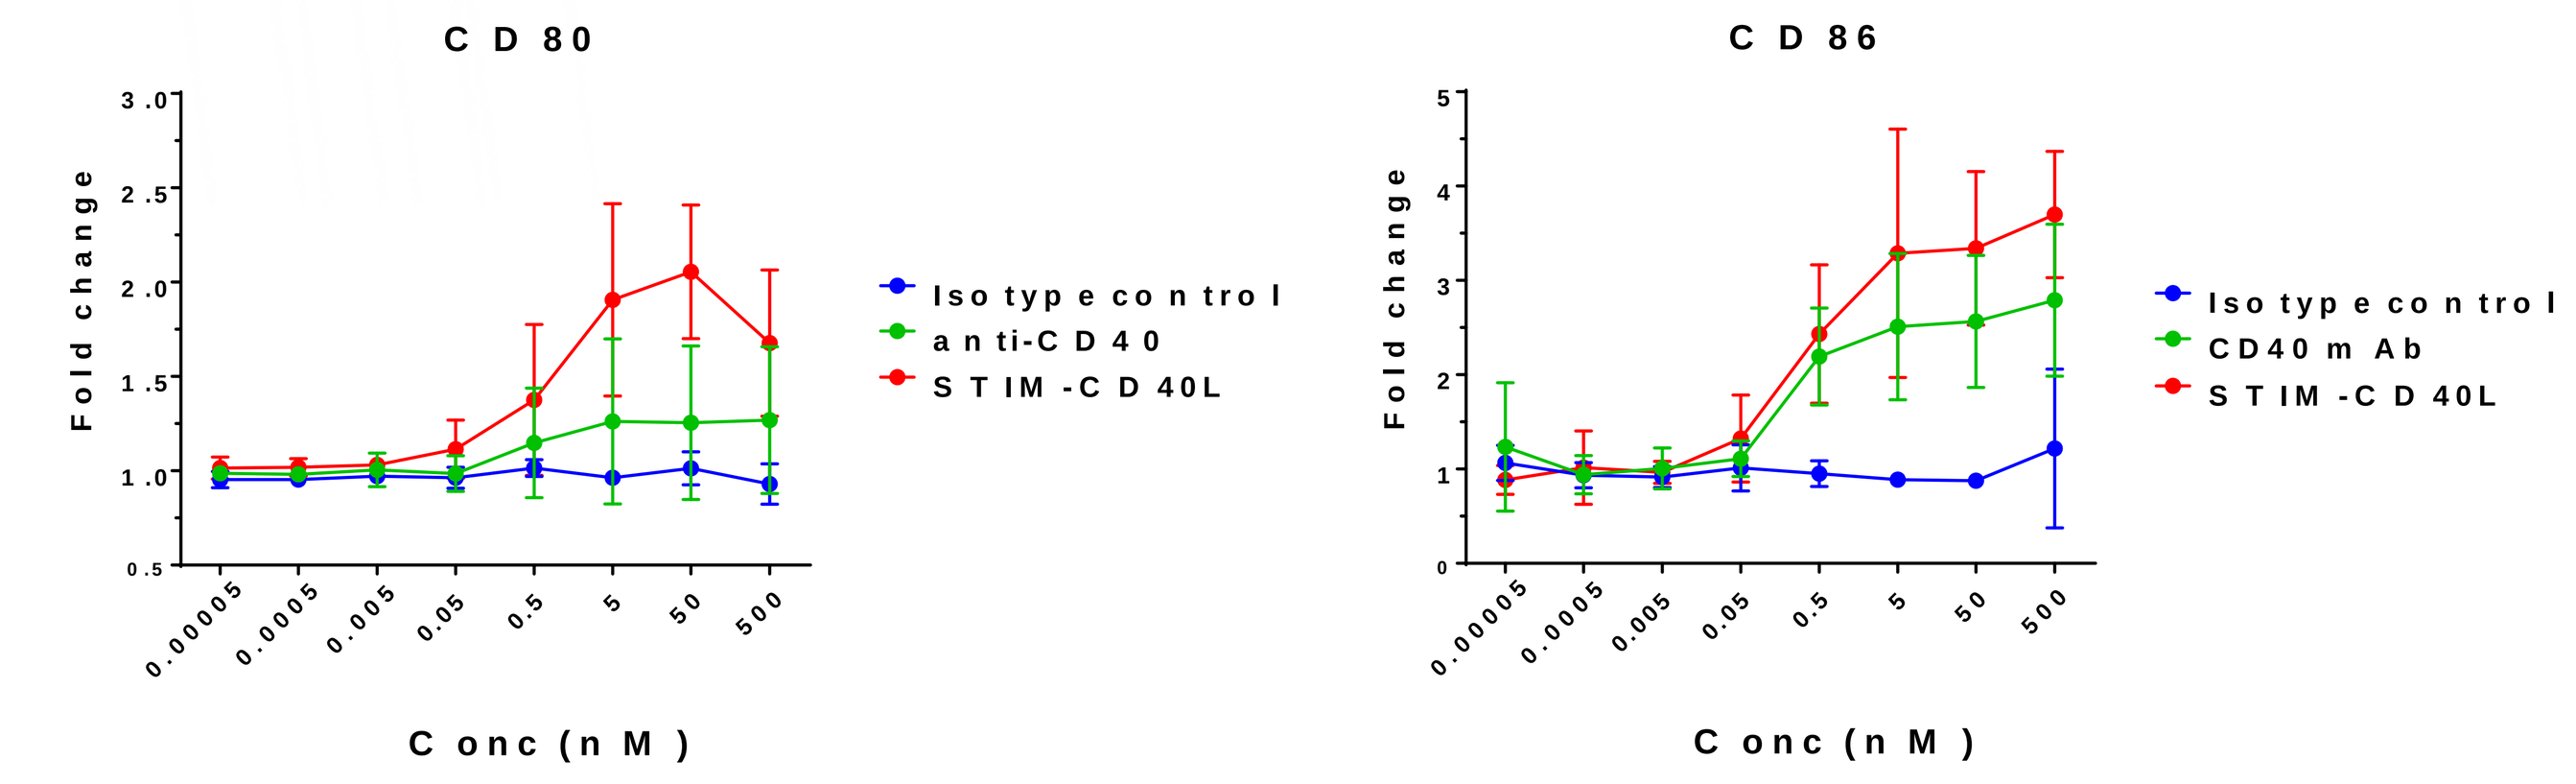
CD80
CD86
Fold change
Conc (nM)
Isotype control
anti-CD40
STIM-CD40L
CD40 mAb

Monocyte-derived DCs purified from whole blood of healthy donors were incubated with STIM-CD40L, anti-CD40 and isotype control for 48 hrs before measuring CD80 and CD86 by flow cytometry.

Human STIMULI molecules induce cytokine production and release

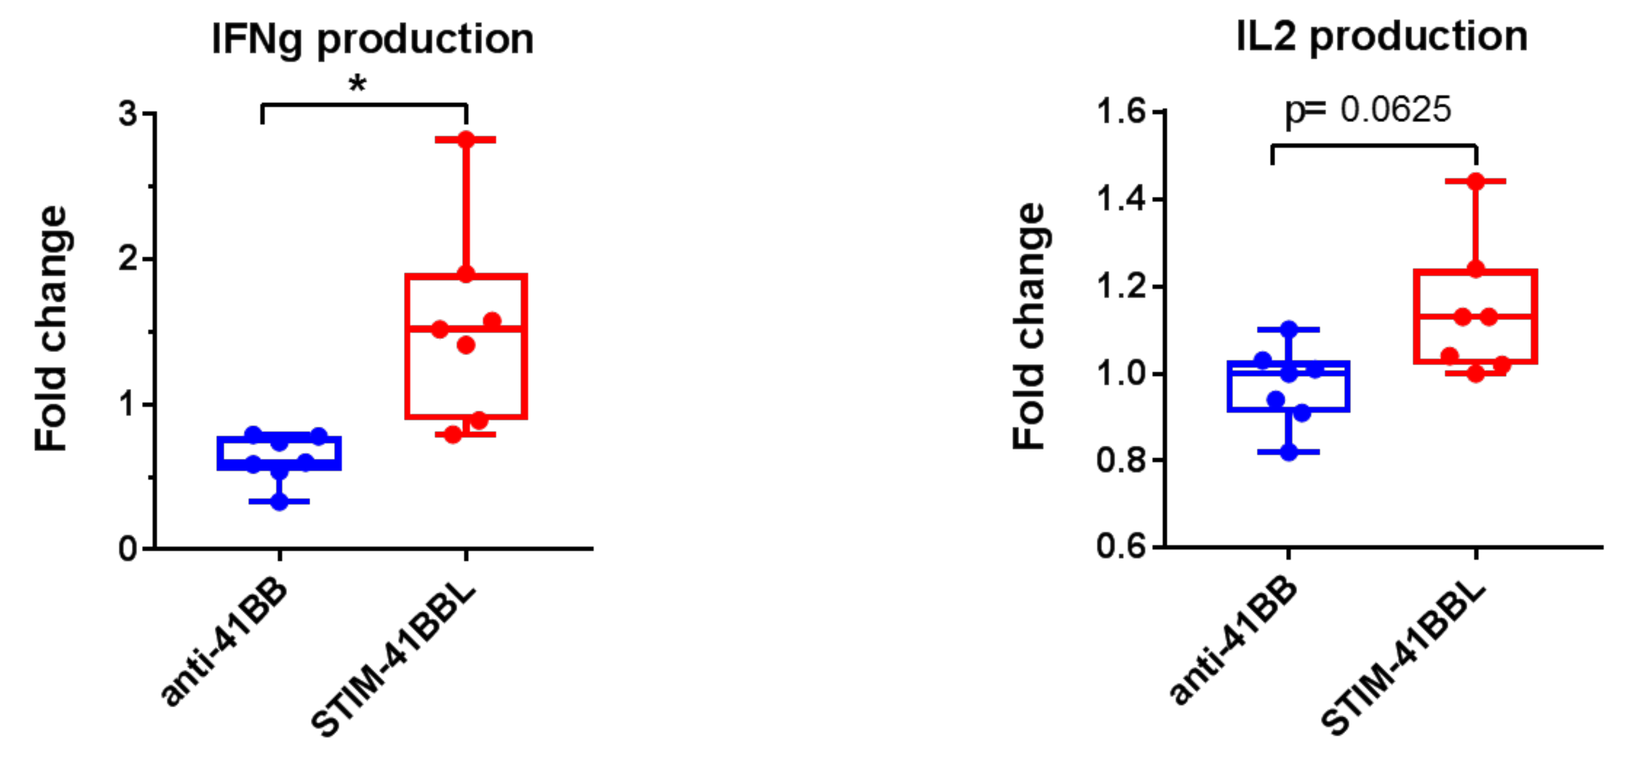
IFNg production
IL2 production
p= 0.0625
Fold change
anti-41BB
STIM-41BBL

CD8+ T cells were isolated from whole blood of healthy donors and stimulated with 1 μg/mL plate-bound CD3 and either 70 nM of STIM-41BBL, anti-41BB and isotype controls. After 72 hr incubation, IFN gamma, and IL2 were measured by ELISA.

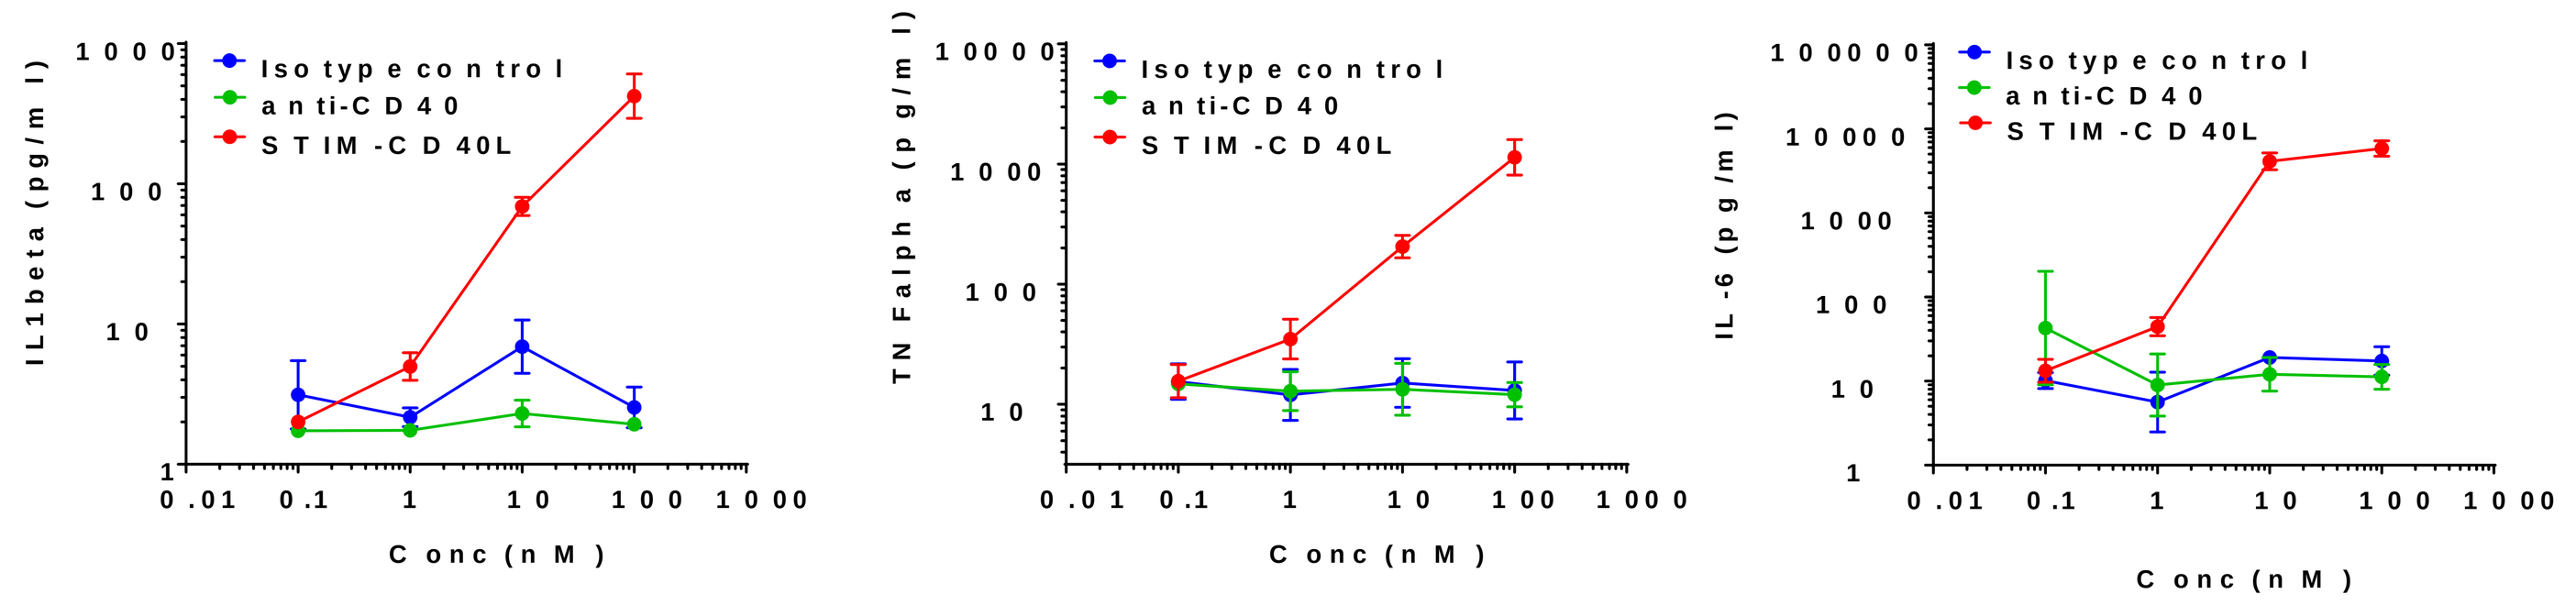
Isotype control
anti-CD40
STIM-CD40L
IL1beta (pg/ml)
TNFalpha (pg/ml)
IL-6 (pg/ml)
Conc (nM)

PBMCs were isolated from whole blood of healthy donors and stimulated with STIM-CD40L, anti-CD40 and isotype controls. After 24 hr incubation, IL-1beta, TNFalpha, and IL-6 were measured using Luminex.

Murine STIM-41BBL promotes T cell activation and cytokine release *in vitro*

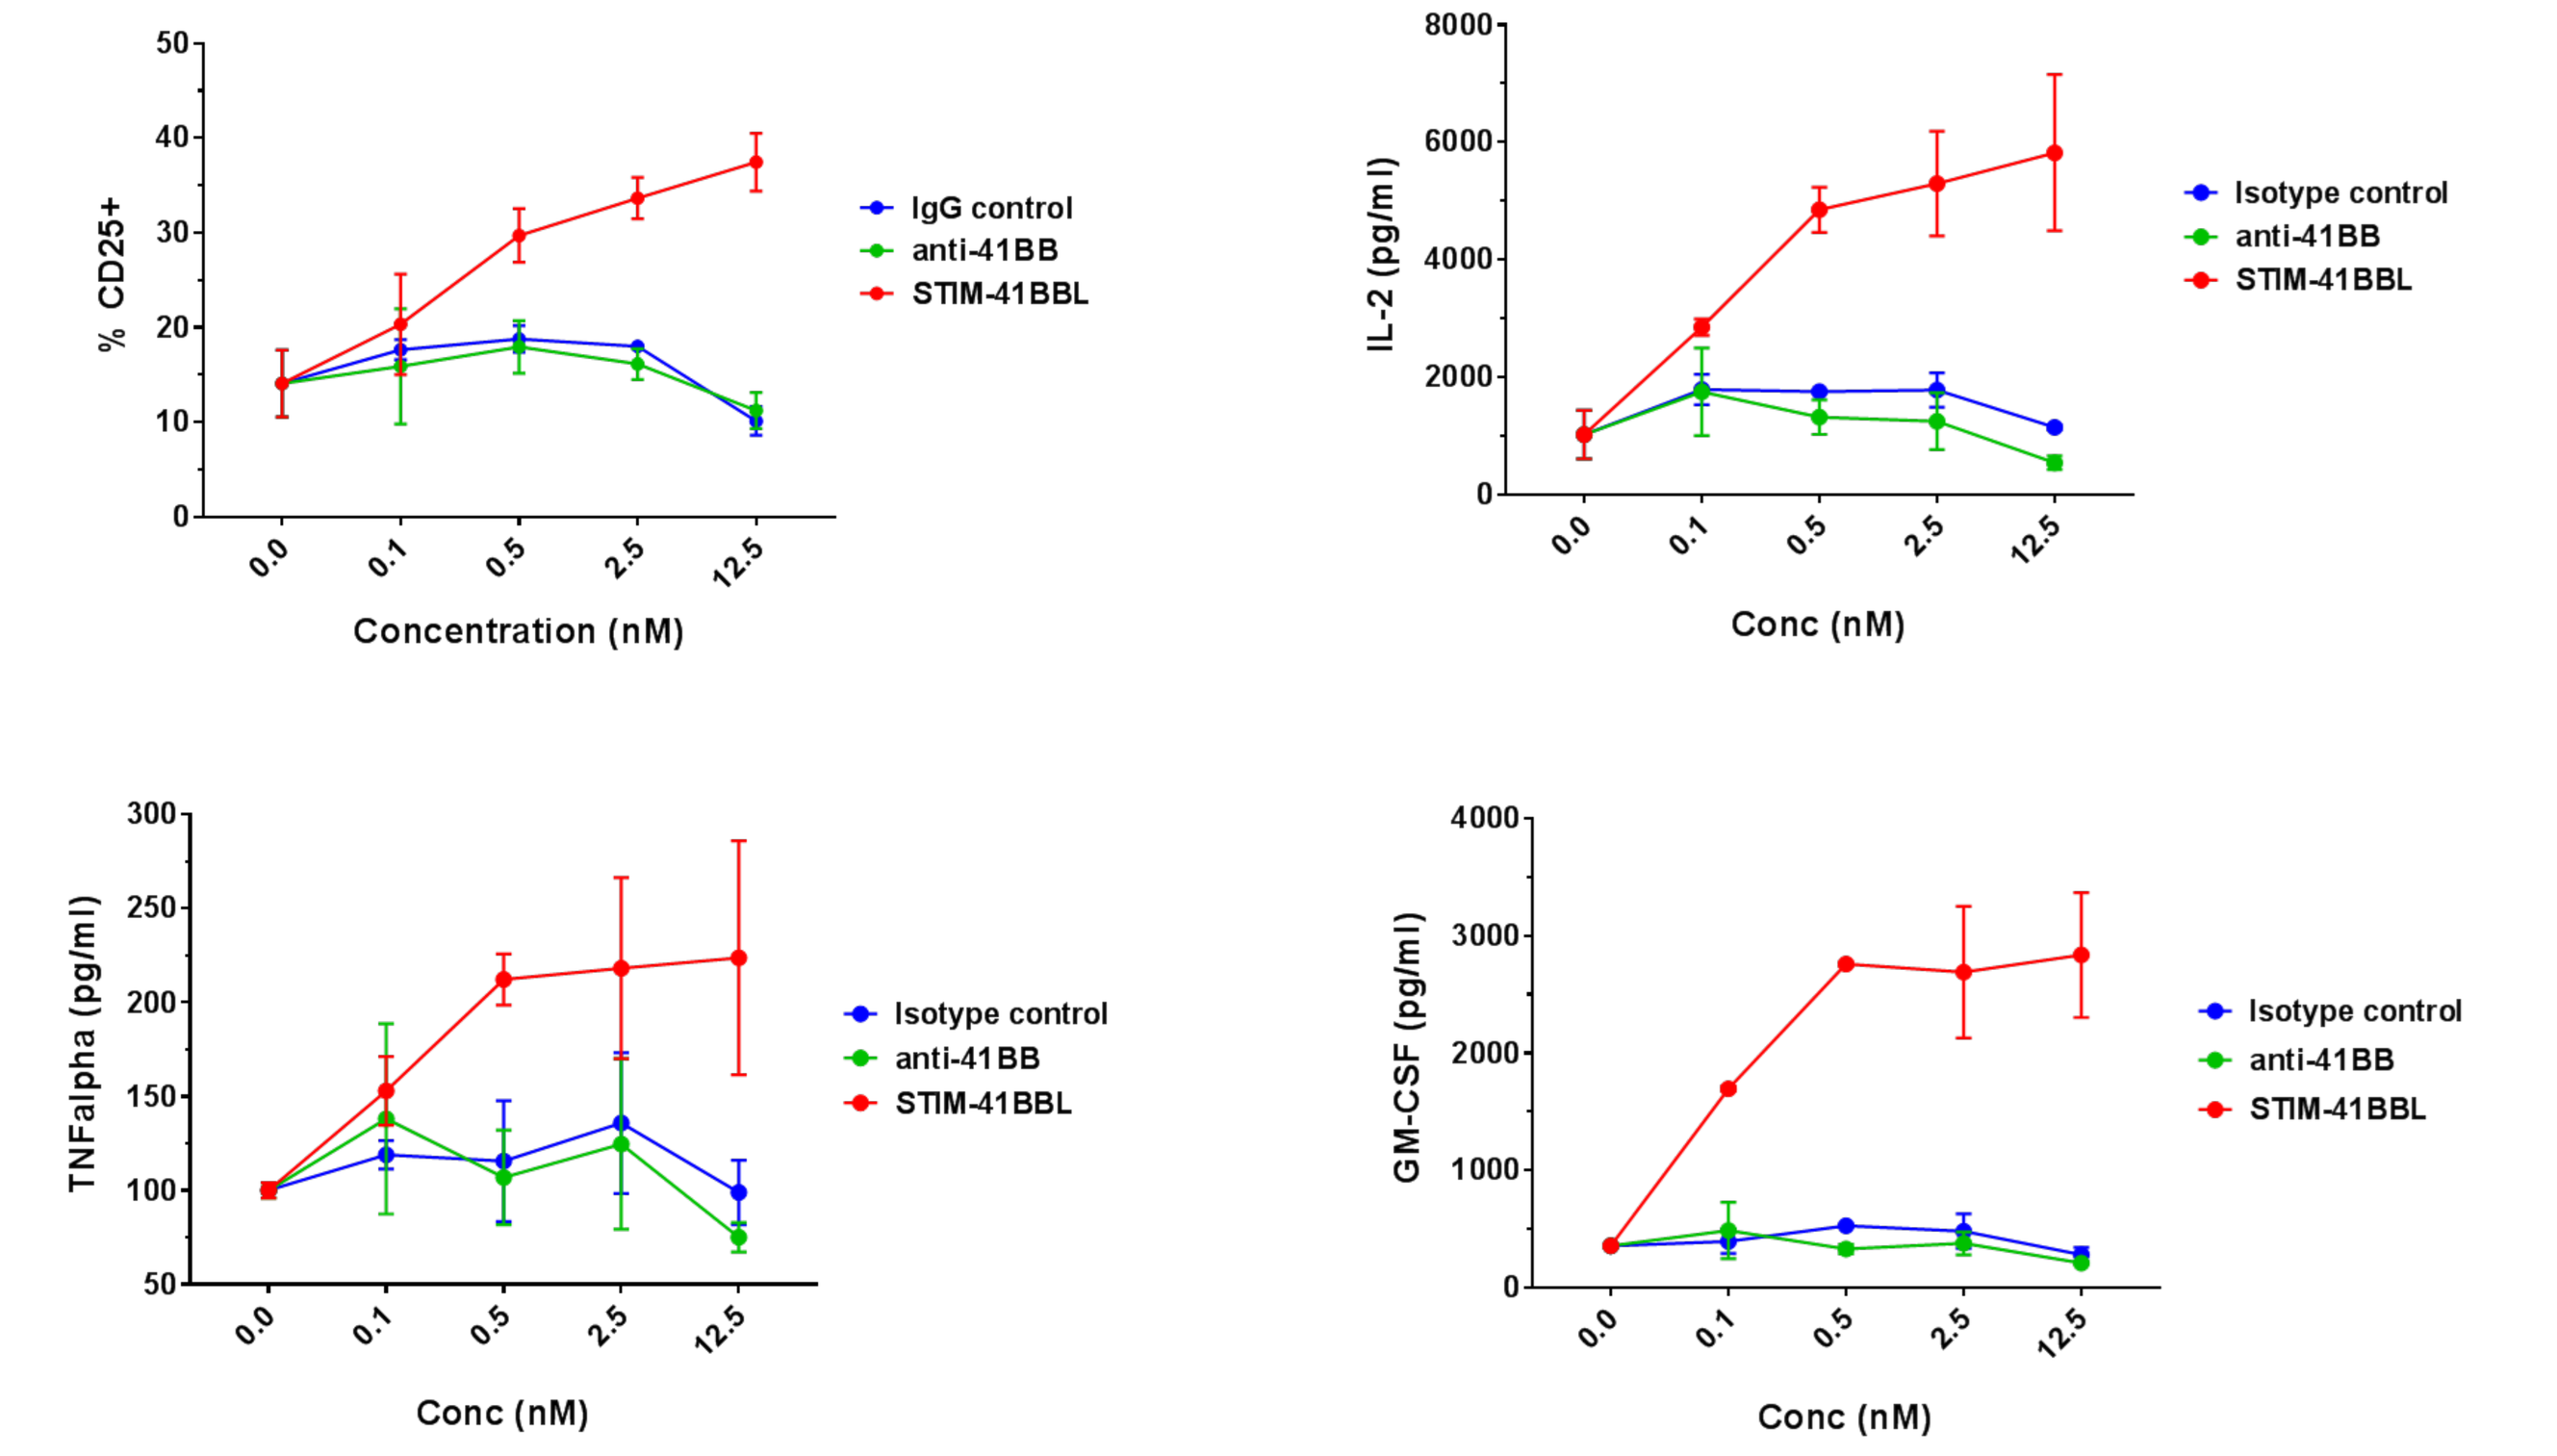
IgG control
Isotype control
anti-41BB
STIM-41BBL
% CD25+
IL-2 (pg/ml)
TNFalpha (pg/ml)
GM-CSF (pg/ml)
Concentration (nM)
Conc (nM)

CD8+ T cells were isolated from mice and stimulated with 1 μg/mL plate-bound CD3 in the presence of STIM-41BBL, anti-41BB and isotype controls. After 72 hrs of incubation, the percentage of CD25+ CD8 cells were measured by flow cytometry while IL-2, TNF-alpha, and GM-CSF were measured by Luminex.

STIM-41BBL stimulates T cell proliferation and activation in OT-1 model

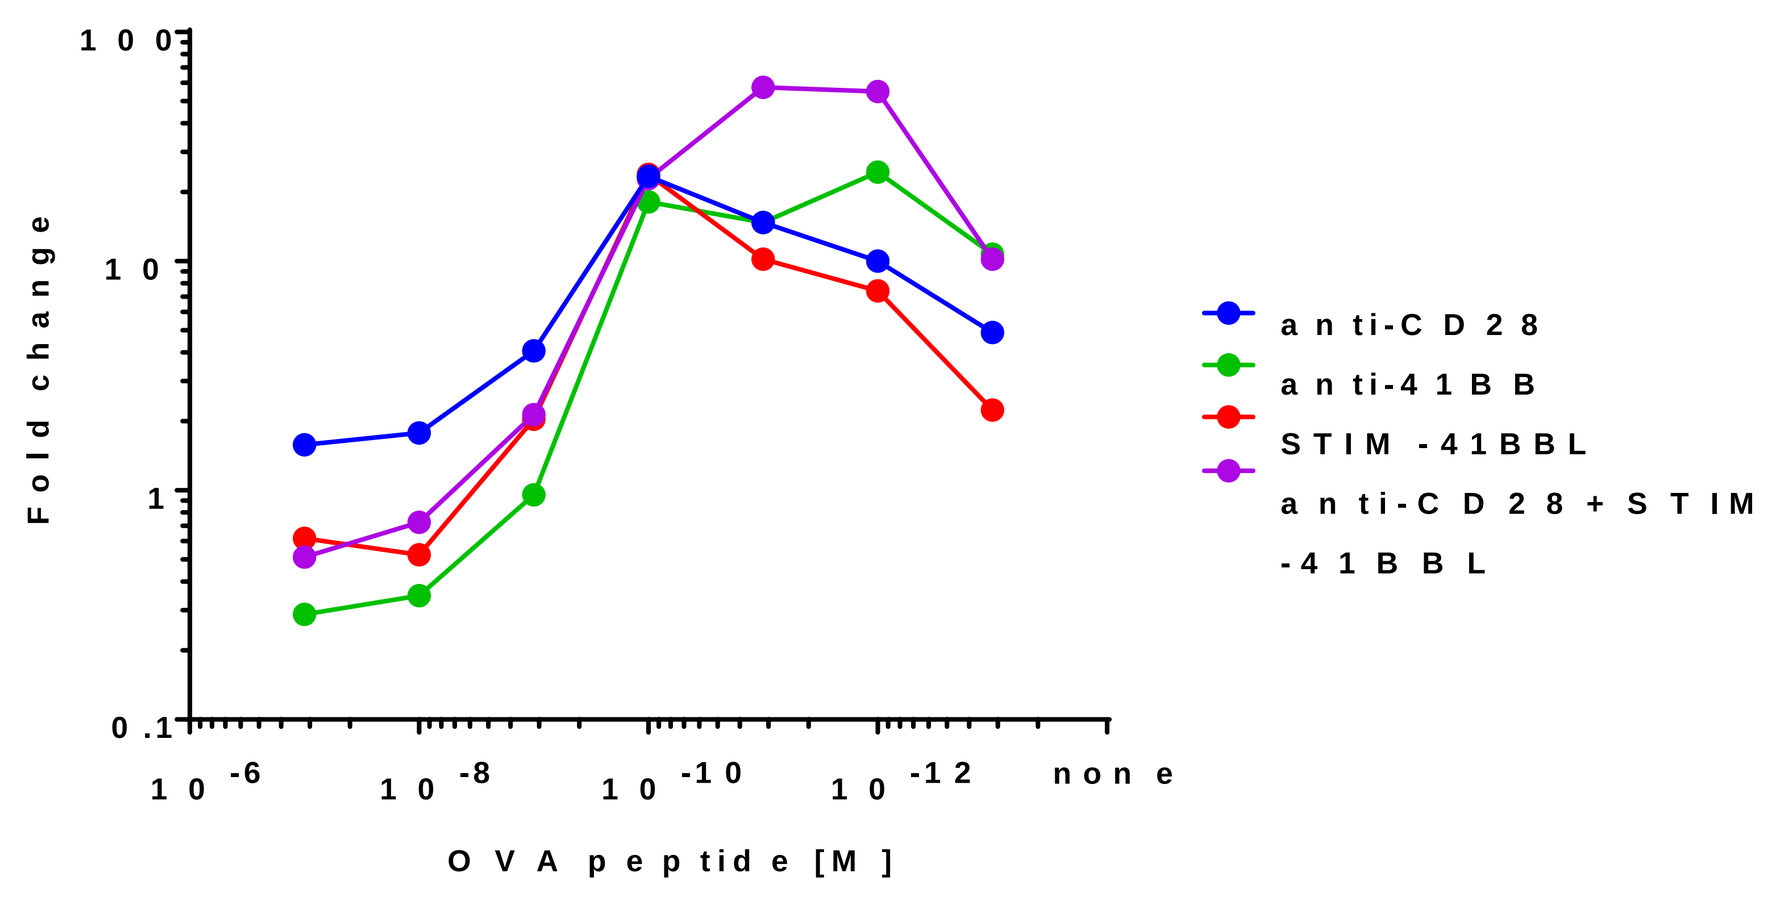
Fold change
OVA peptide [M]
none
anti-CD28
anti-41BB
STIM-41BBL
anti-CD28 + STIM-41BBL

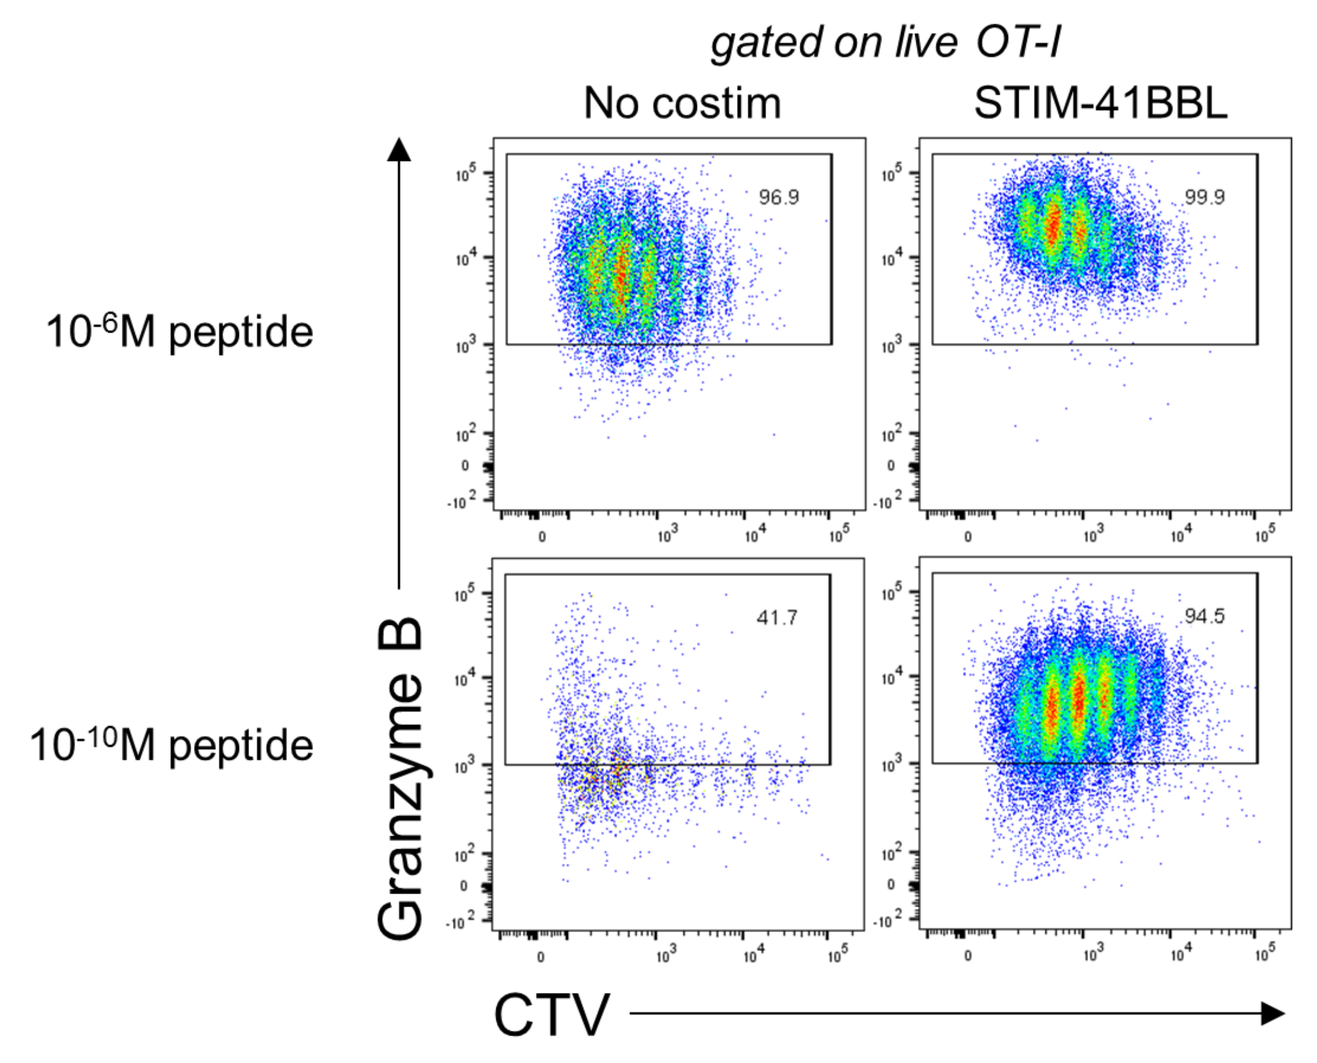
gated on live OT-I
No costim
STIM-41BBL
10⁻⁶M peptide
10⁻¹⁰M peptide
Granzyme B
CTV

Wild-type splenocytes were pulsed with increasing $OVA_{257\text{-}264}$ peptide, treated with mitomycin C and then co-cultured with OT-1 CD8+ T cells labelled with cell trace violet (CTV) and in the absence or presence of STIM-41BBL (10 nM), anti-41BB (10 nM), and anti-CD28 (1 μg/mL). Following 72 hr incubation, OT-1 CD8+ T cells were counted and stained for Granzyme B.

Tumor derived NK cells are preferentially activated by STIM-41BBL in CT26 model

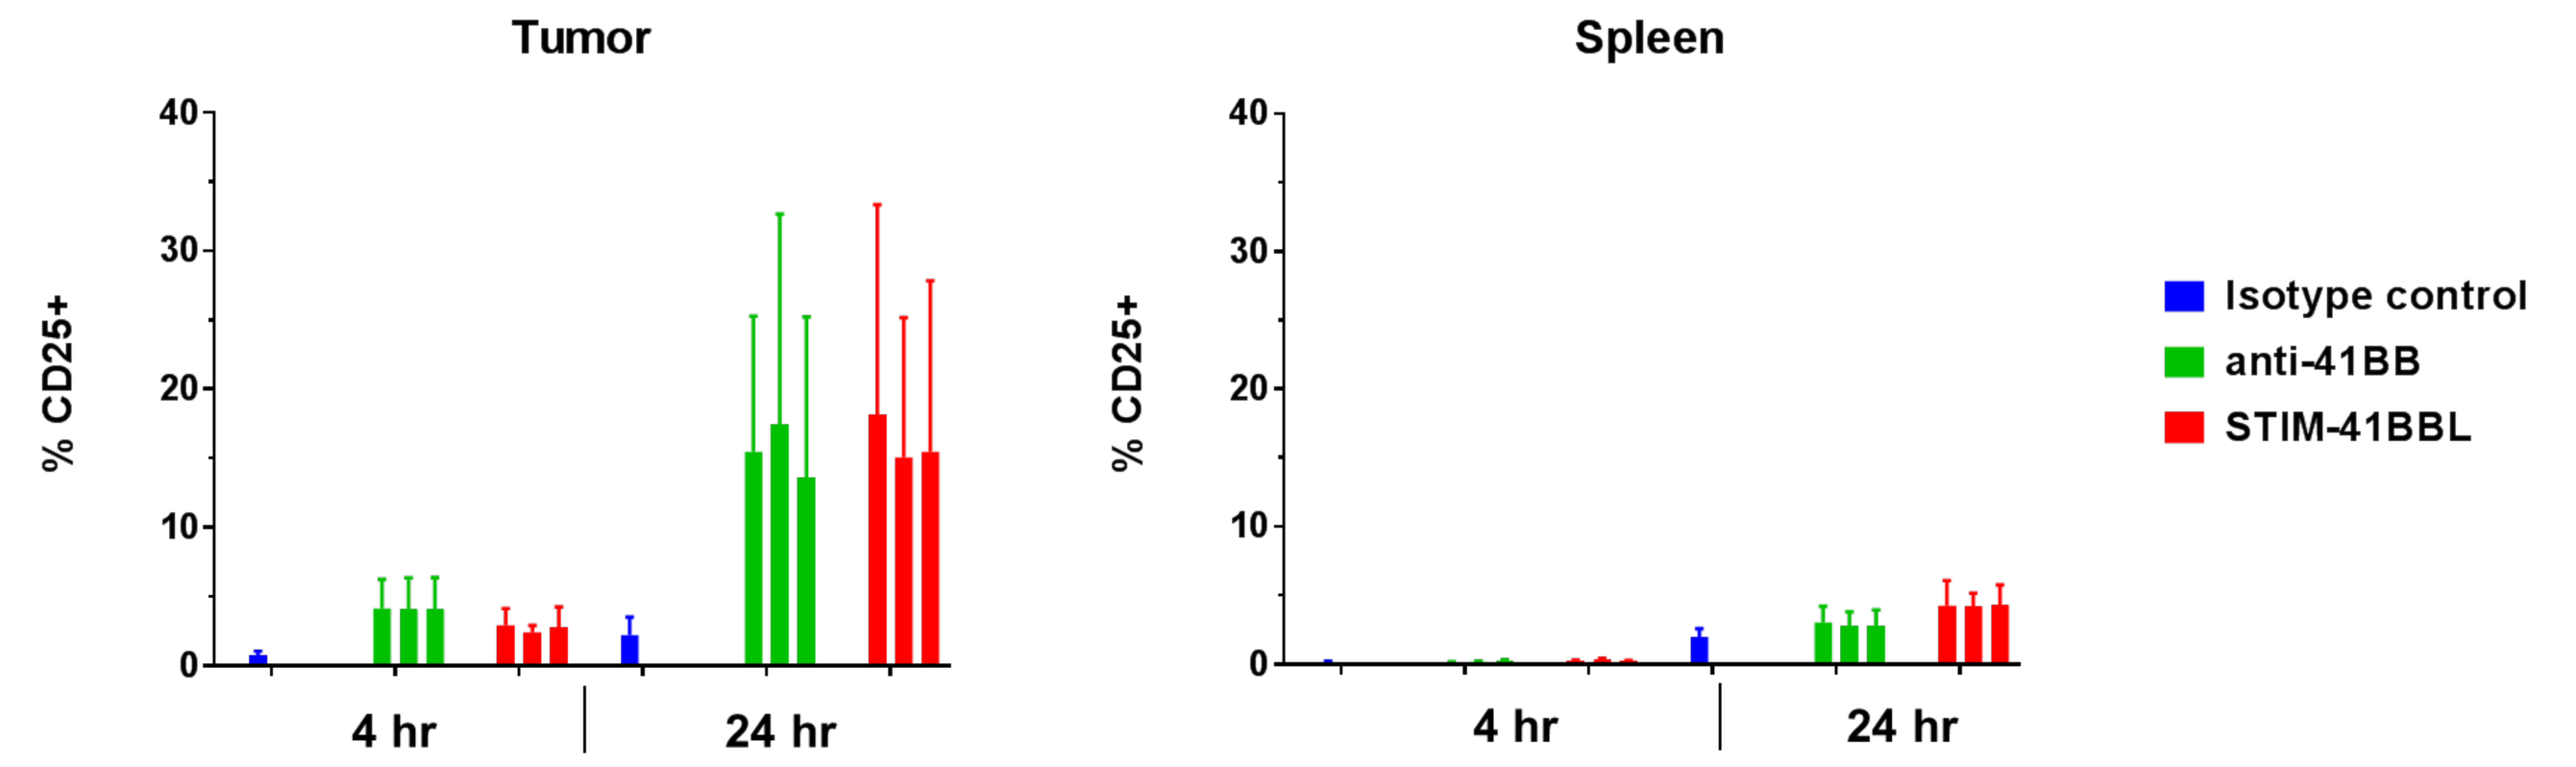
Tumor
Spleen
% CD25+
4 hr
24 hr
Isotype control
anti-41BB
STIM-41BBL

C57BL/6 mice were injected with 1 million CT26 cells. Thirty days post injection, tumors and spleens were harvested. NK cells were isolated and stimulated for 4 and 24 hrs with 10, 100 and 500 nM of isotype control, anti-41BB, and STIM-41BBL. The percentage of CD25+ NK cells were measured by flow cytometry.

STIM-41BBL synergizes with anti-PD1 in a MC38 syngeneic colorectal tumor model

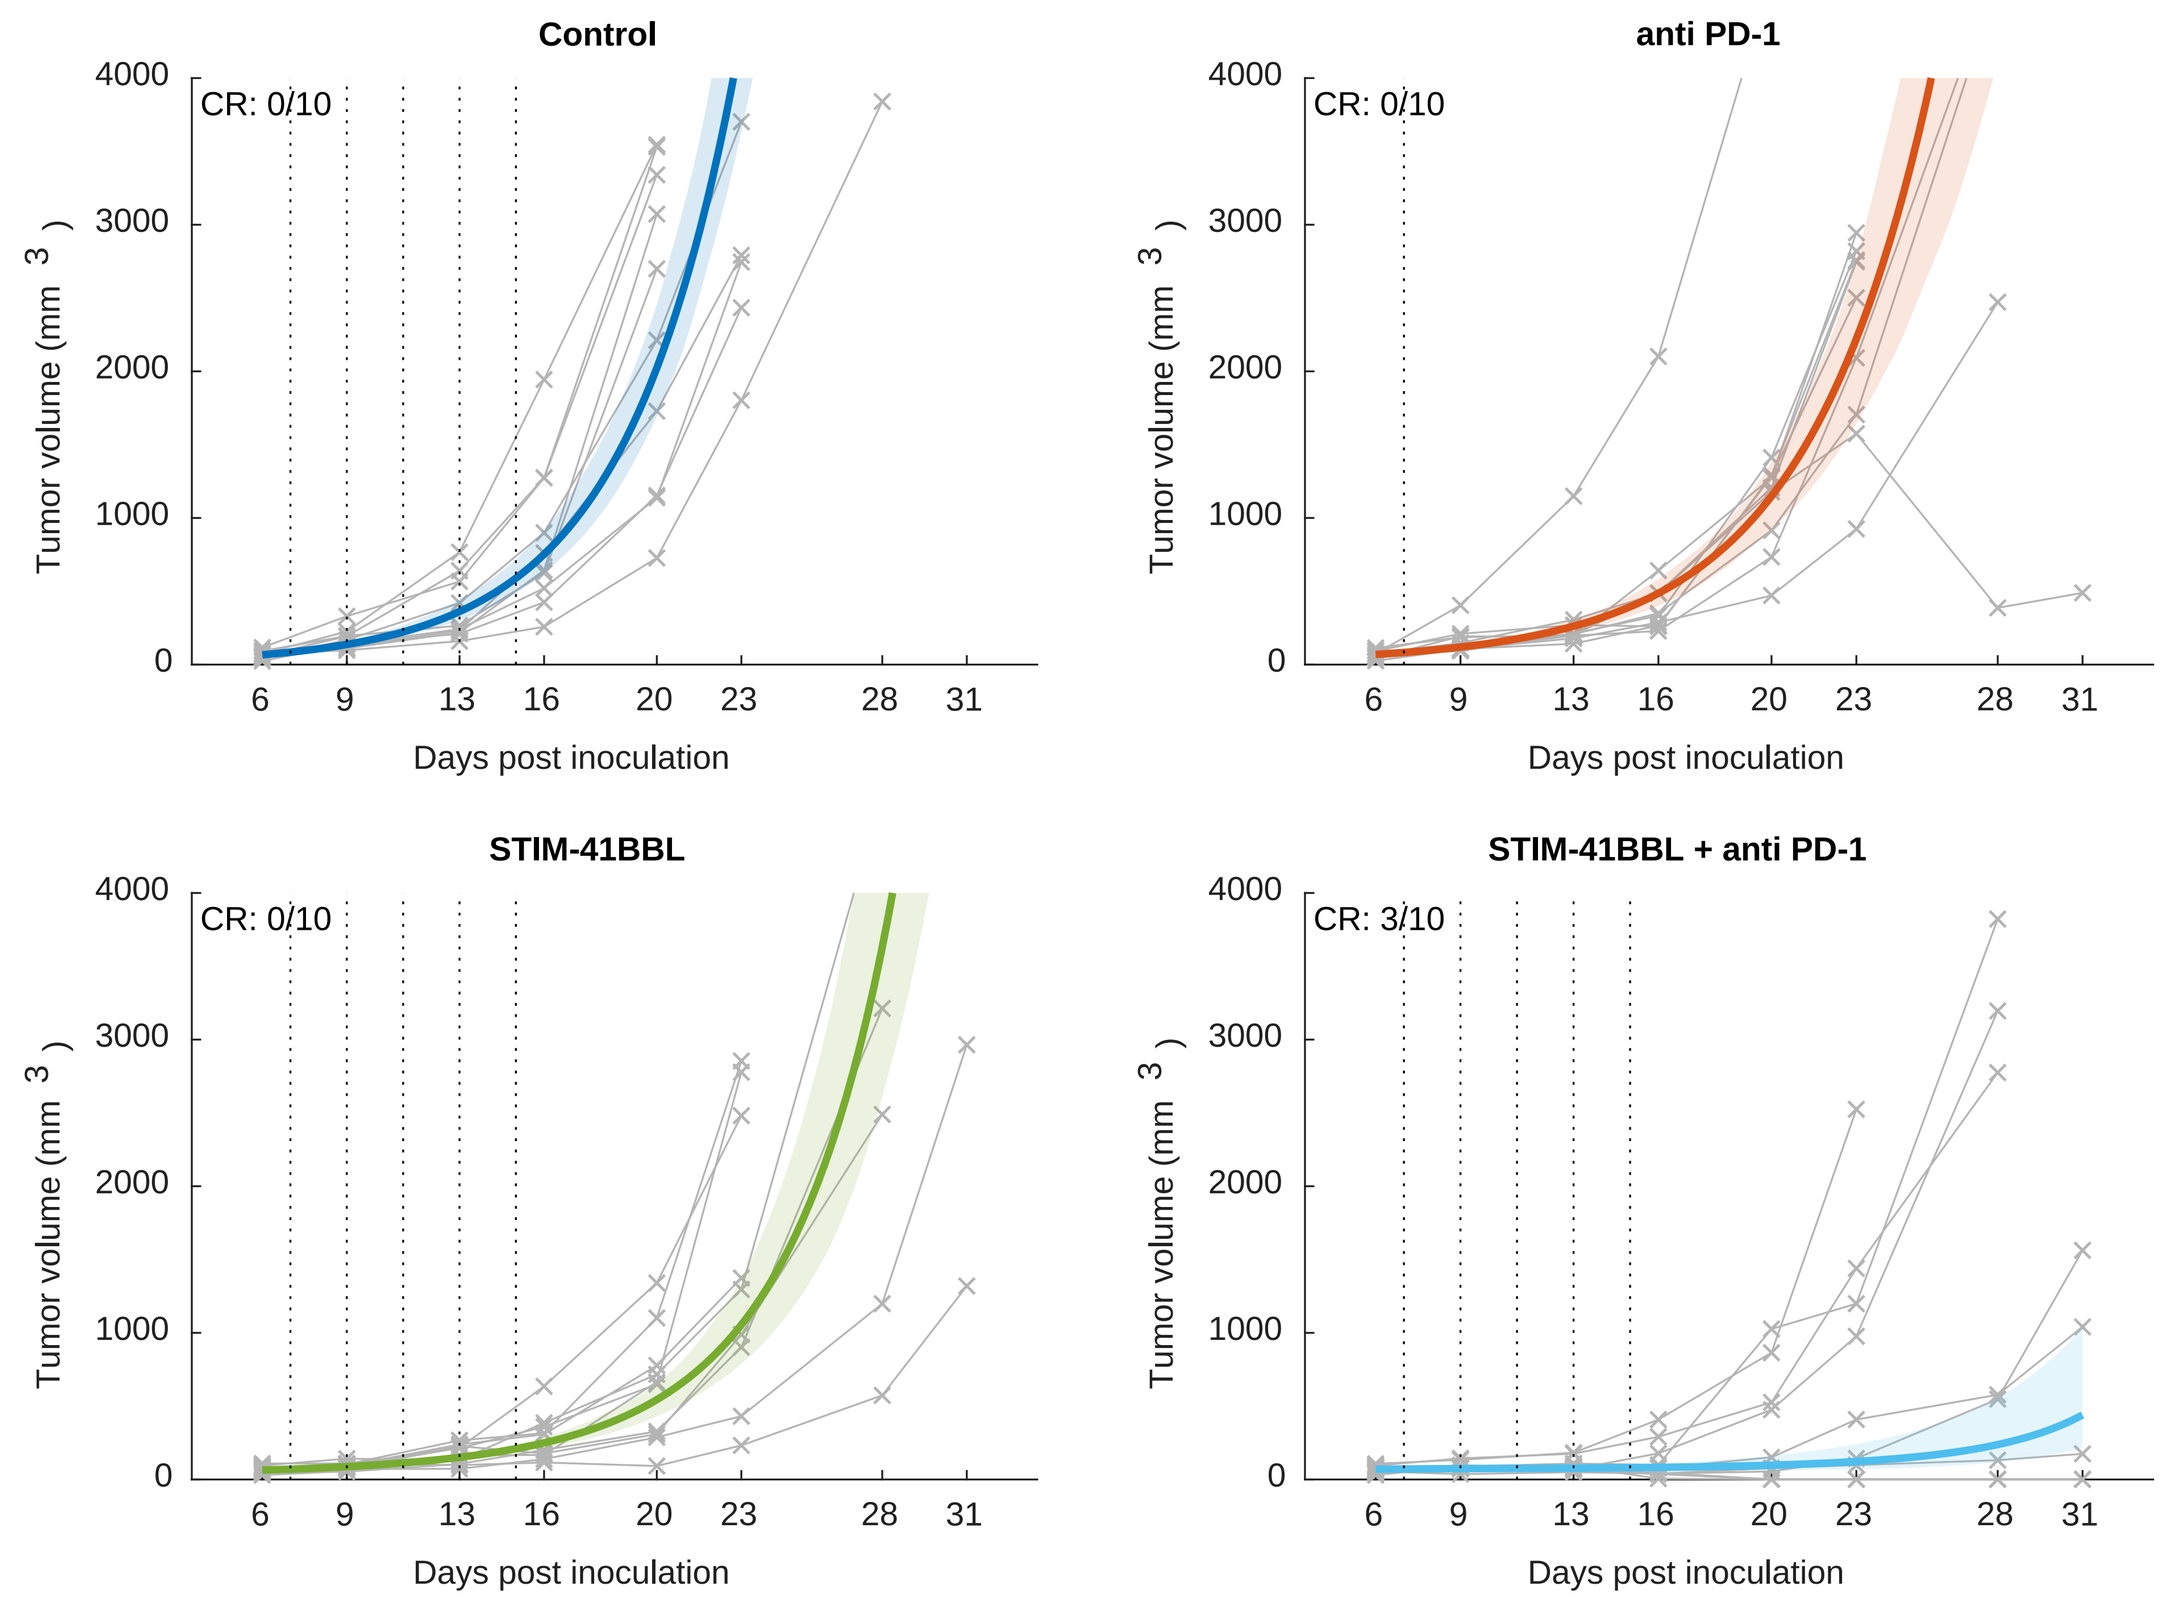
Control
anti PD-1
STIM-41BBL
STIM-41BBL + anti PD-1
CR: 0/10
CR: 0/10
CR: 0/10
CR: 3/10
Tumor volume (mm³)
Days post inoculation

C57BL/6 mice were injected with 1 million MC38 cells. Six days post-injection, mice were randomized and treated with PBS (control), anti-PD-1 (5 mg/kg on D7), STIM-41BBL (10 mg/kg on D7, D9, D11, D13, and D15) or the combination of STIM-41BBL and anti PD-1.

Conclusions

- Human STIMULI molecules showed greater activity than agonistic antibodies both in immune cell activation and in cytokine release.
- Murine STIM-41BBL is synergistic with anti-PD1 in a MC38 syngeneic colorectal tumor model.
- Further *in vivo* work with murine STIMULI molecules is underway to understand the advantages and limitations of format compared to antibodies.
- The bispecific option of the STIMULI format can potentially deliver improved and precise activation to specific immune cell types thereby expanding the therapeutic window.

A PASSION FOR OUTTHINKING CANCER

© 2018 Merrimack Pharmaceuticals, Inc. All rights reserved.

merrimack