Abstract #3375

Development of a second-generation TRAIL agonist and predictive biomarker profile for colorectal cancer

Sara Ghassemifar[1], Yasmin Hashambhoy-Ramsay[1], Haluk Yuzugullu[1], Tamara Utermark[1], Anahita Daruwalla[1], Yvonne Hua[2], Violette Paragas[1], Andreas Raue[1], Neeraj Kohli[1], Maja Razlog[1], Tim Maiwald[1] , Diane Chai[1], Birgit Schoeberl[1], Patrick C. Reynolds[3], Peter K. Sorger[2], Andrew J. Sawyer[1], Daryl C. Drummond[1] and Eric M. Tam[1]

[1] Merrimack Pharmaceuticals, Inc., Cambridge, Massachusetts, USA, [2] HMS LINCS Center, Harvard Medical School, Boston, Massachusetts, [3] Cancer Center, Texas Tech University Health Sciences Center School of Medicine, Lubbock, Texas, USA

Abstract

INTRODUCTION

RAS mutated cancers comprise of 30-50% of colorectal cancers (CRC), for which there are limited treatment options. A promising therapy is treatment with tumor necrosis factor (TNF)-related apoptosis-inducing ligand (TRAIL) which has been found to selectively induce cell death in ~50% of CRC cell lines independent of RAS mutation status. Unfortunately, first generation TRAIL receptor agonists have had limited clinical success due to weak activity, poor bioavailability and the lack of a predictive biomarker.

RESULTS

To address these limitations, we have developed a long circulating TRAIL receptor agonist, MM-201a, and have identified a companion biomarker strategy for CRC. MM-201a consists of an IgG1 Fc fused to an engineered single chain TRAIL (Fc-scTRAIL). We identified three mutations within TRAIL (R130G/N228S/I247V) from thermal challenge of a random mutagenesis yeast library. Incorporation of these mutations into each TRAIL protomer of MM-201a resulted in 10°C improvement in T_M and a prolonged serum half-life of 39 hours in mice. The activity of MM-201a was observed to be superior to TRAIL and agonist DR4 and DR5 antibodies, and comparable to ABBV621 in the cancer cell lines tested. To identify predictive biomarkers for MM-201a, we used continuous logic gate analysis on a publicly available dataset of cell survival for 27 CRC cell lines treated with cross-linked TRAIL (Nair et al. 2015, PMID: 25902490). Using this dataset and gene expression data from Cancer Cell Line Encyclopedia (CCLE), we explored combinations of 33 genes in the apoptosis pathway and identified a two gene biomarker signature of high DR4 and low cIAP-1 which correlated with cell line sensitivity to TRAIL. Using a biomarker score of [DR4]*[1-cIAP1], we were then able to predict the response of 7/9 new CRC cell lines and 5/5 CRC patient derived xenograft (PDX) models to MM-201a both *in vitro* and *in vivo*. Our 2-gene biomarker is also highly prevalent in CRC tumors (59%) compared to normal (12%) according to TCGA data.

CONCLUSIONS

MM-201a is a potent death receptor agonist with suitable biophysical properties and pharmacokinetics for therapeutic development. In addition, our unique 2-gene biomarker signature in CRC is highly predictive in preclinical models and prevalent in patients. We believe that these two achievements together warrant clinical investigation of MM-201a in CRC.

For more information about MM-201, please see our other poster.
Session PO.MCB02.03 - BCL-2 Family Proteins
2491 / 1 - Engineering and preclinical activity of MM-201, a best-in-class TRAIL receptor agonist

MM-201a – engineered Fc single chain TRAIL fusion protein

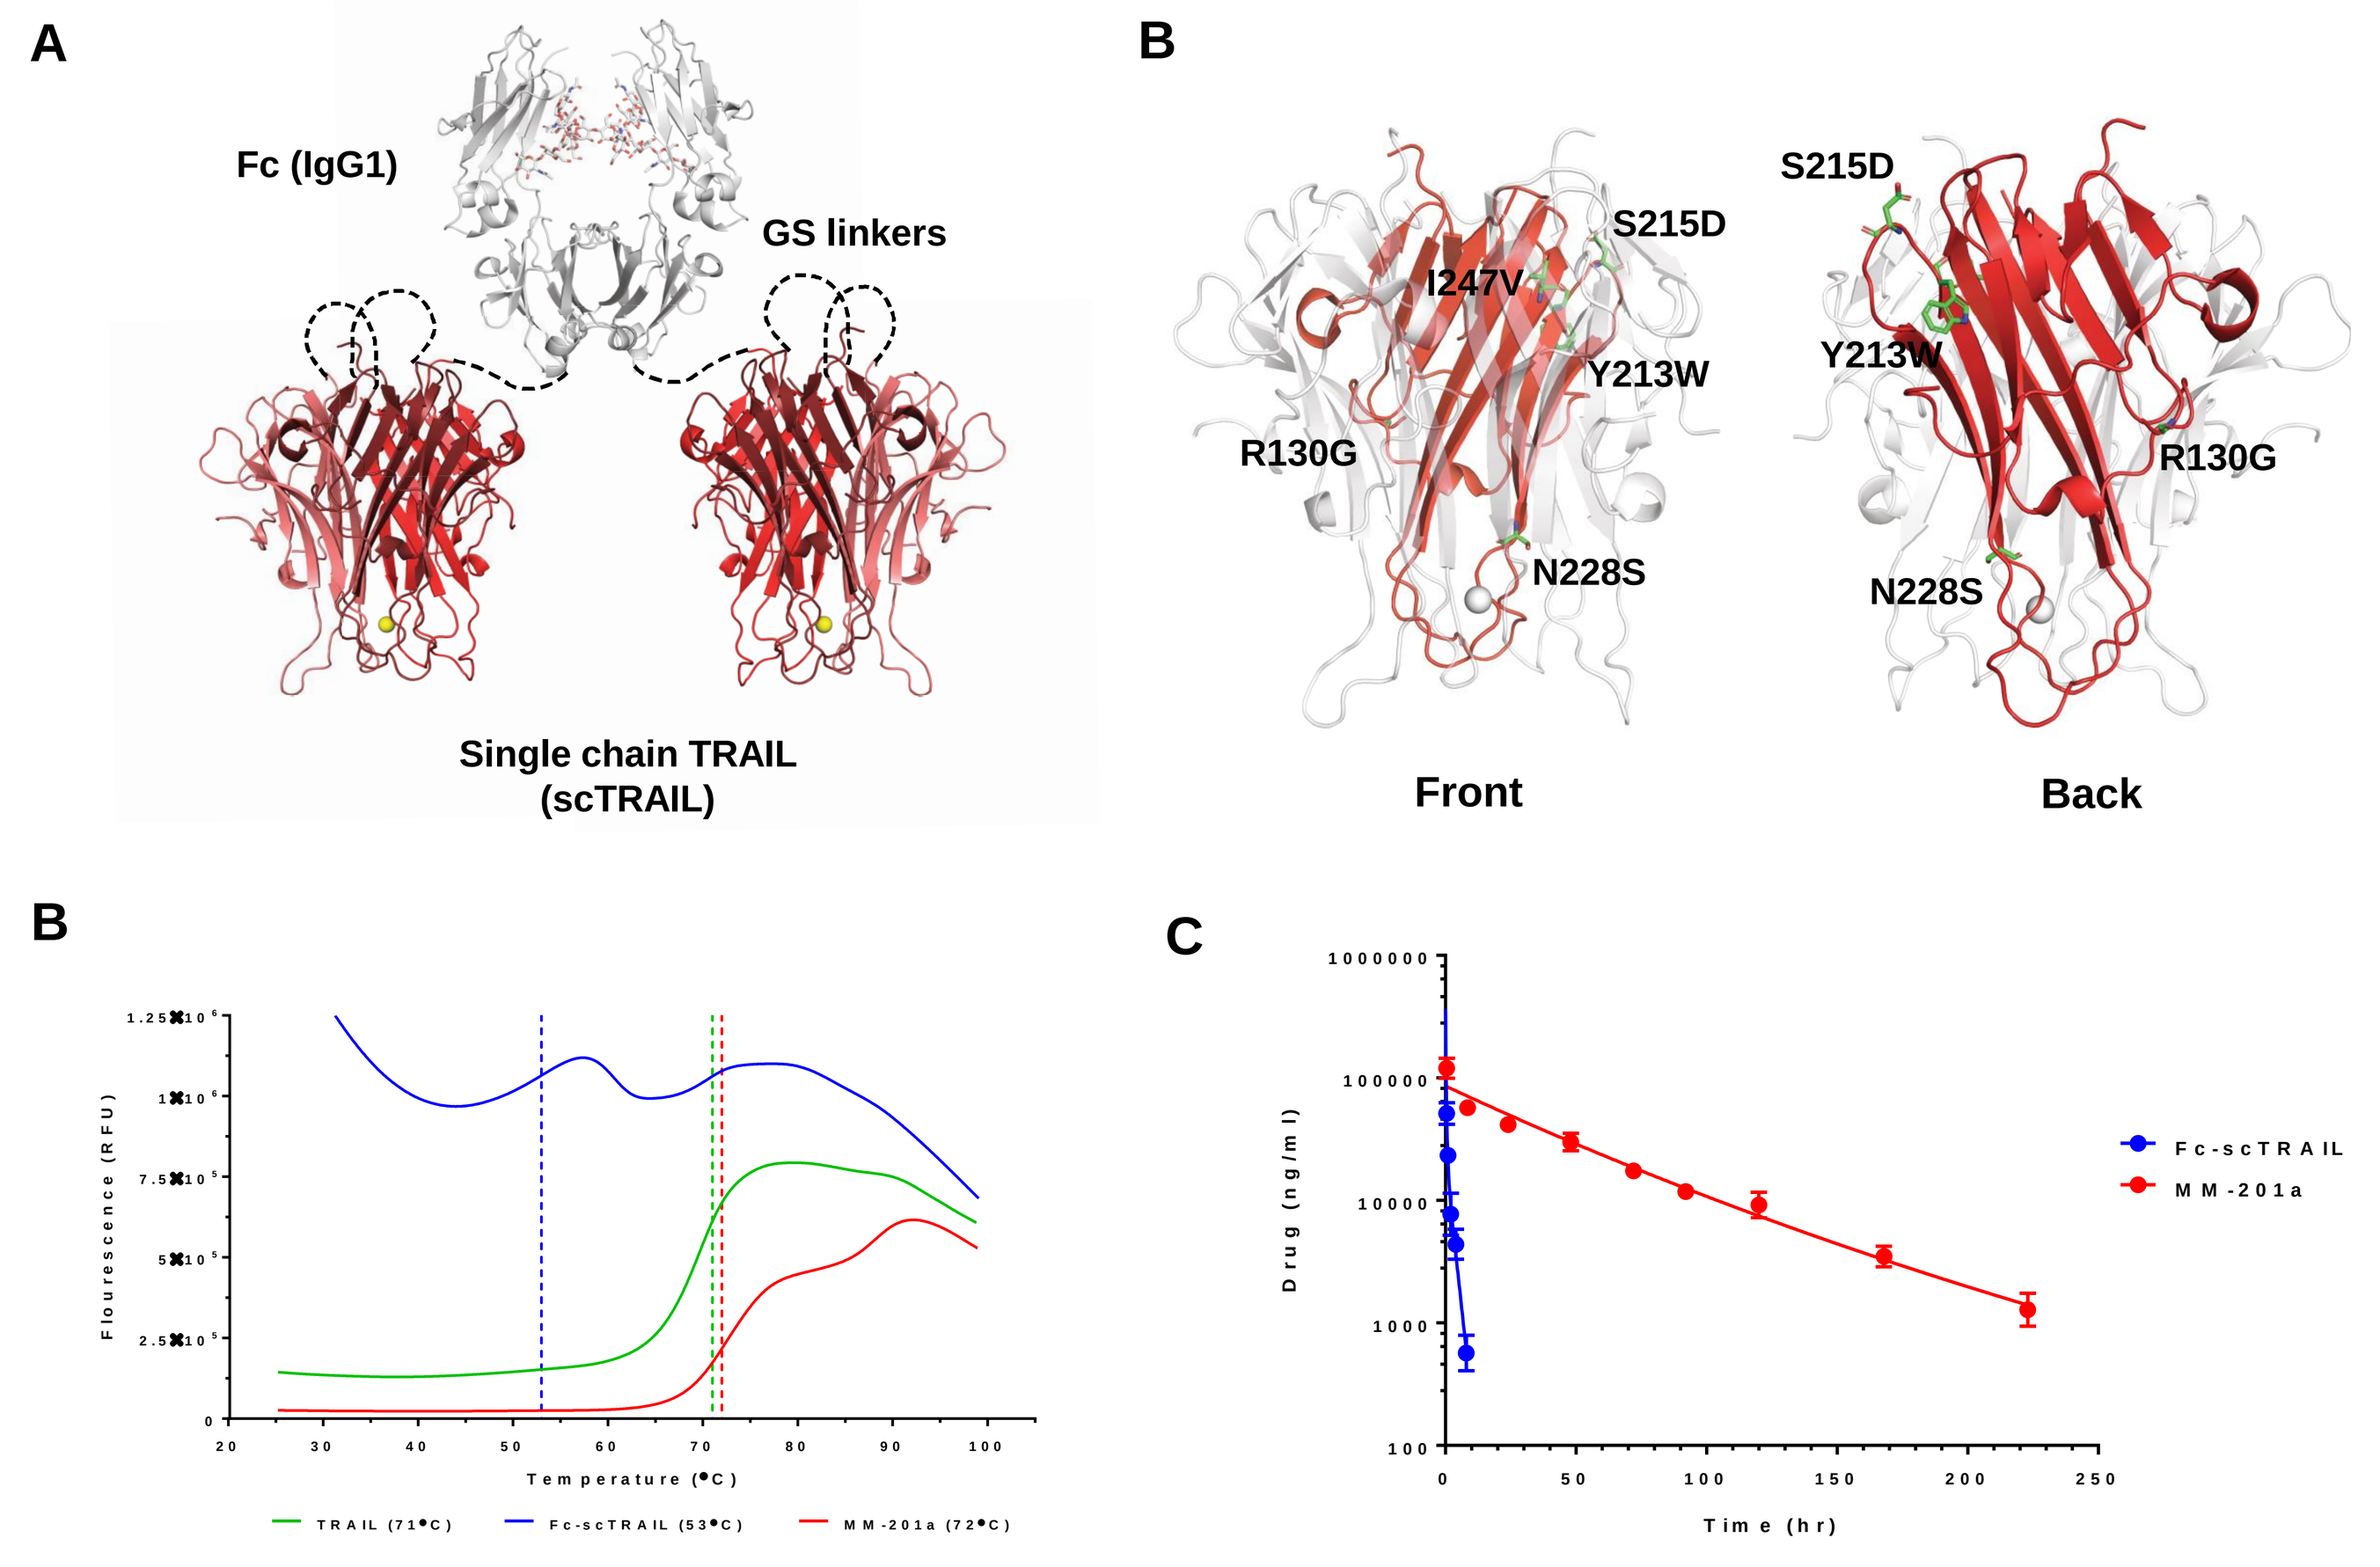
A
Fc (IgG1)
GS linkers
Single chain TRAIL (scTRAIL)
B
S215D
I247V
Y213W
R130G
N228S
Front
Back
B
Temperature (°C)
Flourescence (RFU)
TRAIL (71°C)
Fc-scTRAIL (53°C)
MM-201a (72°C)
C
Drug (ng/ml)
Time (hr)
Fc-scTRAIL
MM-201a

Figure 1. Engineering MM-201a for improved thermostability and pharmacokinetics. (A) Format of the Fc-scTRAIL fusion protein. (B) MM-201a mutations. R130G, N228S, and I247V were identified from thermal challenge (60°C) of a random mutagenesis TRAIL library. Y213W and S215D were previously identified from phage display (Kelley et al. 2005, PMID: 15520016). (C) Differential scanning fluorimetry (DSF) analysis of TRAIL, Fc-scTRAIL and MM-201a at 0.5 mg/ml. (D) Pharmacokinetics of Fc-scTRAIL and MM-201a (5 mg/kg) in mice. Serum drug concentration was measured using DR5 ELISA. The serum half-life determined to be 45 min for Fc-scTRAIL and 39 hrs for MM-201a.

MM-201a compares favorably with competitor death receptor agonists

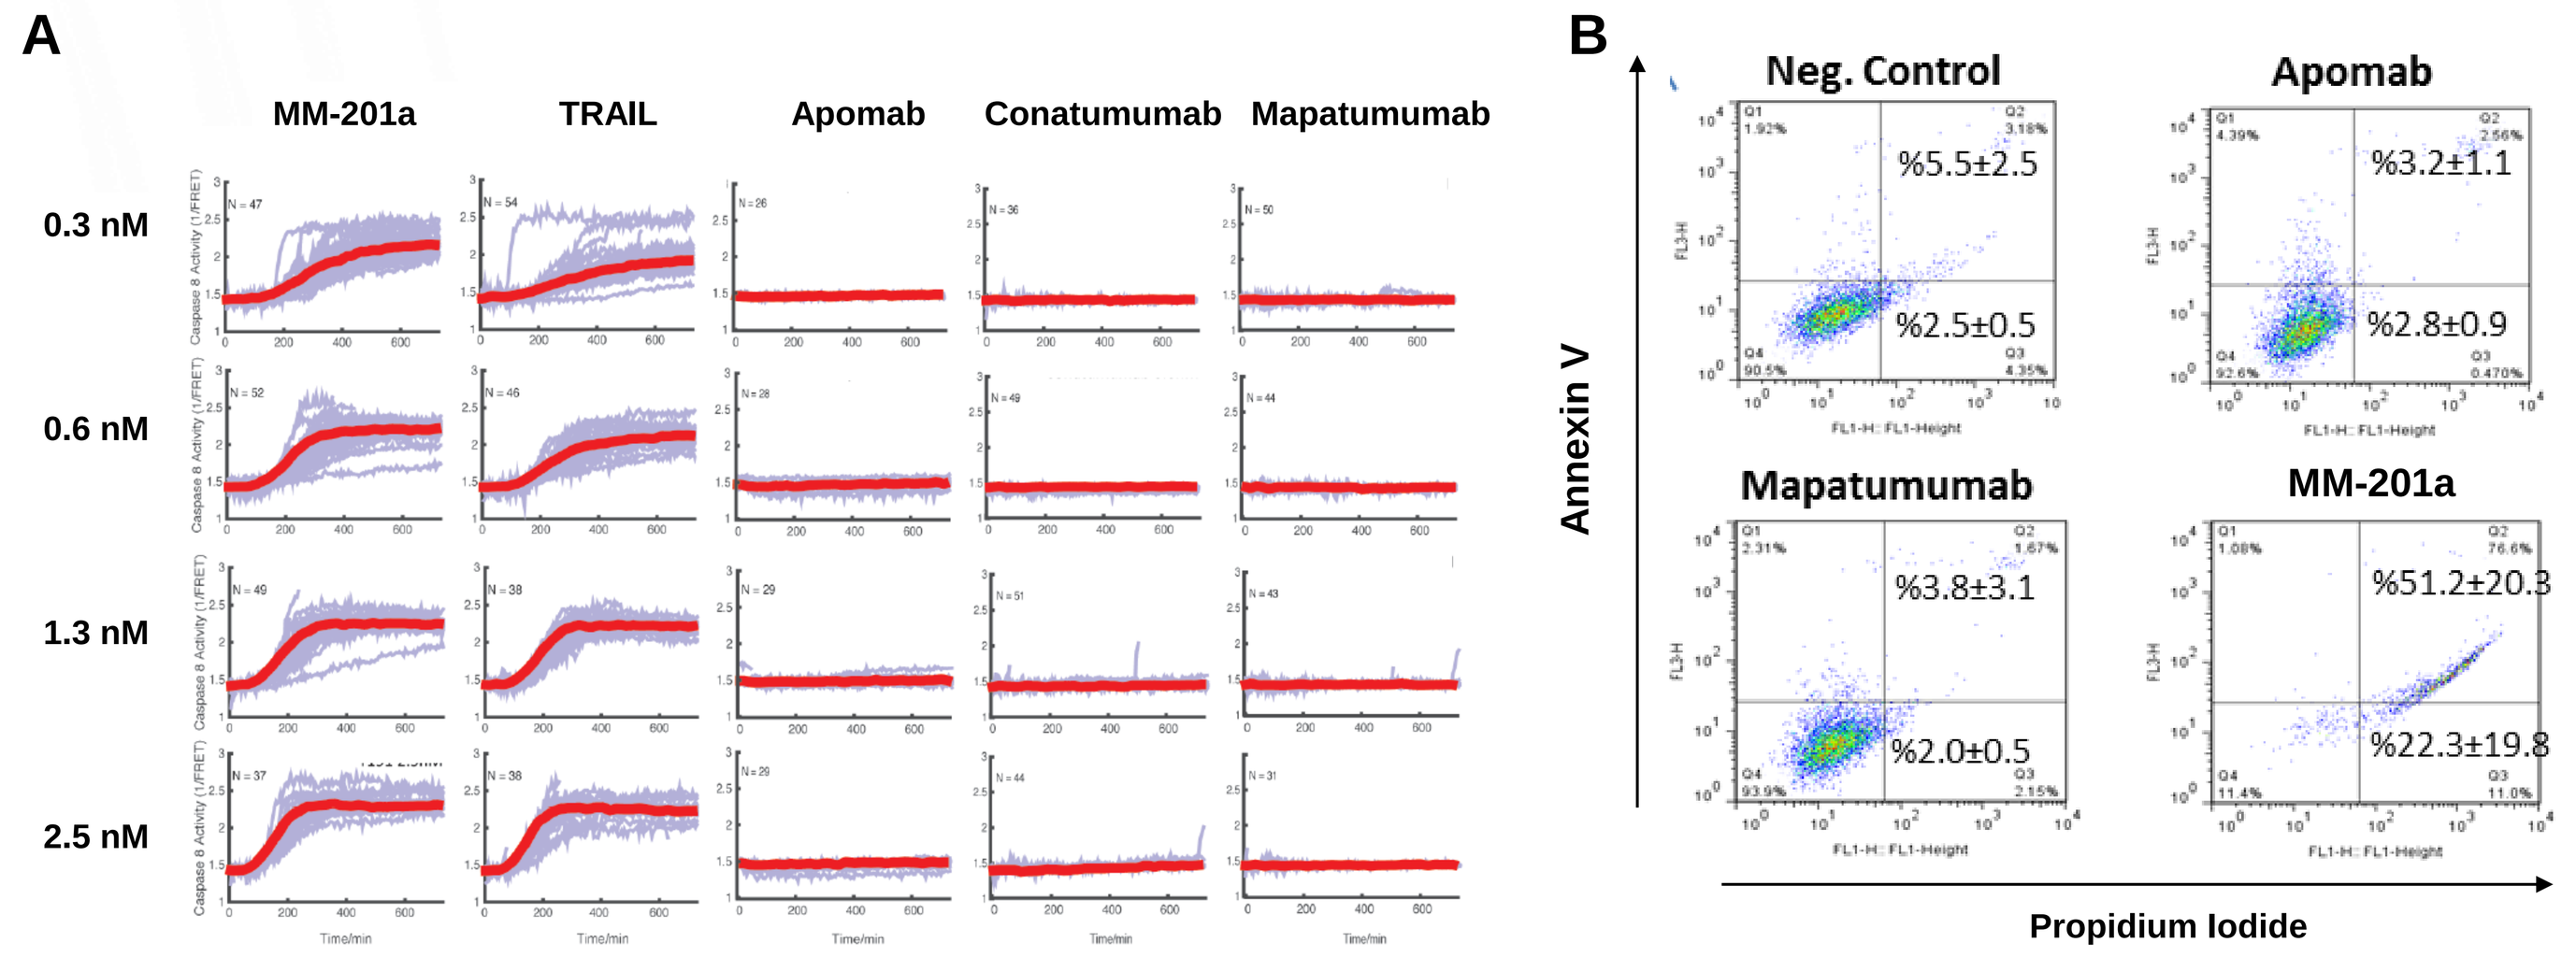
A
MM-201a
TRAIL
Apomab
Conatumumab
Mapatumumab
0.3 nM
0.6 nM
1.3 nM
2.5 nM
B
Neg. Control
Apomab
Mapatumumab
MM-201a
%5.5±2.5
%2.5±0.5
%3.2±1.1
%2.8±0.9
%3.8±3.1
%2.0±0.5
%51.2±20.3
%22.3±19.8
Annexin V
Propidium Iodide

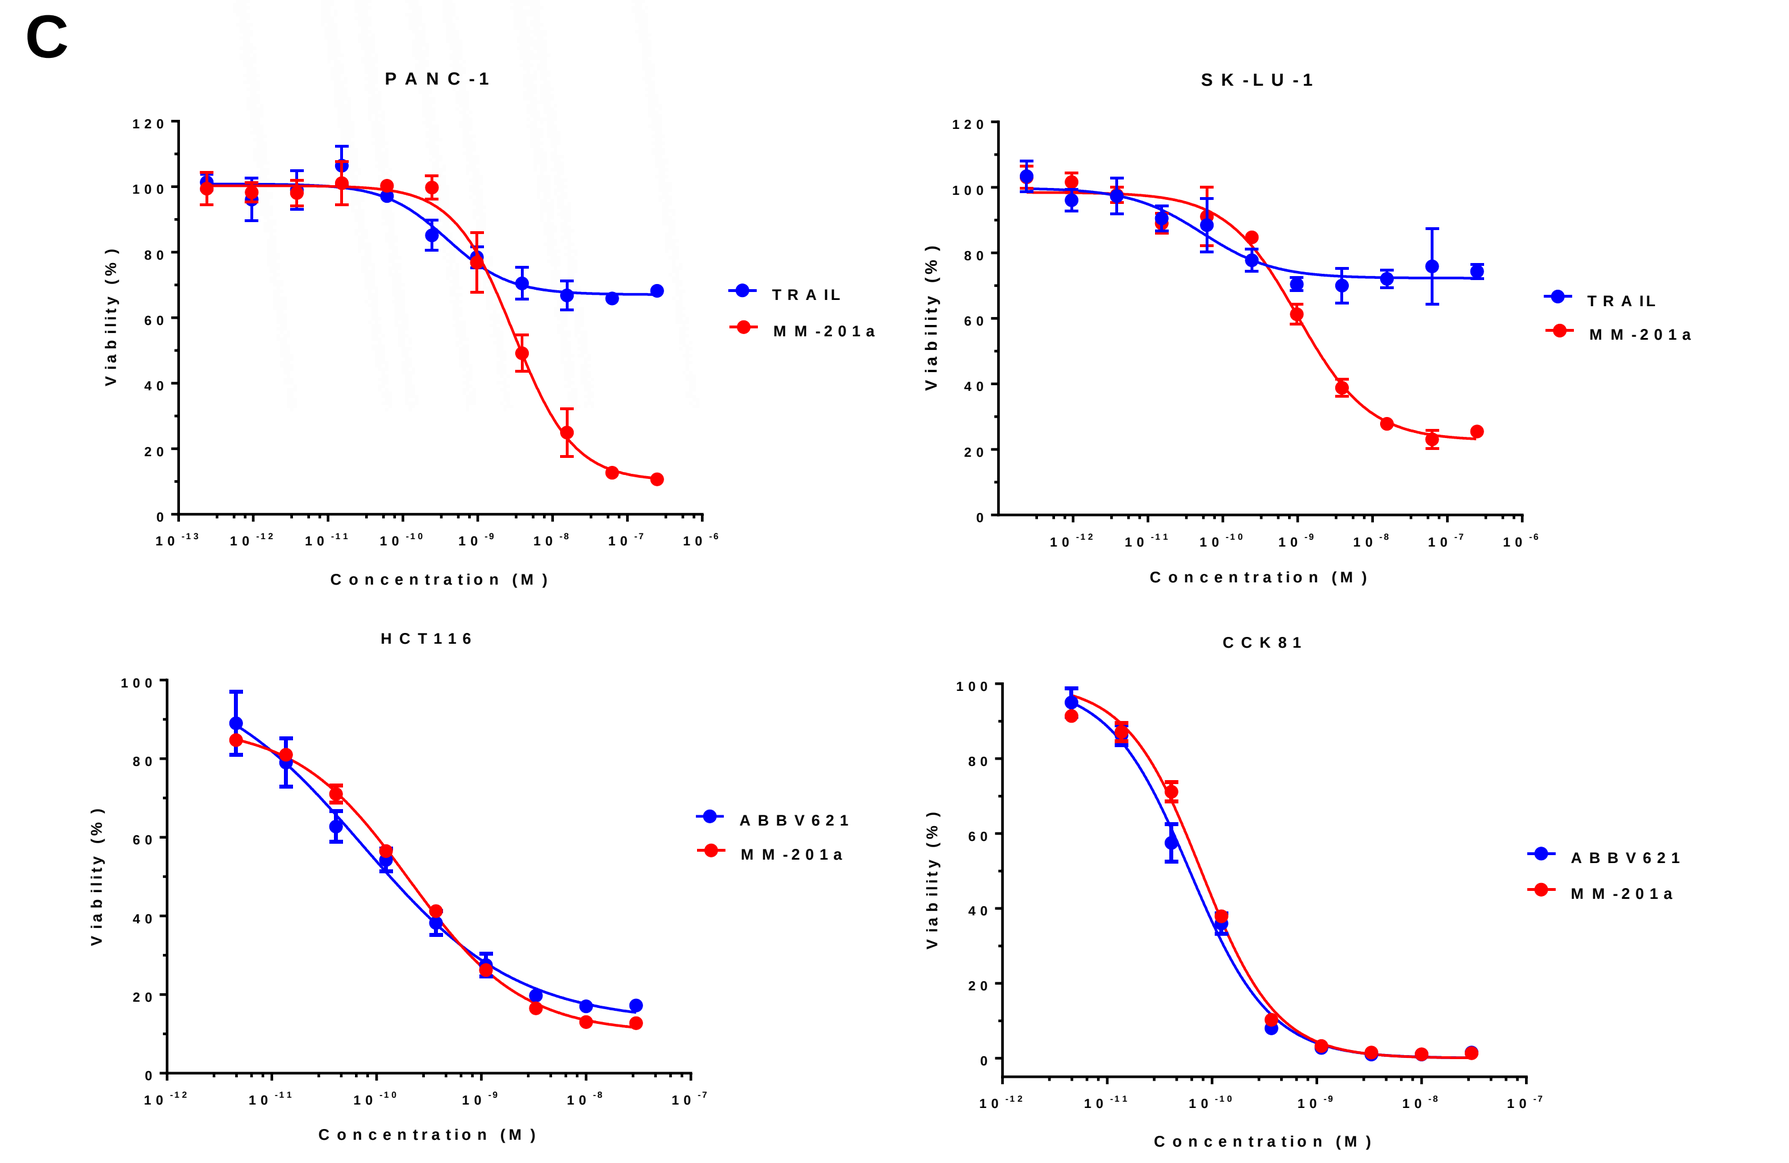
C
PANC-1
SK-LU-1
HCT116
CCK81
TRAIL
MM-201a
ABBV621
Viability (%)
Concentration (M)

Figure 2. Comparison of MM-201a and competitor death receptor agonists (A) HELA cells containing caspase 8 activity reporter were imaged every 3 mins after treatment with MM-201a, TRAIL, anti-DR5 (Apomab and Conatumumab), anti-DR4 (Mapatumumab). (B) COLO205 cells were treated for 1 hr with 30 nM of MM-201a, Apomab, and Mapatumumab prior to staining. (C) Cells were treated with MM-201a and either TRAIL or ABBV621 for 24 hr prior CTG assay.

Computational logic gate model identifies DR4 and cIAP1 as biomarkers for CRC

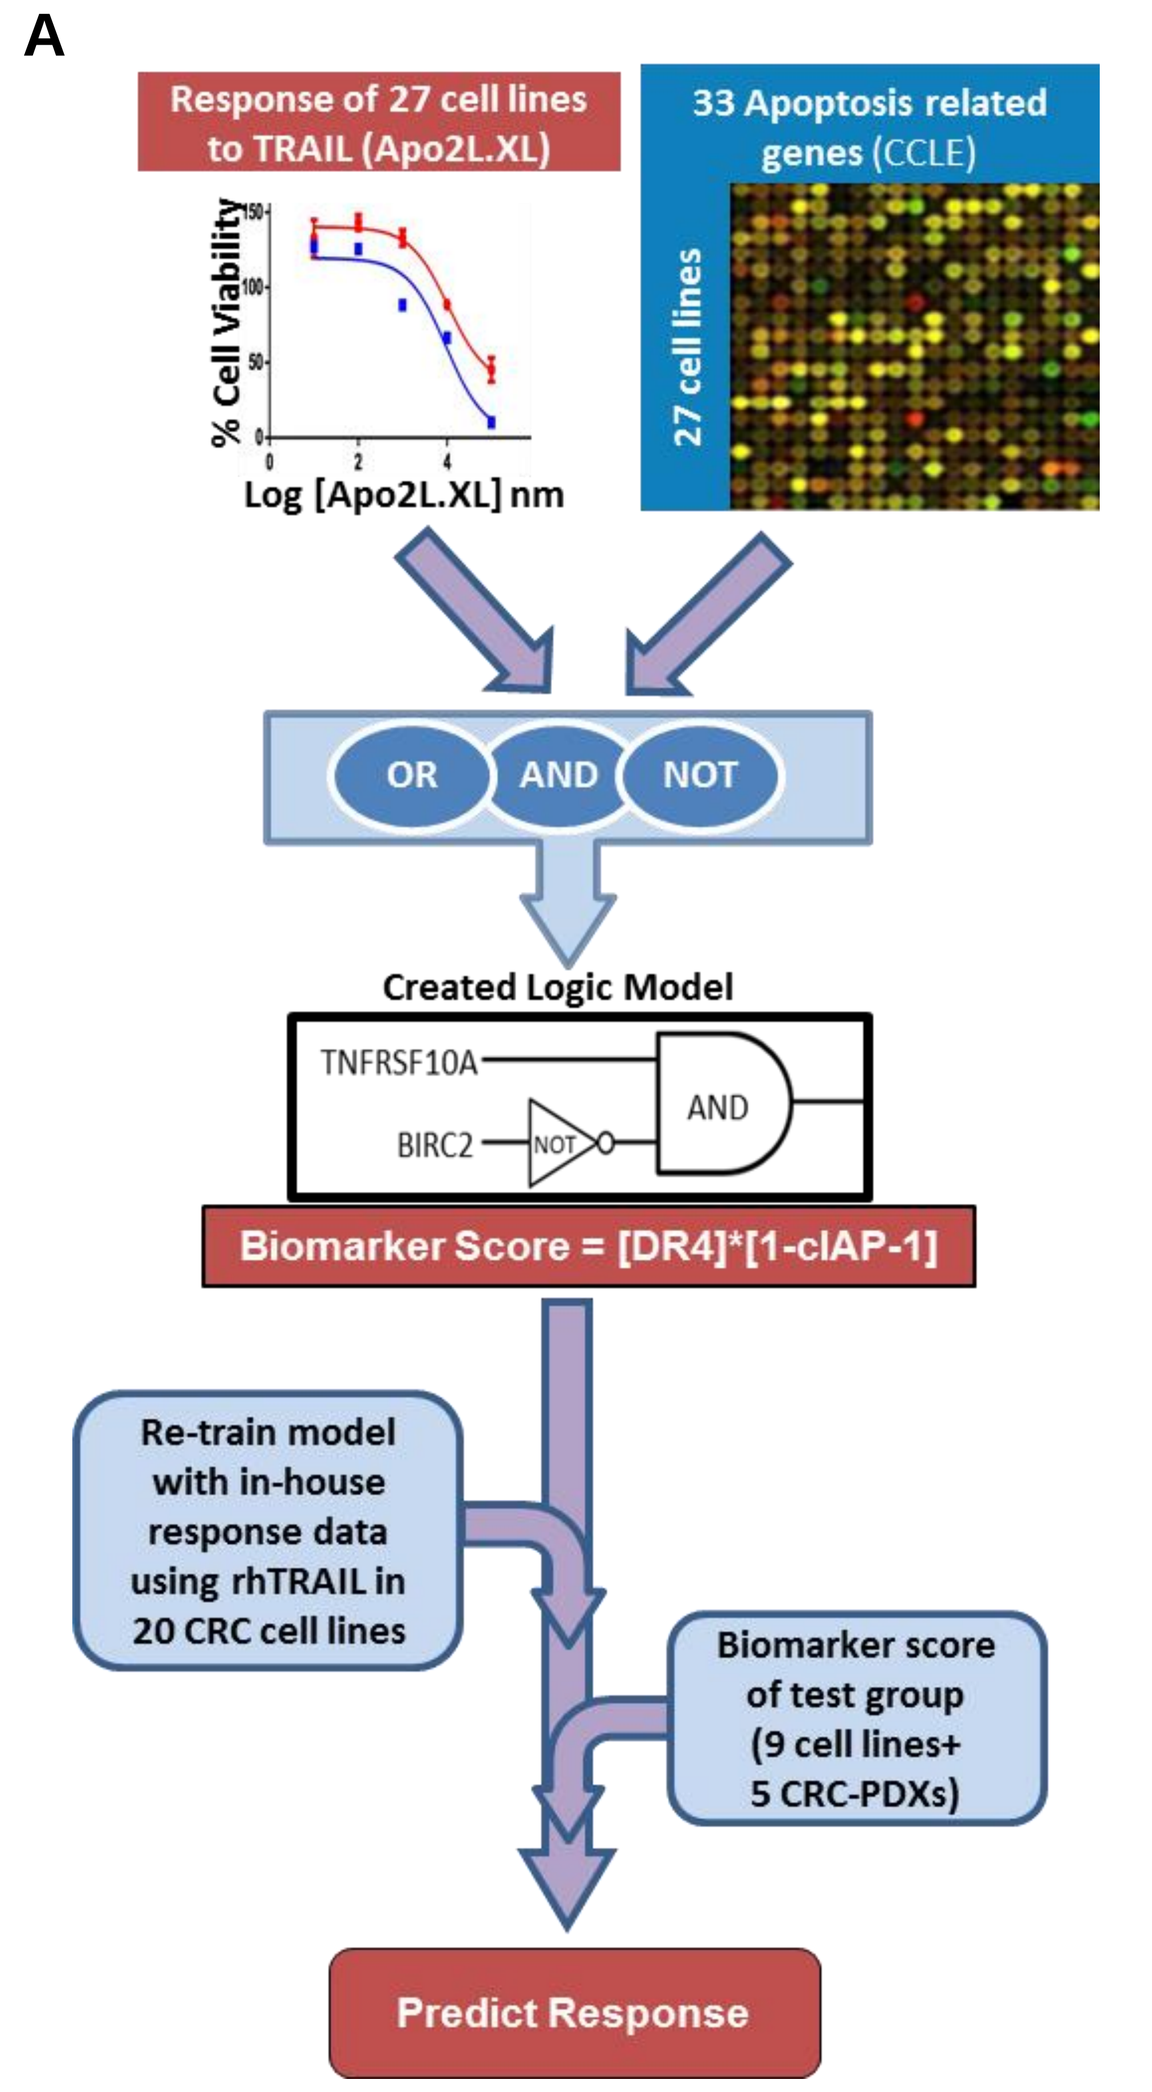
A
Response of 27 cell lines to TRAIL (Apo2L.XL)
% Cell Viability
Log [Apo2L.XL] nm
33 Apoptosis related genes (CCLE)
27 cell lines
OR
AND
NOT
Created Logic Model
TNFRSF10A
BIRC2
NOT
AND
Biomarker Score = [DR4]*[1-cIAP-1]
Re-train model with in-house response data using rhTRAIL in 20 CRC cell lines
Biomarker score of test group (9 cell lines+ 5 CRC-PDXs)
Predict Response

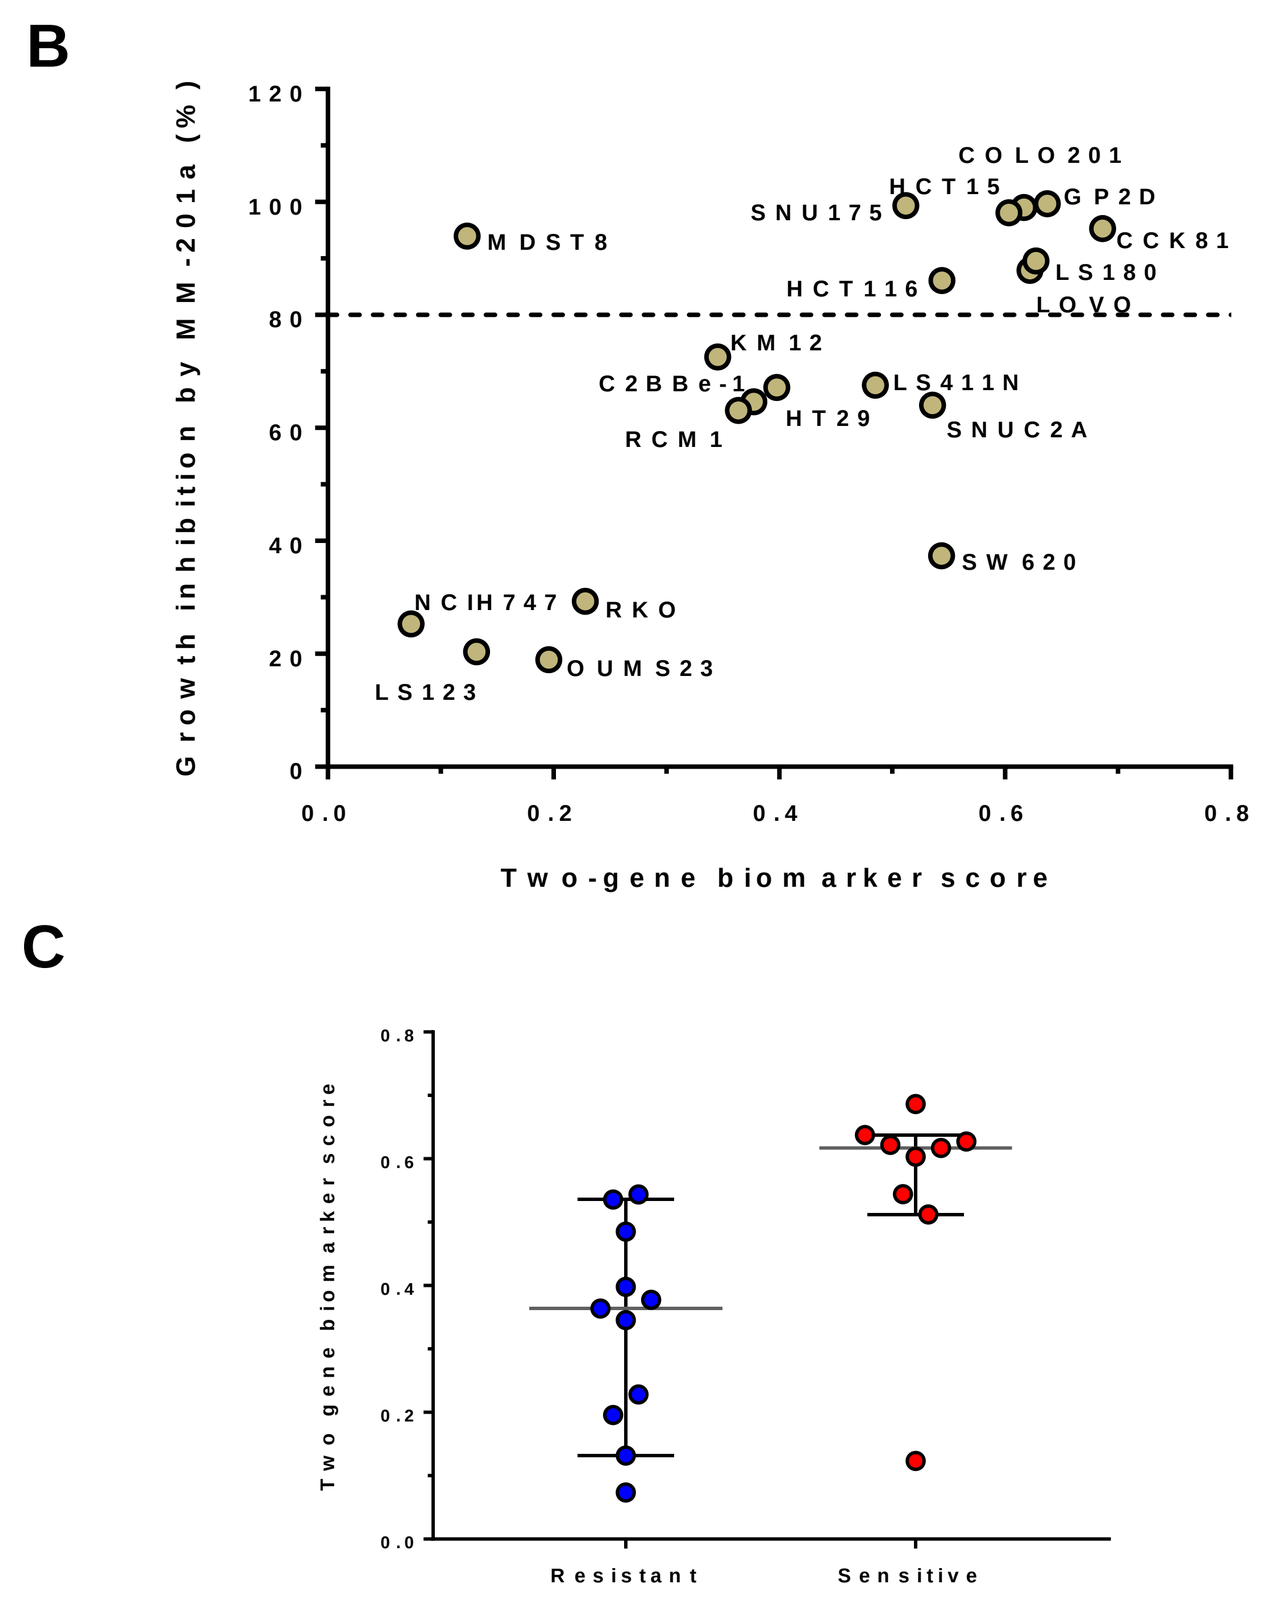
B
Growth inhibition by MM-201a (%)
Two-gene biomarker score
C
Two gene biomarker score
Resistant
Sensitive

Figure 3. Computational logic gate model analysis of public and in-house CRC datasets. (A) Logic model was developed by correlating TRAIL response in CRC cell lines (Nair et al. 2015) to apoptotic gene expression from CCLE. The combination of DR4 and "not" cIAP1 was chosen from 2145 combinations. (B) In-house validation of model: correlation of cell-line response to MM-201a and 2-gene biomarker score. (C) Sensitive cells were classified as having > 80% growth inhibition after 24 hr treatment with 10 nM MM-201a.

Two-gene biomarker predicts MM-201a sensitivity in CRC cell lines

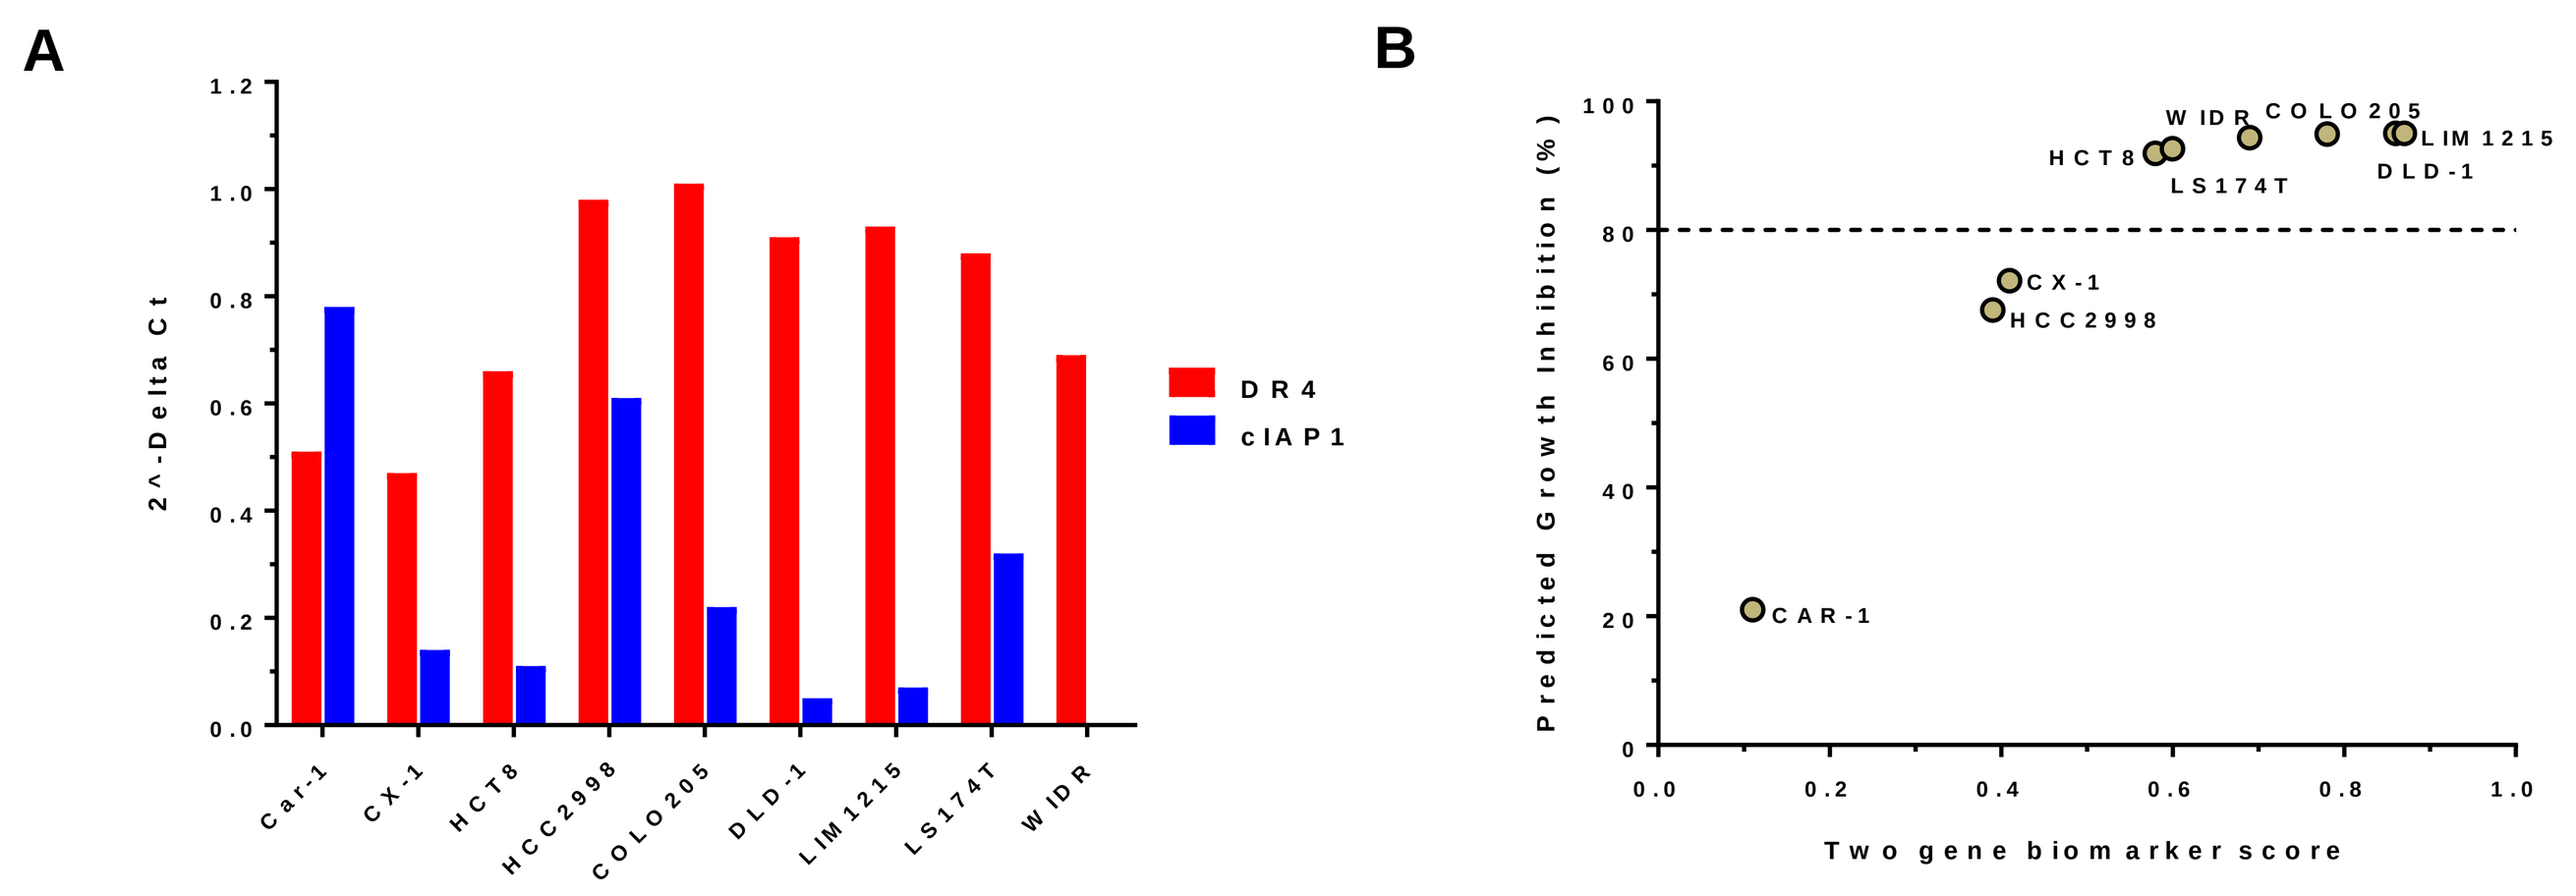
A
2^-Delta Ct
DR4
cIAP1
B
Predicted Growth Inhibition (%)
Two gene biomarker score

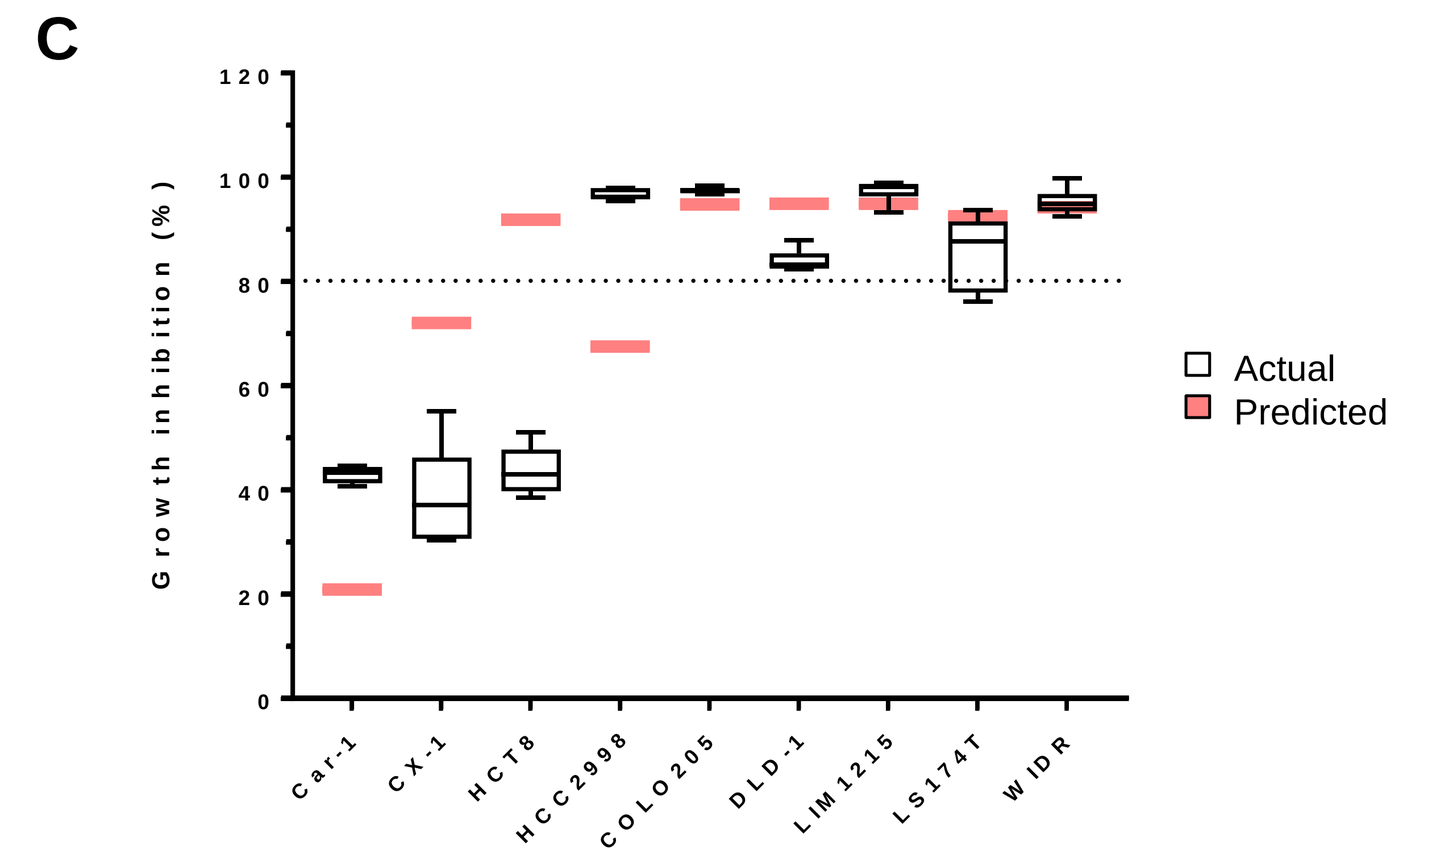
C
Growth inhibition (%)
Actual
Predicted

Figure 4. Two-gene biomarker signature predicts in vitro cellular viability response to MM-201a in CRC cell lines. (A) Relative expression of DR4 and cIAP1 in 9 new CRC cell lines (CCLE data not available) was determined by qPCR. (B) 2-gene biomarker score was calculated and growth inhibition (%) in response to MM-201a was predicted. Sensitive cell lines were classified as having > 80% growth inhibition (C) Actual growth inhibition (n=3, box plots) was measured by CTG after treatment with MM-201a (10 nM) for 24 hrs. The predicted and actual growth inhibition matched in 7/9 cell lines.

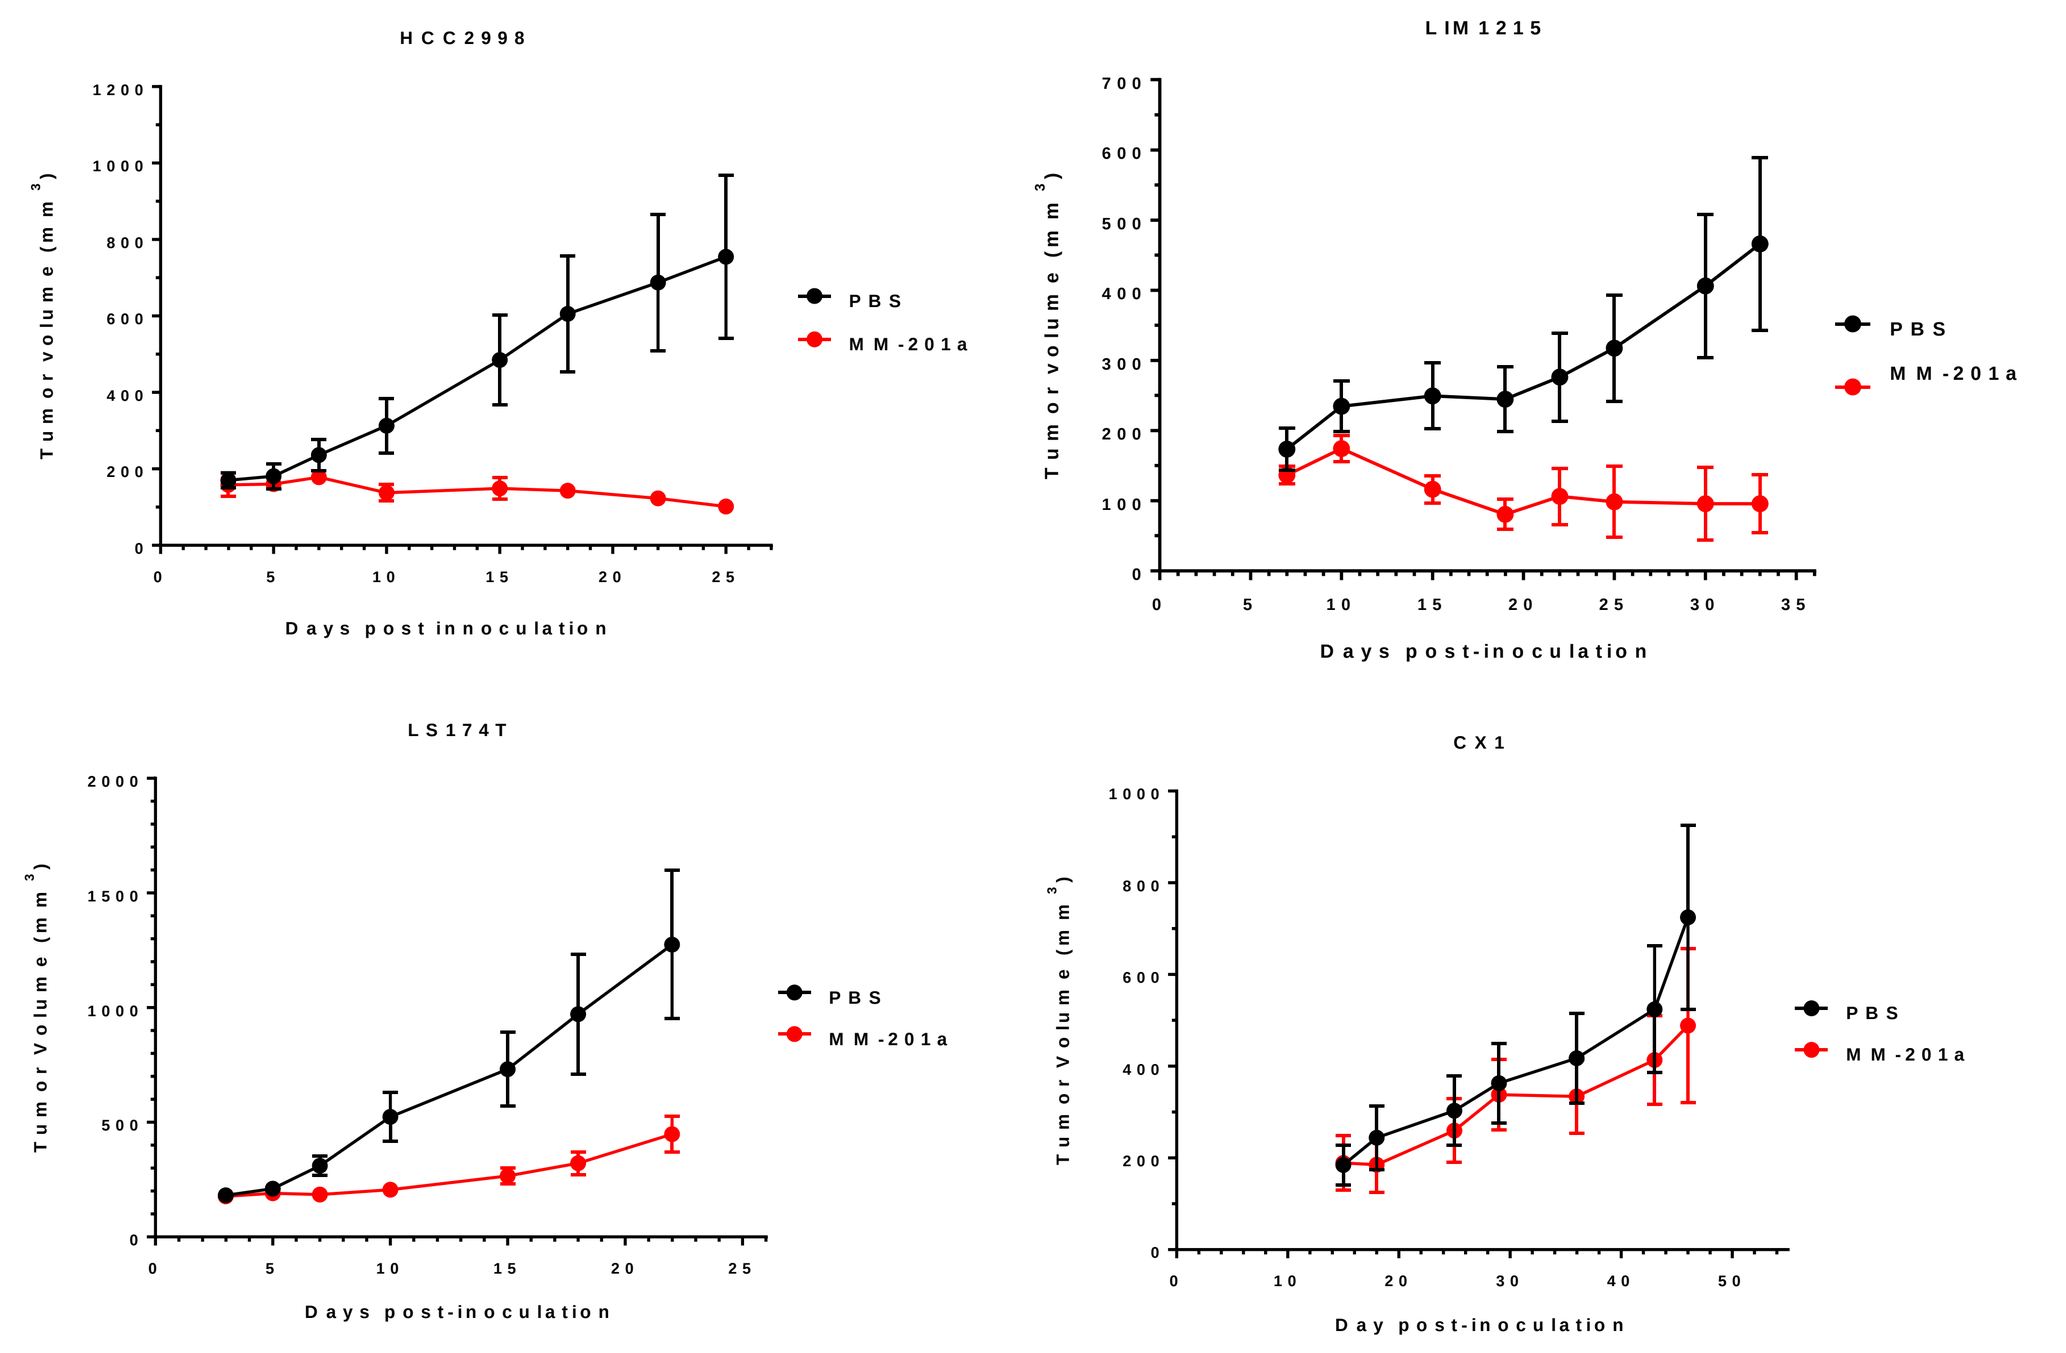
HCC2998
LIM1215
LS174T
CX1
PBS
MM-201a
Tumor volume (mm3)
Days post-inoculation

Figure 5. Two-gene biomarker is predictive of MM-201a activity in CRC xenograft models. Nude mice carrying cell derived tumors were treated with vehicle or T191 (5 mg/kg, IP, 1 x dose). Tumor volumes are expressed as the mean ± SEM and group size was 5-8 mice/arm.

MM-201a synergizes with IAP inhibitors

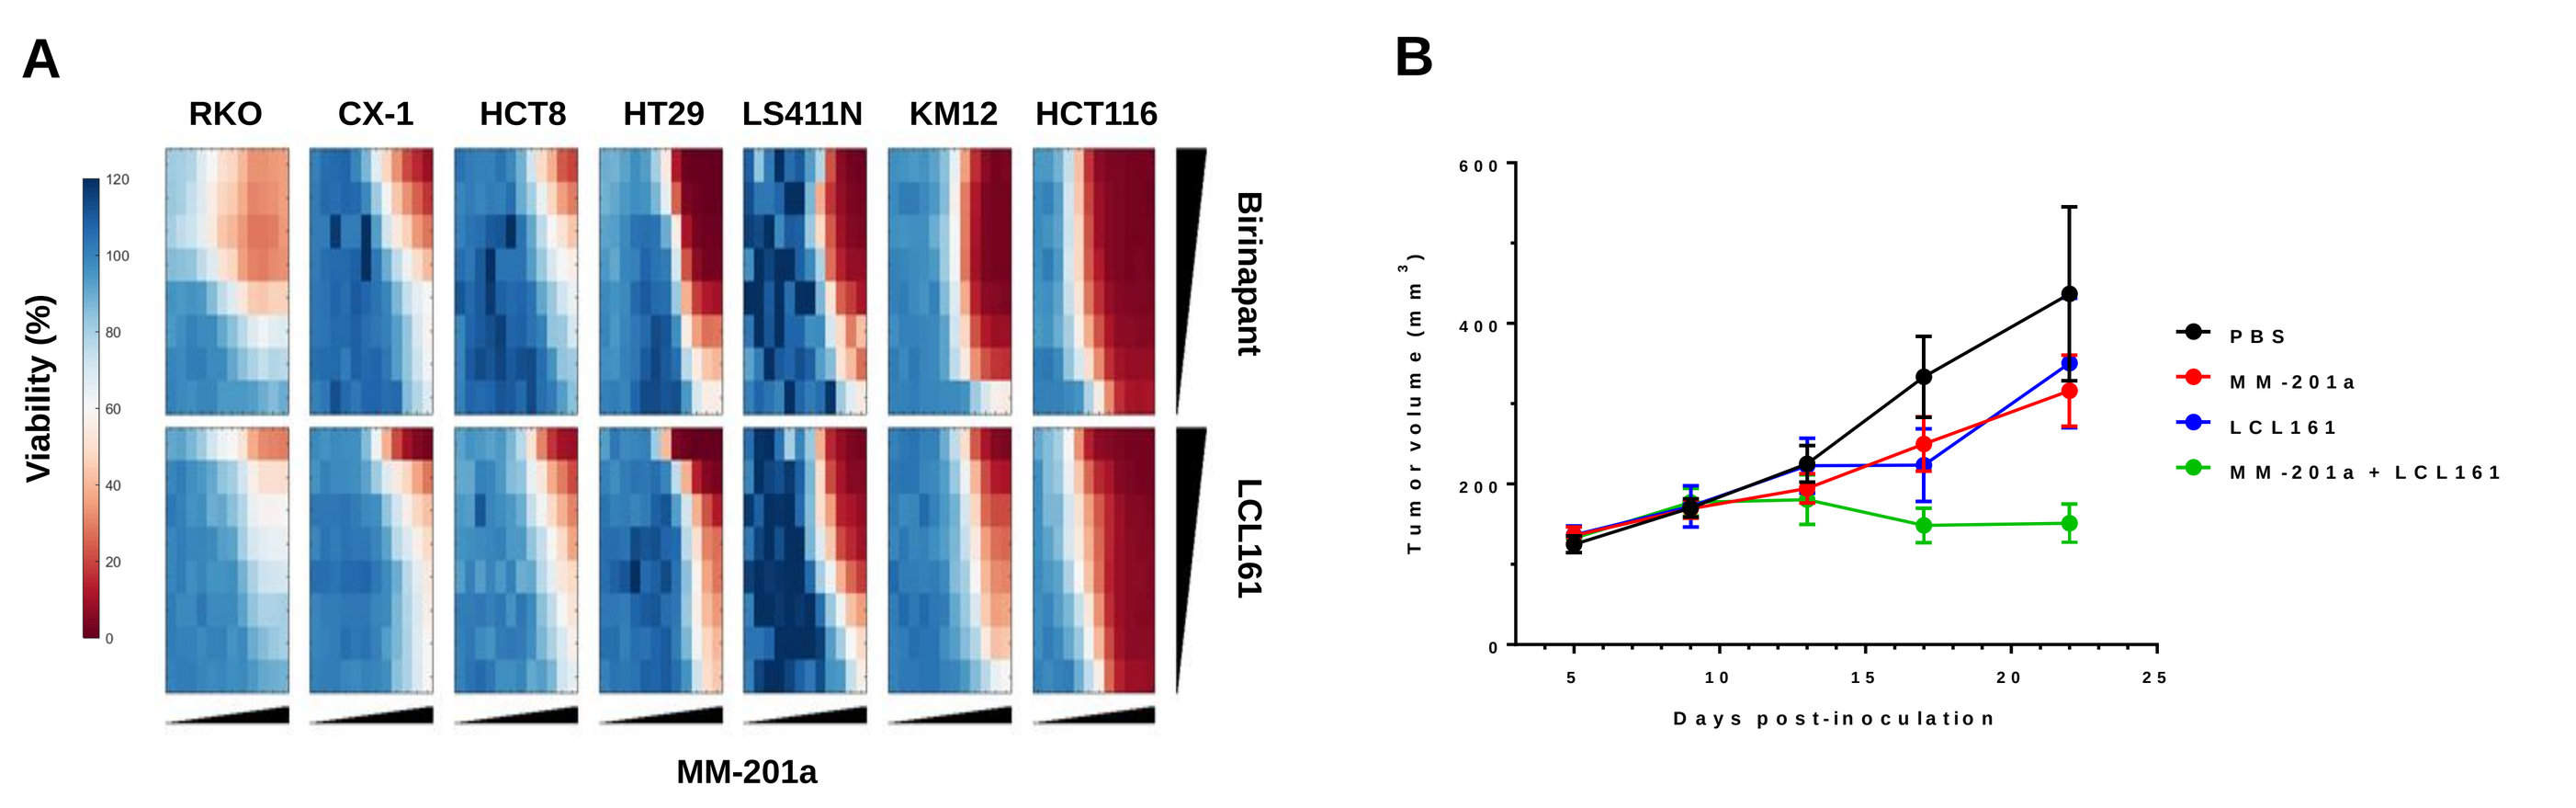
A
RKO
CX-1
HCT8
HT29
LS411N
KM12
HCT116
Viability (%)
Birinapant
LCL161
MM-201a
B
Tumor volume (mm3)
Days post-inoculation
PBS
MM-201a
LCL161
MM-201a + LCL161

Figure 6. MM-201a synergized with IAP inhibitors in CRC cell lines. (A) Heatmap representation of cell viability after 24 hr treatment with MM-201a (1:3 dilution starting from 30 nM) and IAP (Birinapant and LCL161) (1:4 dilution starting from 6 µM. (B) RKO xenograft model. Mice were dosed with T191 (5 mg/kg, IP, once/week), LCL161 (30 mg/kg, PO, twice/week) or the combination. Data are expressed as the mean ± SEM (n=8).

Two-gene biomarker predicts MM-201a sensitivity in CRC PDX models

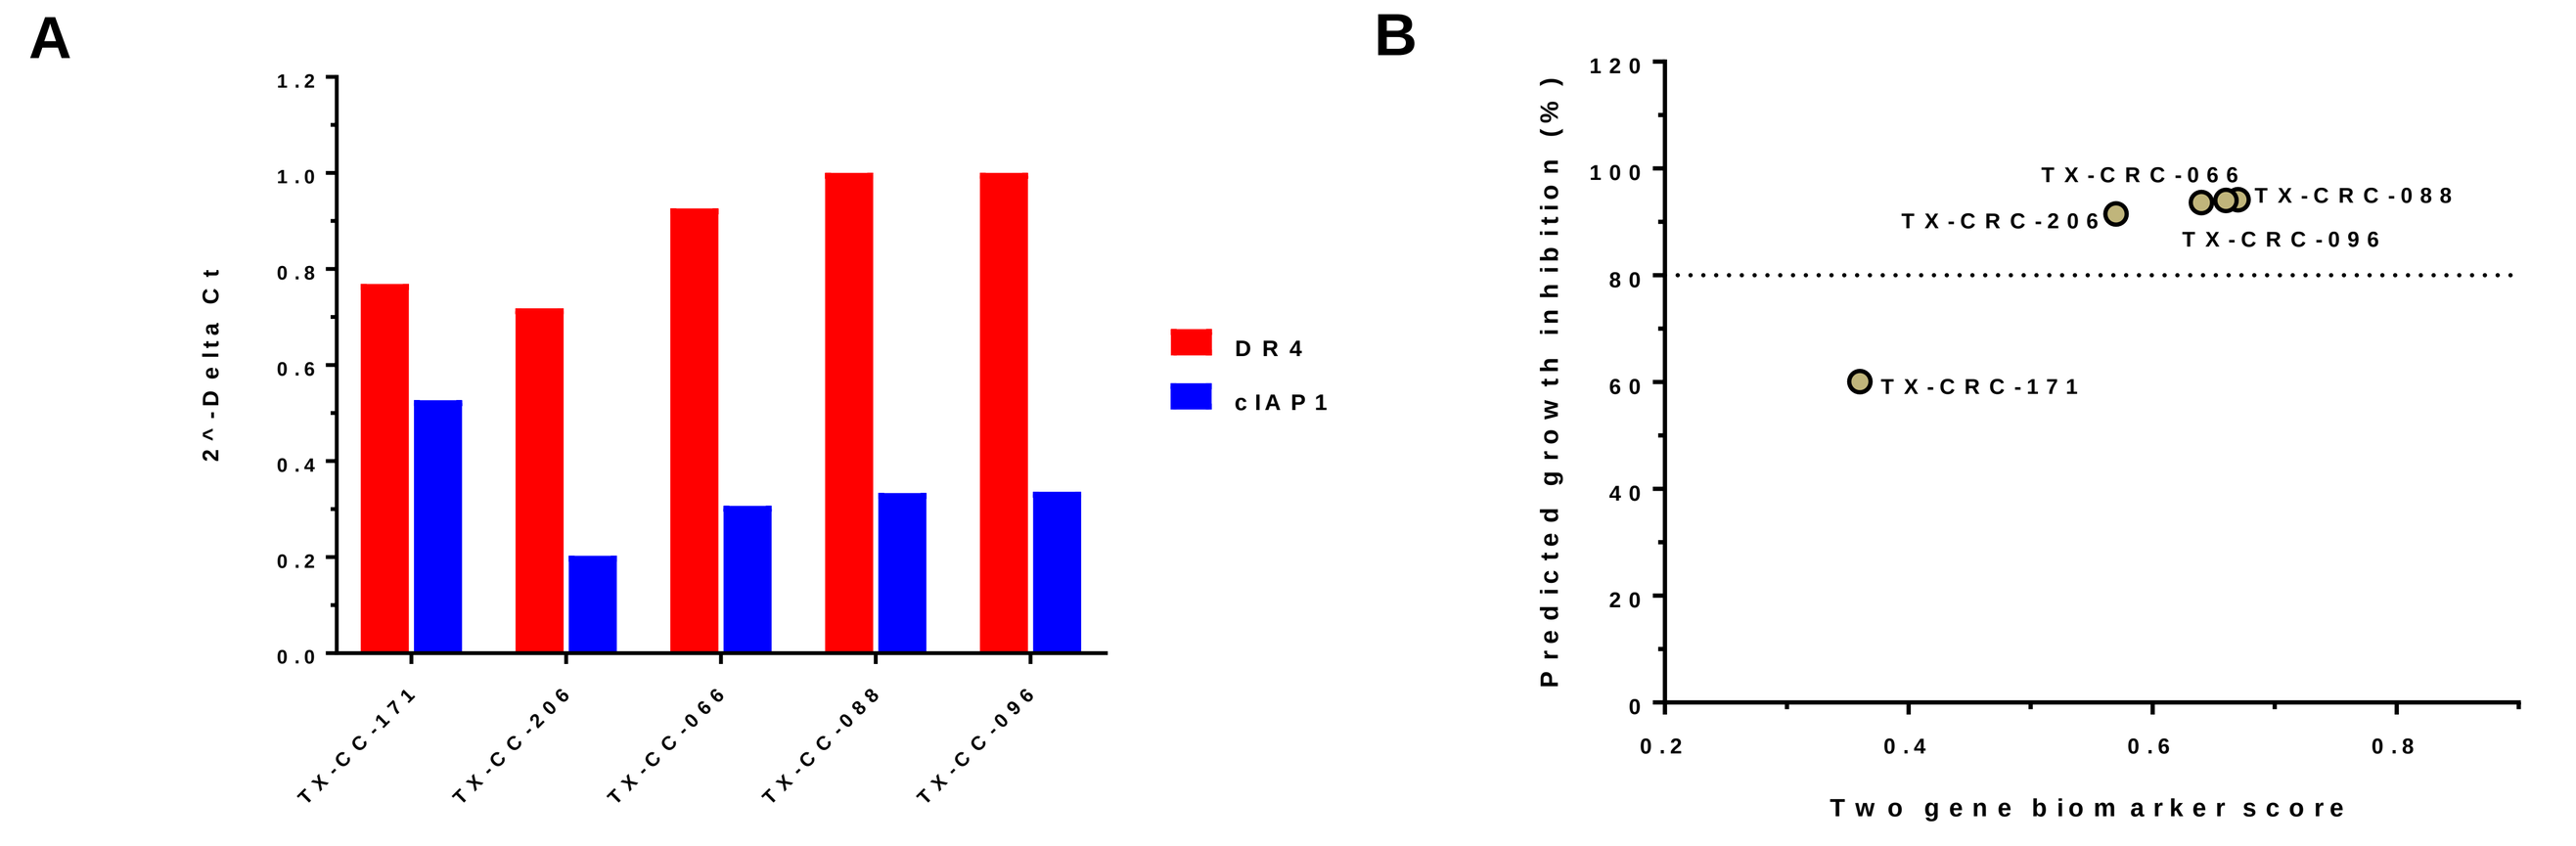
A
2^-Delta Ct
DR4
cIAP1
B
Predicted growth inhibition (%)
Two gene biomarker score

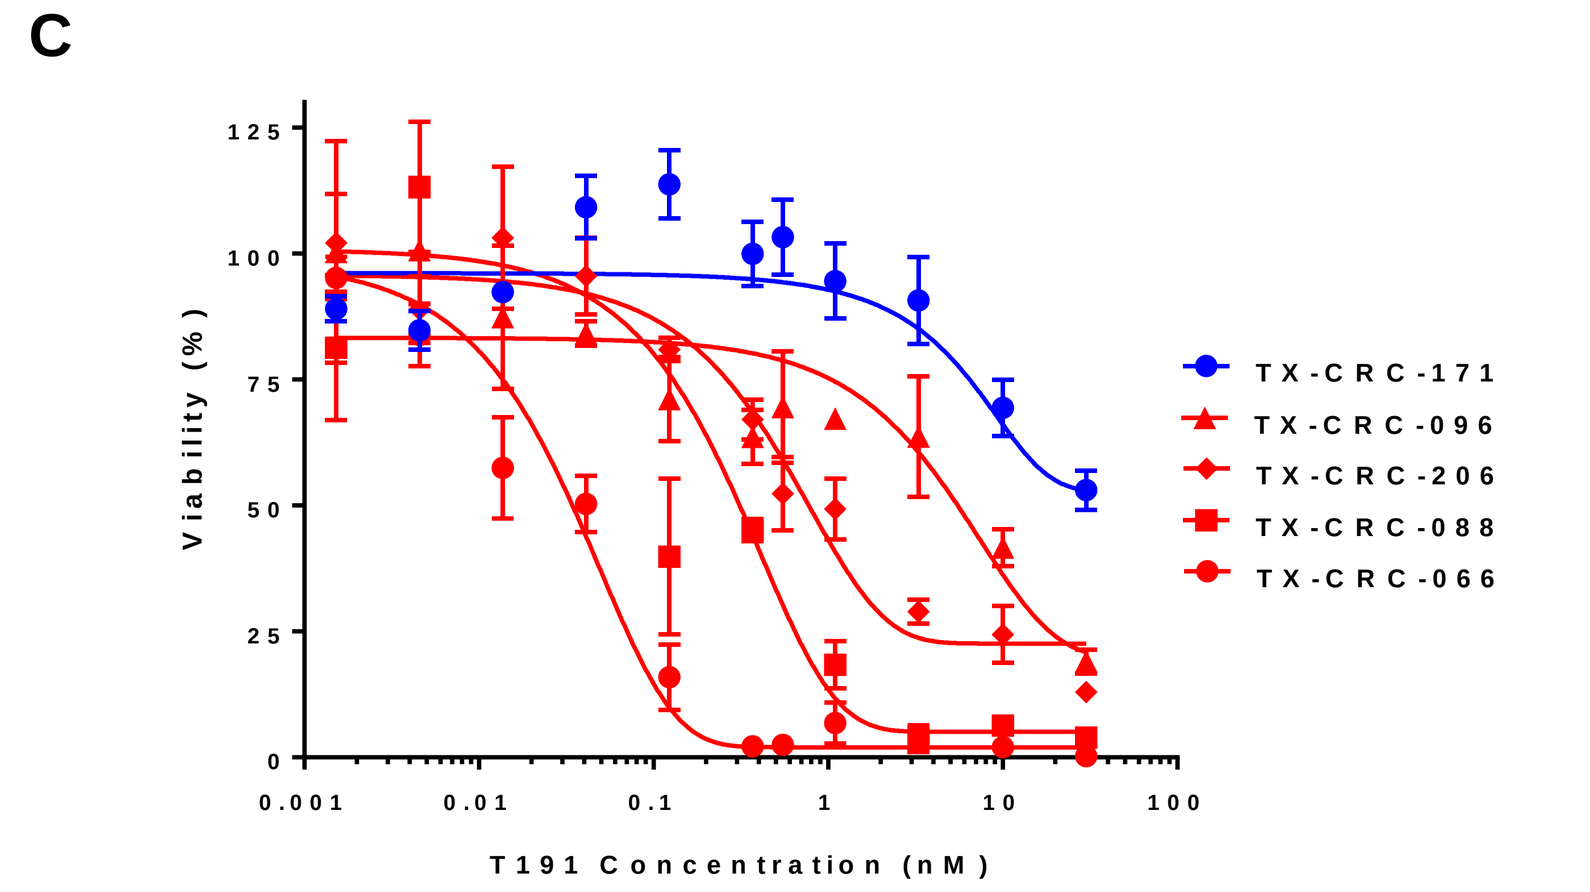
C
Viability (%)
T191 Concentration (nM)
TX-CRC-171
TX-CRC-096
TX-CRC-206
TX-CRC-088
TX-CRC-066

Figure 7. Two-gene biomarker signature predicts in vitro cellular viability response to MM-201a in CRC PDX organoids. (A) Relative expression of DR4 and cIAP1 in CRC PDX models were determined by qPCR. (B) 2-gene biomarker score was calculated and growth inhibition (%) in response to MM-201a was predicted. Sensitive models were classified as having > 80%. growth inhibition. (C) Actual growth inhibition (n=3, box plots) was measured by CTG after treatment with MM-201a for 24 hrs. The predictions match experimental growth inhibition in 5/5 PDX models.

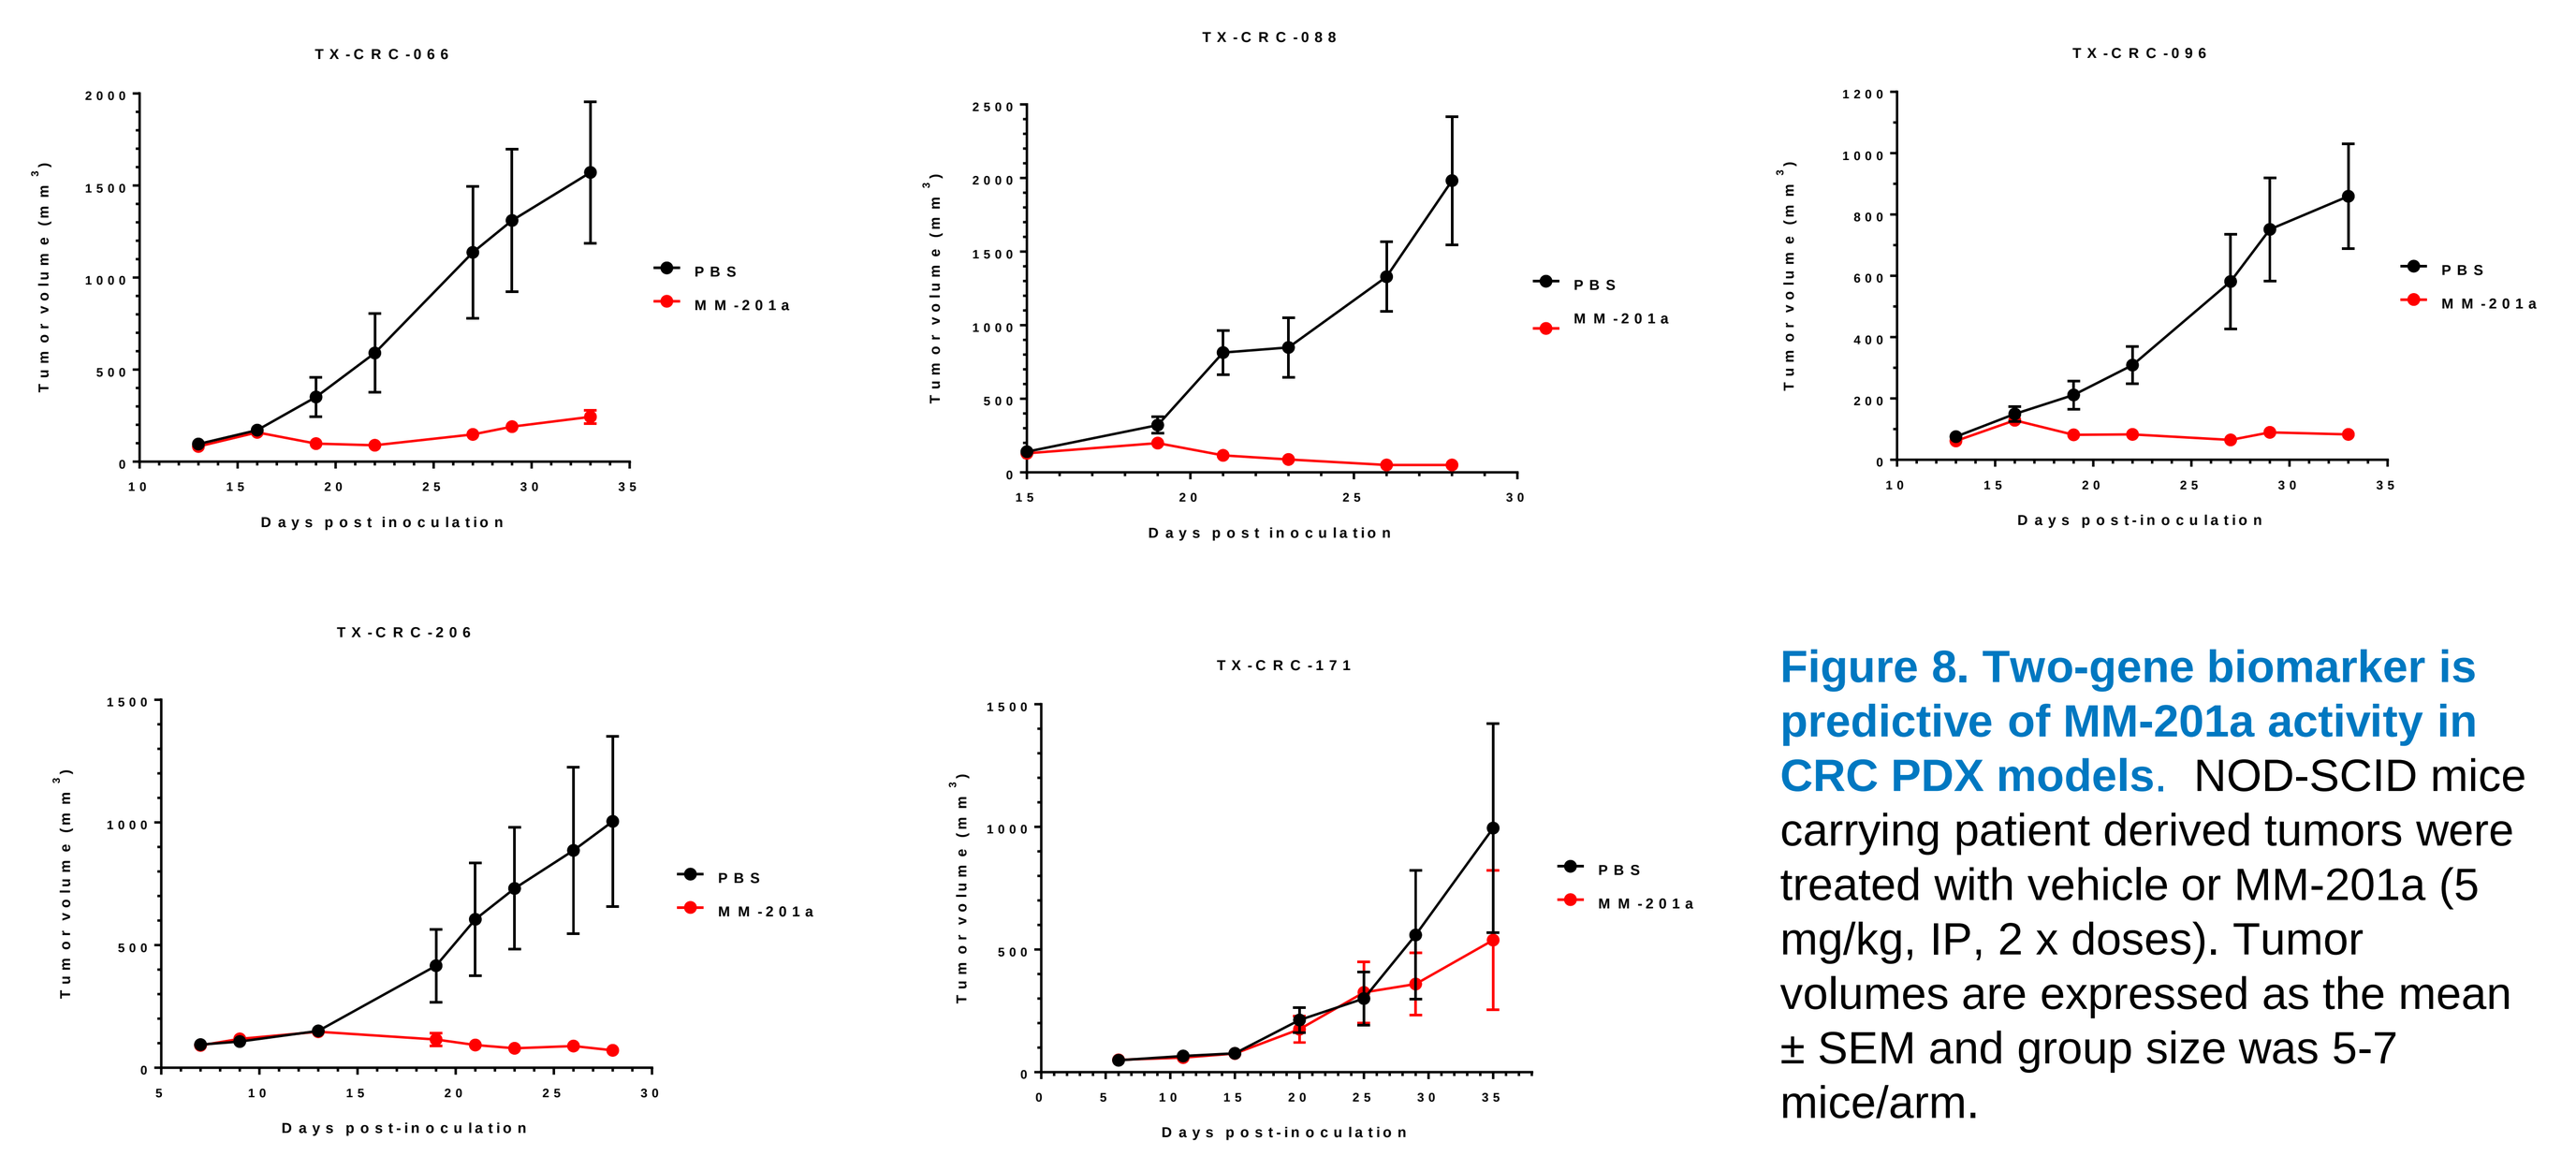
TX-CRC-066
TX-CRC-088
TX-CRC-096
TX-CRC-206
TX-CRC-171
PBS
MM-201a
Tumor volume (mm3)
Days post-inoculation

Figure 8. Two-gene biomarker is predictive of MM-201a activity in CRC PDX models. NOD-SCID mice carrying patient derived tumors were treated with vehicle or MM-201a (5 mg/kg, IP, 2 x doses). Tumor volumes are expressed as the mean ± SEM and group size was 5-7 mice/arm.

Analysis of two-gene biomarker in TCGA

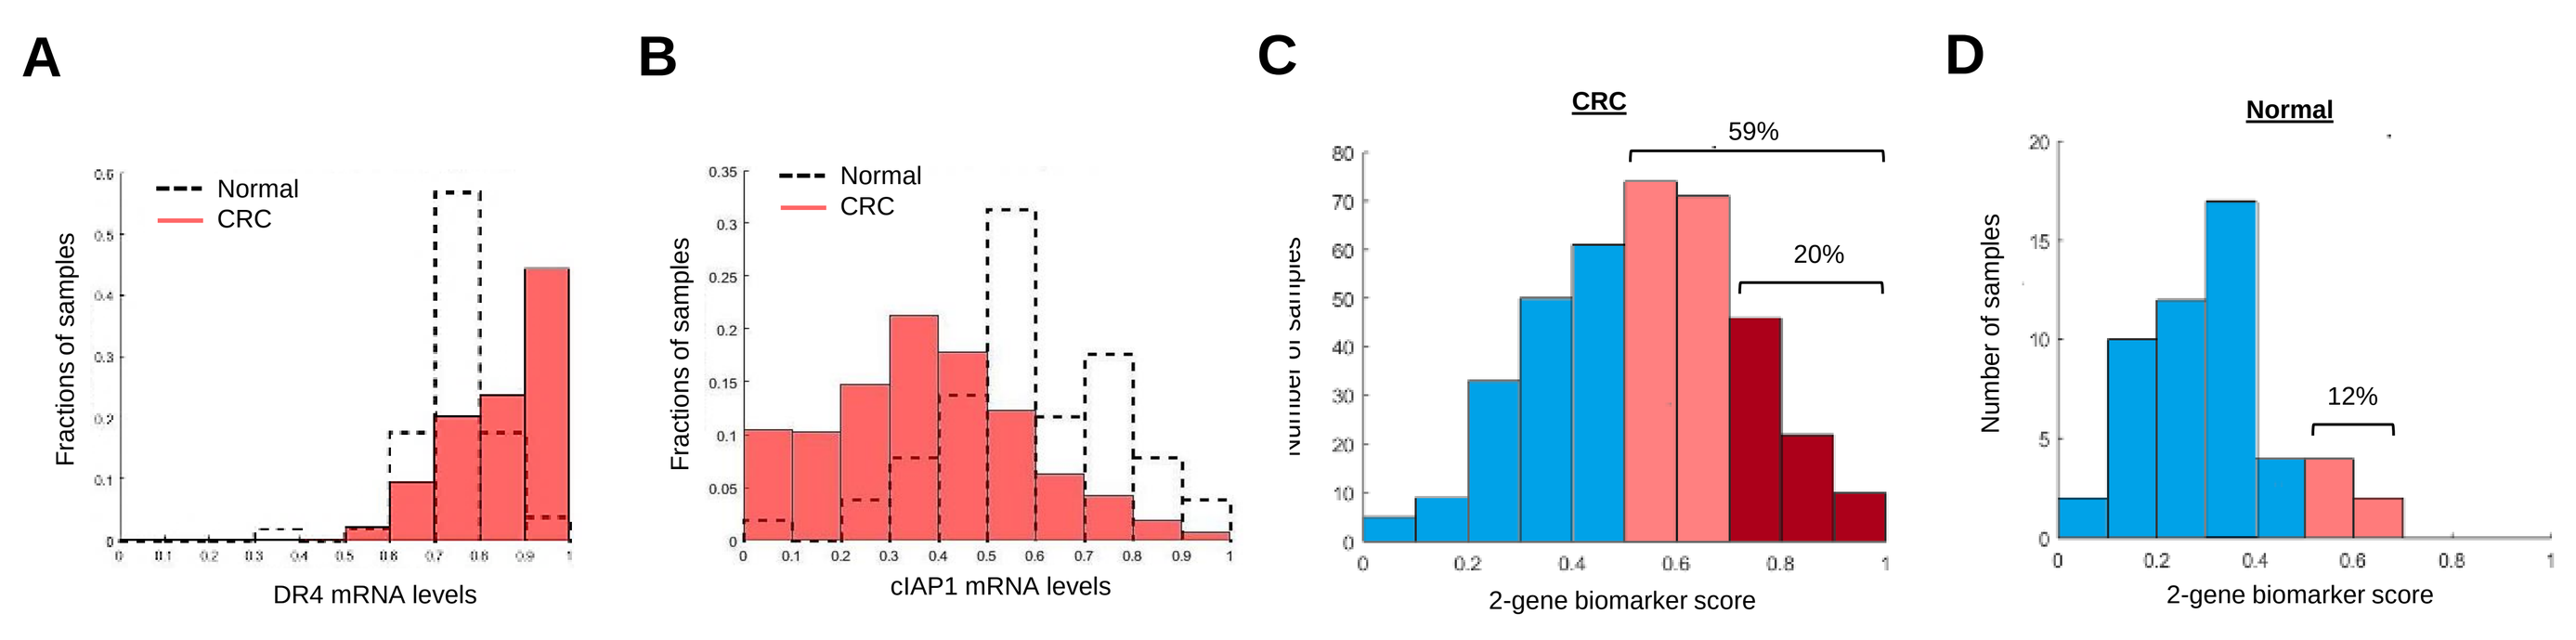
A
Normal
CRC
Fractions of samples
DR4 mRNA levels
B
cIAP1 mRNA levels
C
CRC
59%
20%
2-gene biomarker score
D
Normal
12%
Number of samples

Figure 9. Two gene biomarker score predicts TRAIL therapy would be beneficial to large patient population in CRC. DR4 and cIAP1 mRNA levels from CRC patient samples in the TCGA were analyzed. DR4 and cIAP1 expression was normalized across all samples in all indications. (A) Comparison of DR4 mRNA levels in CRC tumors and normal samples. (B) Comparison of cIAP1 mRNA levels in CRC tumors and normal samples. (C) In CRC, 59% of samples have a score > 0.5 (75% reduction in cell viability) and 20% of samples have a score > 0.7 (90% reduction in cell viability). (D) In normal samples, 12% have a score > 0.5, and none have score > 0.7.

Nair et al. Proc Natl Acad Sci U S A. 2015 May 5;112(18):5679-84
Kelley et al. J Biol Chem. 2005 Jan 21;280(3):2205-12

© 2019 Merrimack Pharmaceuticals, Inc. All rights reserved.