Abstract: ENA-0373

MM-401: A novel antibody that targets TNFR2, a key regulator of the tumor immunosuppressive microenvironment

A. Raue, R. Fulton, J. Sampson, A. Camblin, J. Richards, Y. Jiao, L. Xu, C. Wong, A. Koshkaryev, V. Paragas, L. Luus, S. Ghassemifar, M. Razlog, A.S. Dugast, F. Depis, M. Muda, E. Tam, D.C. Drummond

Merrimack Pharmaceuticals, Inc., Cambridge MA, USA

Summary

Background: Despite the dramatic anti-tumor responses observed for immune checkpoint inhibition in subsets of patients, there remains an unmet medical need for the larger patient population. Resistance to checkpoint inhibitors include multiple immunosuppressive mechanisms that are present in the tumor microenvironment, many of which are linked to regulatory T cells (Tregs). Here, we have identified TNF receptor 2 (TNFR2) as a key regulator of the immunosuppressive microenvironment and have observed significant preclinical anti-tumor responses using a monoclonal antibody targeting that receptor.

Materials and methods: Human and mouse tissues were profiled for TNFR2 expression in the tumor versus periphery. Antibodies against murine TNFR2 were generated by screening a human antibody library and by rabbit immunization. Antibodies were characterized for affinity, ability to block TNF and for developability. A selected number of antibodies were expressed as murine IgG2a and evaluated for activity in multiple syngeneic mouse tumor models. The mechanism of action of the most potent murine surrogate antibody (Y9) was investigated further both *in vitro* and *in vivo*.

Results: TNFR2 was highly expressed in both human and murine intratumoral Tregs compared to other T cell subpopulations. Notably, TNFR2 expression in the periphery was low for all T cell populations. Our murine surrogate antibody Y9 provided co-stimulatory signaling that led to proliferation and activation of T cells *in vitro*. *In vivo,* Y9 showed robust anti-tumor activity at a single injection of 300 µg in multiple syngeneic mouse tumor models, including anti-PD-1 resistant models. While our surrogate antibody led to significant tumor regression, it did not lead to significant depletion of cell types, such as Tregs, as observed frequently for antibodies targeting other T cell receptors. Consistently, our anti-TNFR2 antibody compared favorably in a preclinical toxicity model to other antibodies targeting other T cell receptors. After treatment with Y9 we observed a broad reversal of immunosuppression in the tumor. Activity correlated with the downregulation of several suppressive markers, such as PD-1, GARP/LAP, and CTLA-4, and induced long-term anti-tumor memory. The mechanism of action required CD8+ T cell and NK cell responses and led to long-term memory in re-challenge experiments.

Why Target TNFR2?

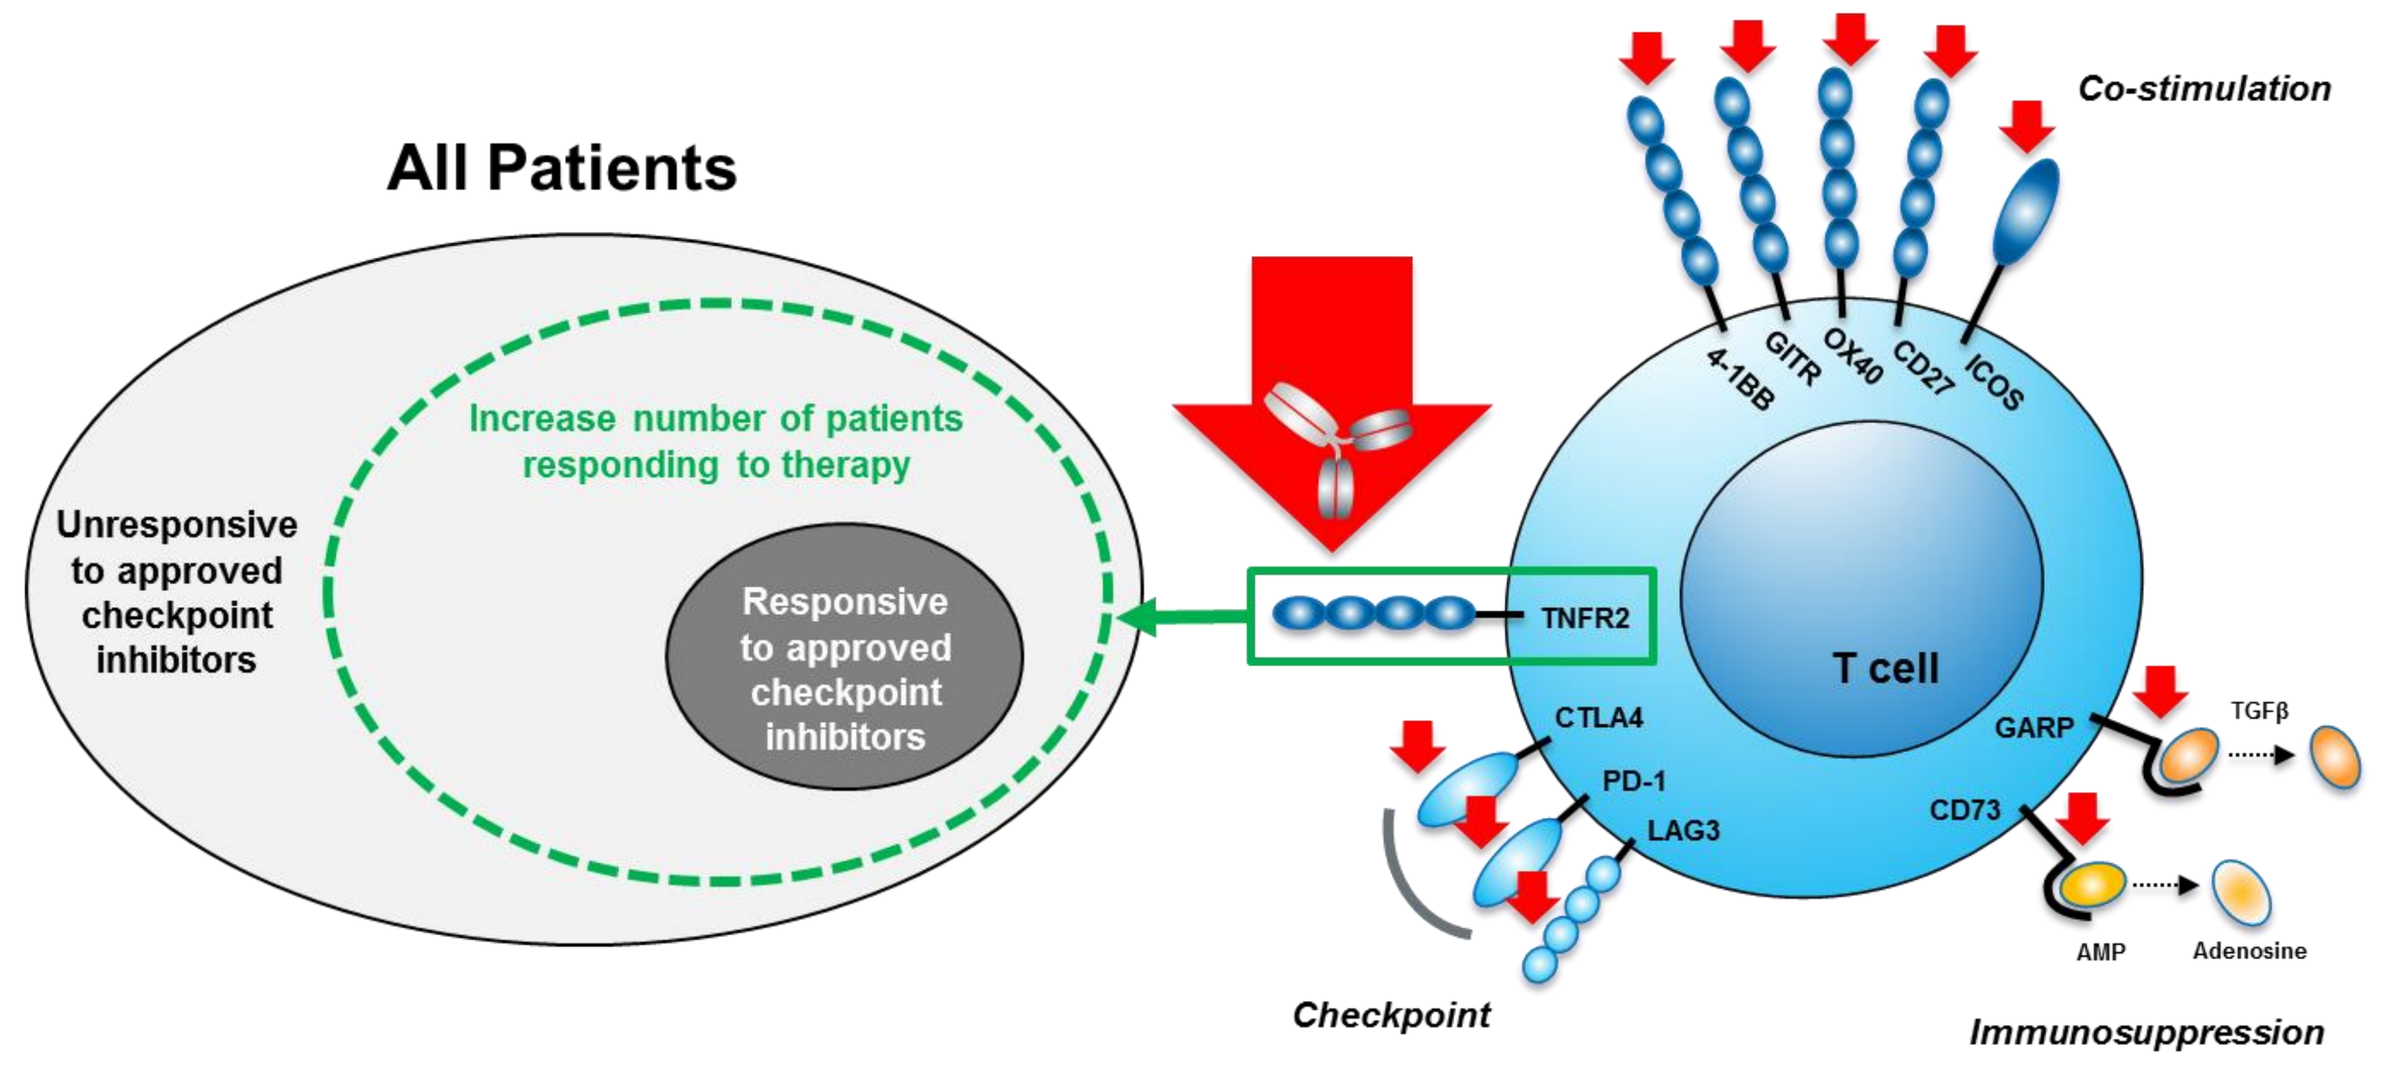
All Patients
Unresponsive to approved checkpoint inhibitors
Increase number of patients responding to therapy
Responsive to approved checkpoint inhibitors
Co-stimulation
4-1BB
GITR
OX40
CD27
ICOS
TNFR2
T cell
CTLA4
PD-1
LAG3
GARP
CD73
TGFβ
AMP
Adenosine
Checkpoint
Immunosuppression

- Unmet need remains for majority of patients despite approved checkpoint inhibitors
- Multiple parallel T cell-linked immunosuppressive mechanisms facilitate tumor evasion
- Anti-TNFR2 mAb reverses immunosuppression in mouse models, enabling robust anti-tumor response

TNFR2 Immune Profile in Human Tumors

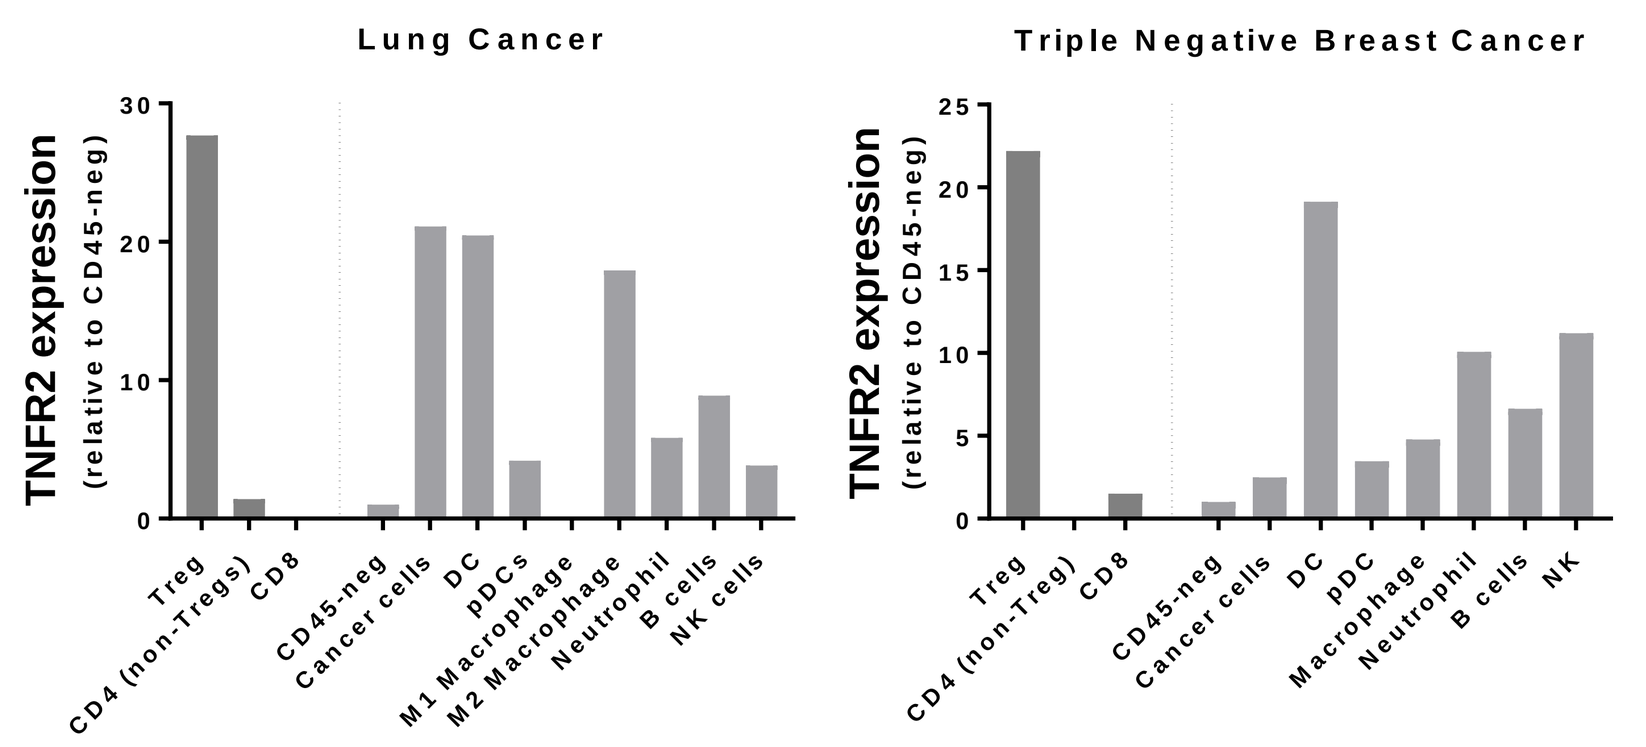
Lung Cancer
Triple Negative Breast Cancer
TNFR2 expression (relative to CD45-neg)
Treg
CD4 (non-Tregs)
CD8
CD45-neg
Cancer cells
DC
pDCs
M1 Macrophage
M2 Macrophage
Neutrophil
B cells
NK cells

- TNFR2 is highly expressed in human intratumoral Tregs vs. other T cells
- Data from human tumors consistent with data from murine syngeneic models

Characterization of Murine Surrogate Anti-TNFR2 Antibody (Clone Y9)

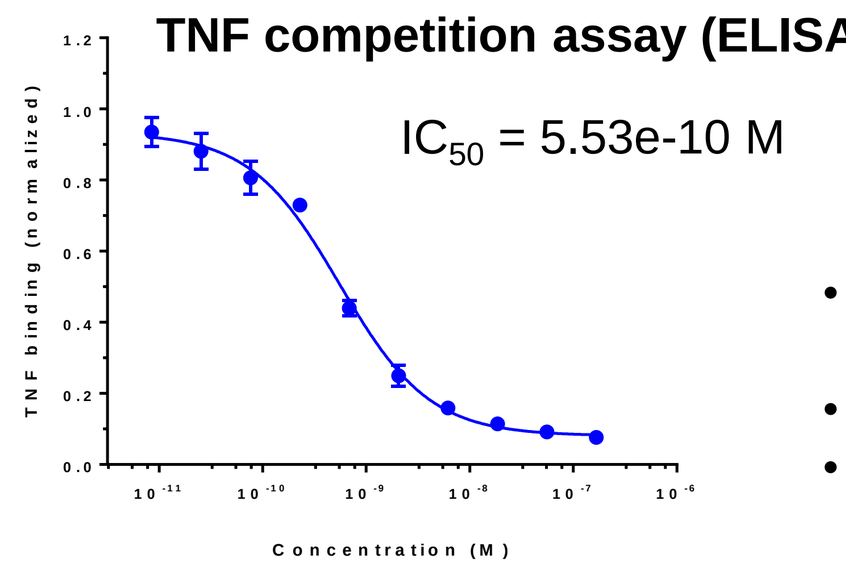
TNF competition assay (ELISA)
IC50 = 5.53e-10 M
TNF binding (normalized)
Concentration (M)

- Y9 (mIgG2a) binds murine TNFR2 with an affinity of 0.5 nM as measured by biolayer interferometry (BLI)
- Y9 is a potent murine anti-TNFR2 antibody and binds well defined critical epitope
- Y9 is a TNF competitor, but TNF competition is not required for activity

Anti-TNFR2 Antibody is Active in PD-1/PD-L1 Resistant and Sensitive Syngeneic Mouse Tumor Models

PD-1 resistant: MBT-2 (Bladder Cancer) **PD-1 sensitive:** Sa1/N (Sarcoma)

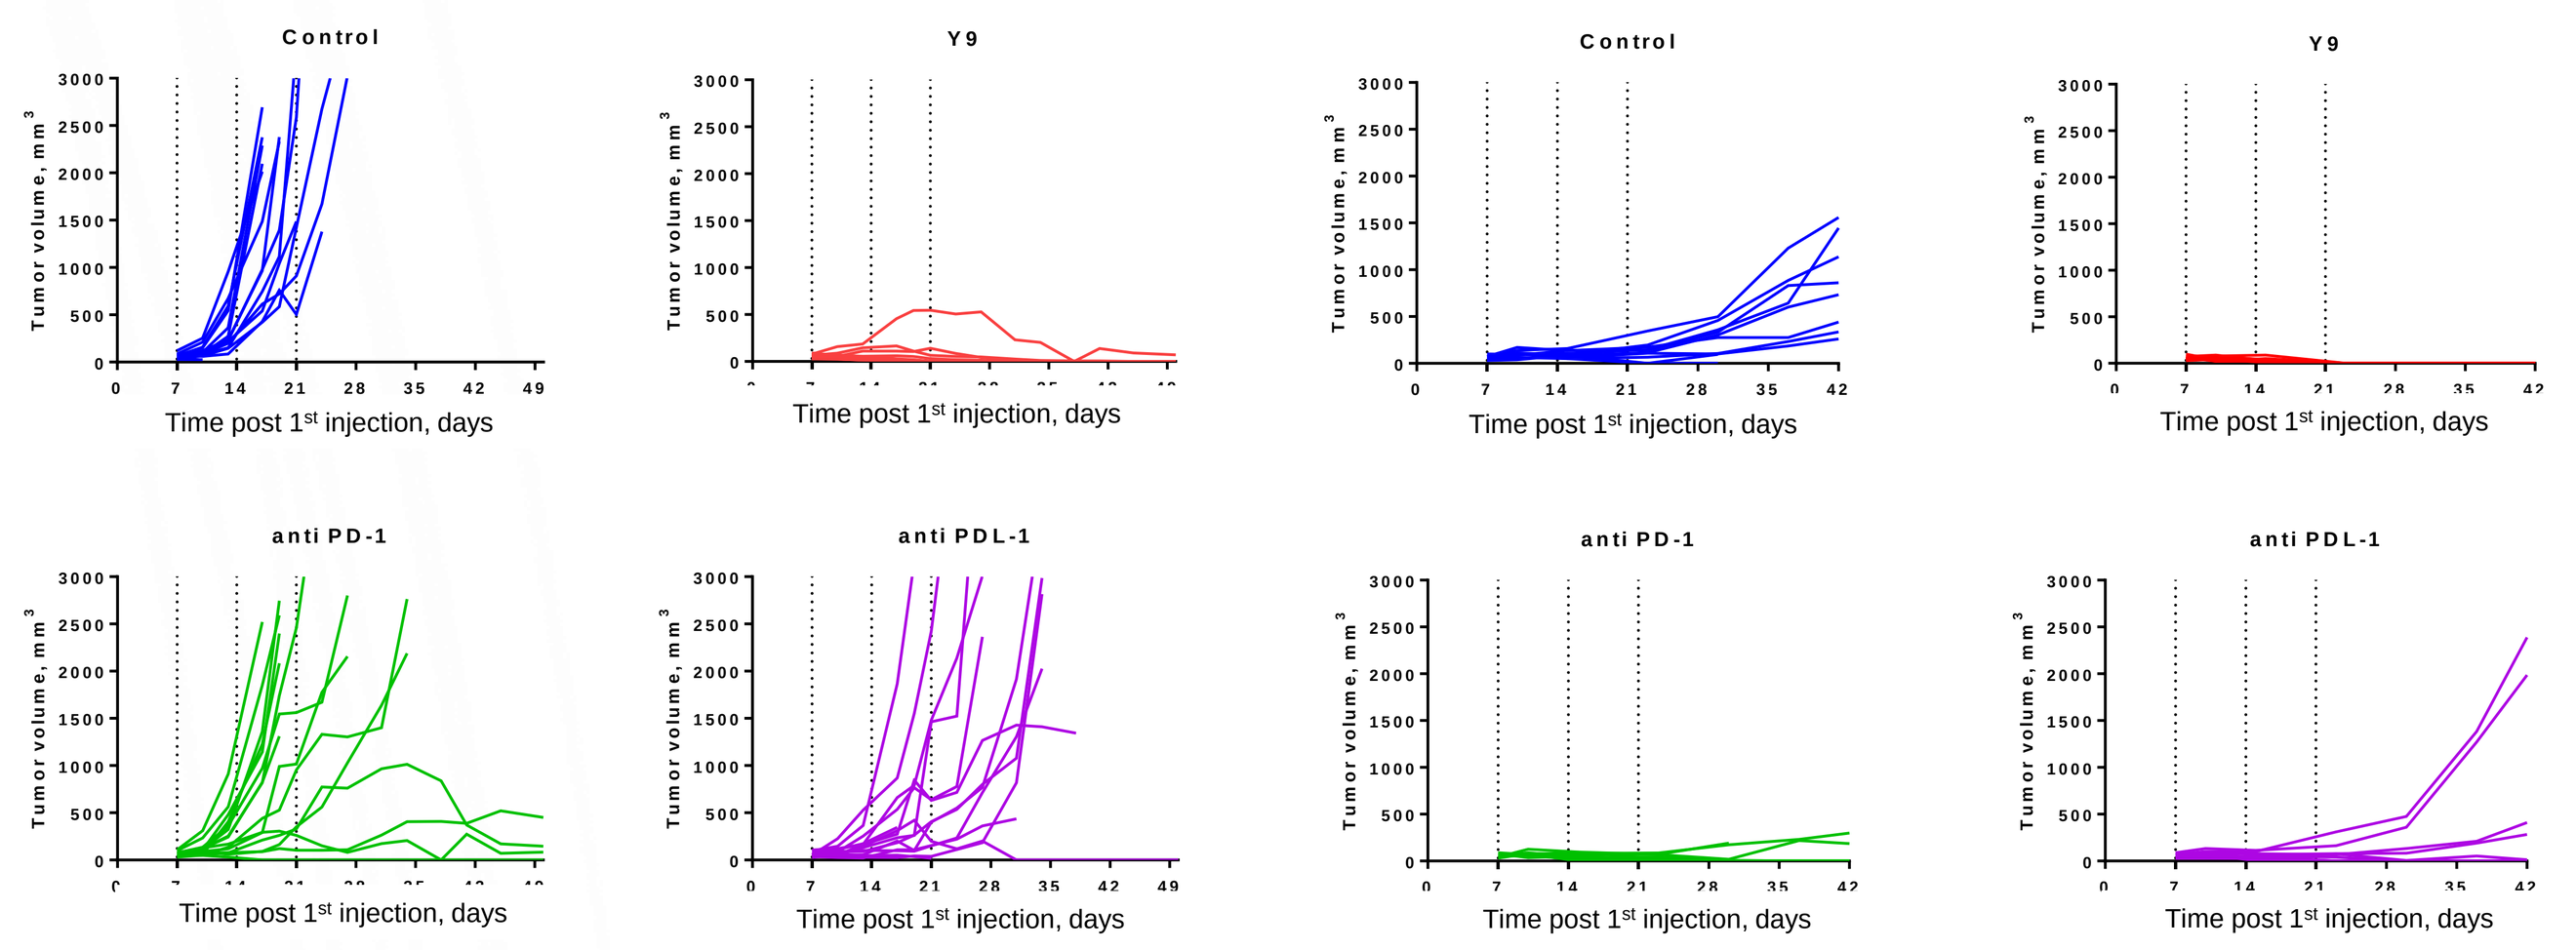
Control
Y9
anti PD-1
anti PDL-1
Tumor volume, mm3
Time post 1st injection, days

- 3 x 300 µg dose of murine surrogate anti-TNFR2 antibody (Y9)

Previously Treated Mice Show Immune Memory to Tumor Re-Challenge

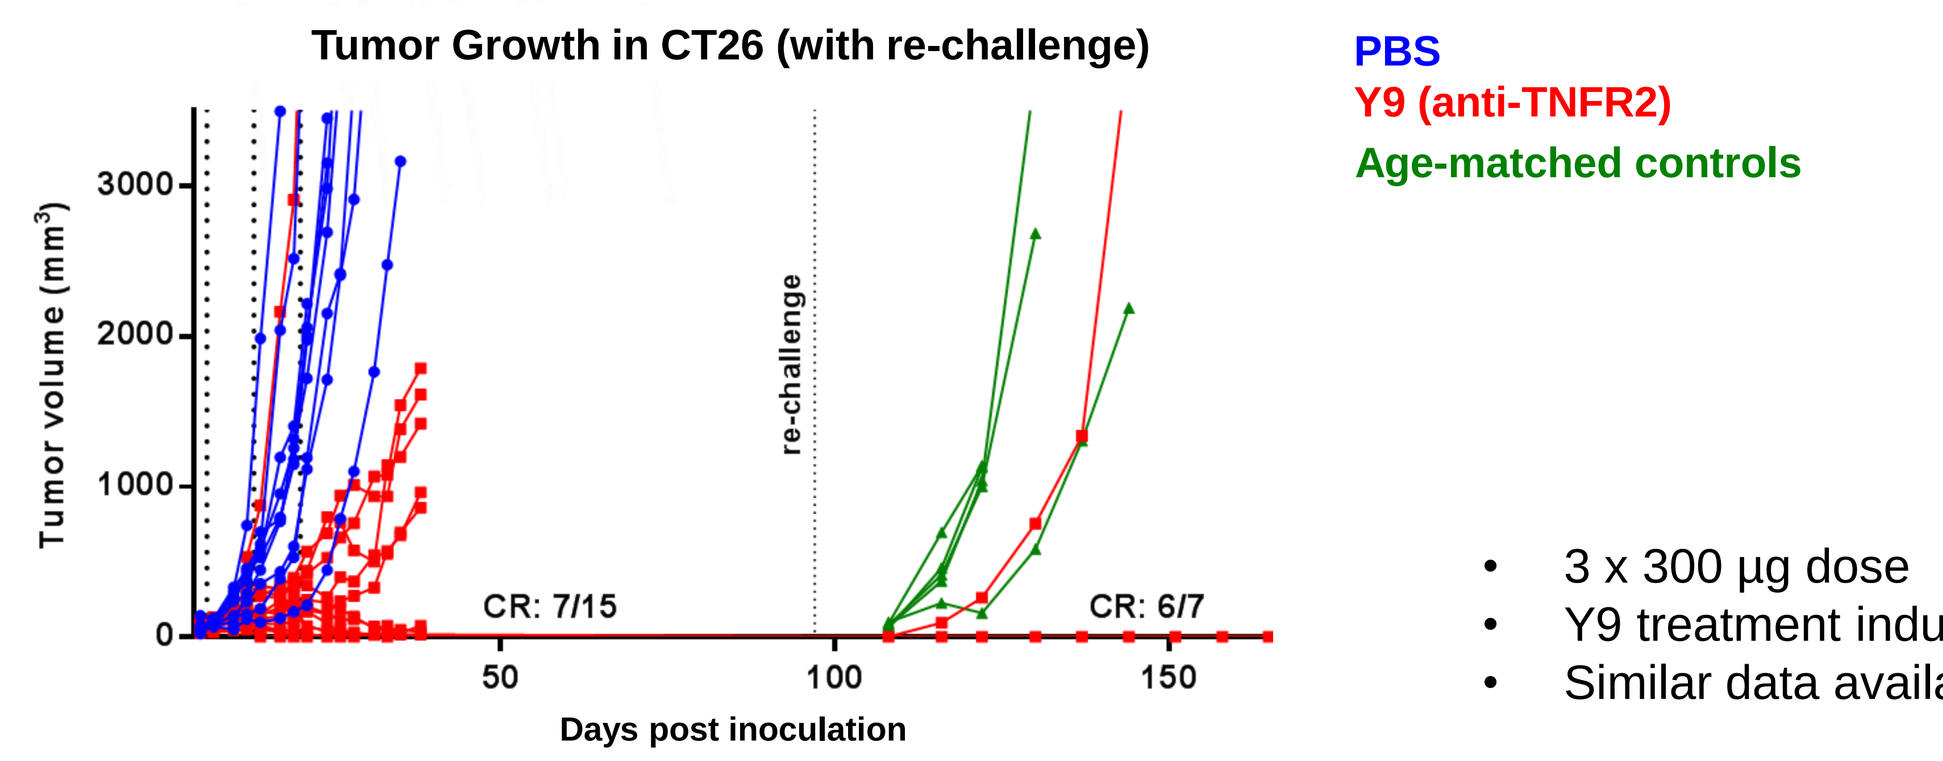
Tumor Growth in CT26 (with re-challenge)
PBS
Y9 (anti-TNFR2)
Age-matched controls
Tumor volume (mm3)
re-challenge
CR: 7/15
CR: 6/7
Days post inoculation

- 3 x 300 µg dose
- Y9 treatment induced long-term anti-tumor memory
- Similar data available for WEHI164 and EMT6

Anti-TNFR2 and Anti-PD1 Antibody Combination Leads to Superior Survival in Syngeneic Mouse Tumor Models

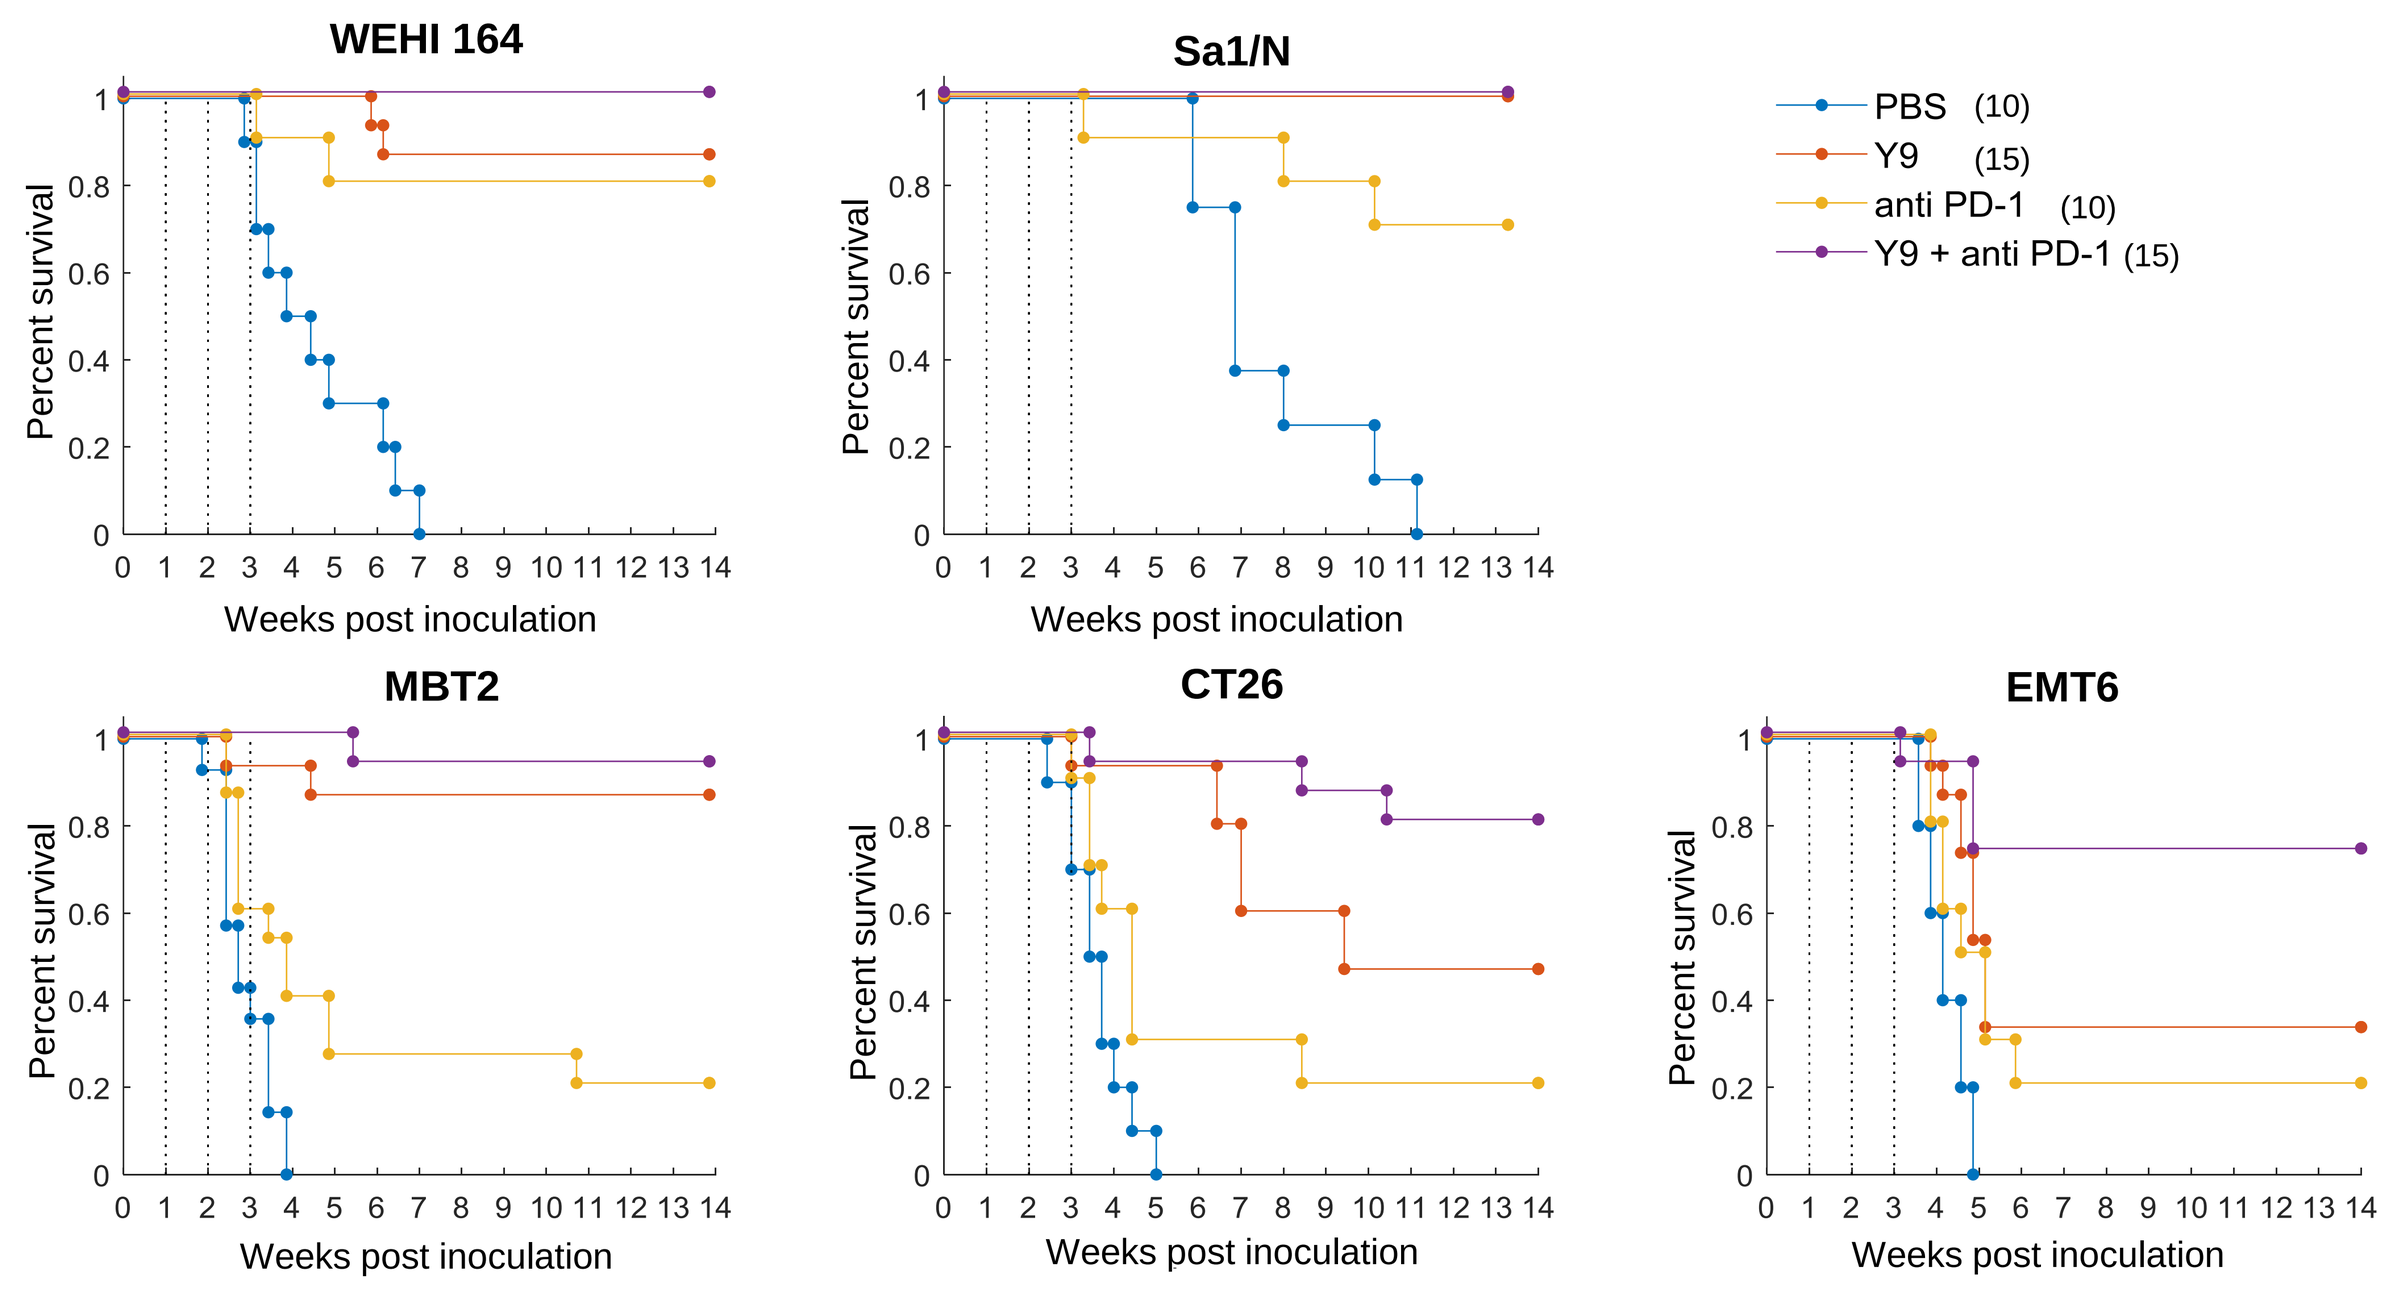
WEHI 164
Sa1/N
MBT2
CT26
EMT6
PBS (10)
Y9 (15)
anti PD-1 (10)
Y9 + anti PD-1 (15)
Percent survival
Weeks post inoculation

- 3 x 300 µg dose
- Similar data for PD-L1 available

Anti-TNFR2 Antibody has Favorable Toxicity Profile Compared to Anti-CTLA4 in Mouse Models

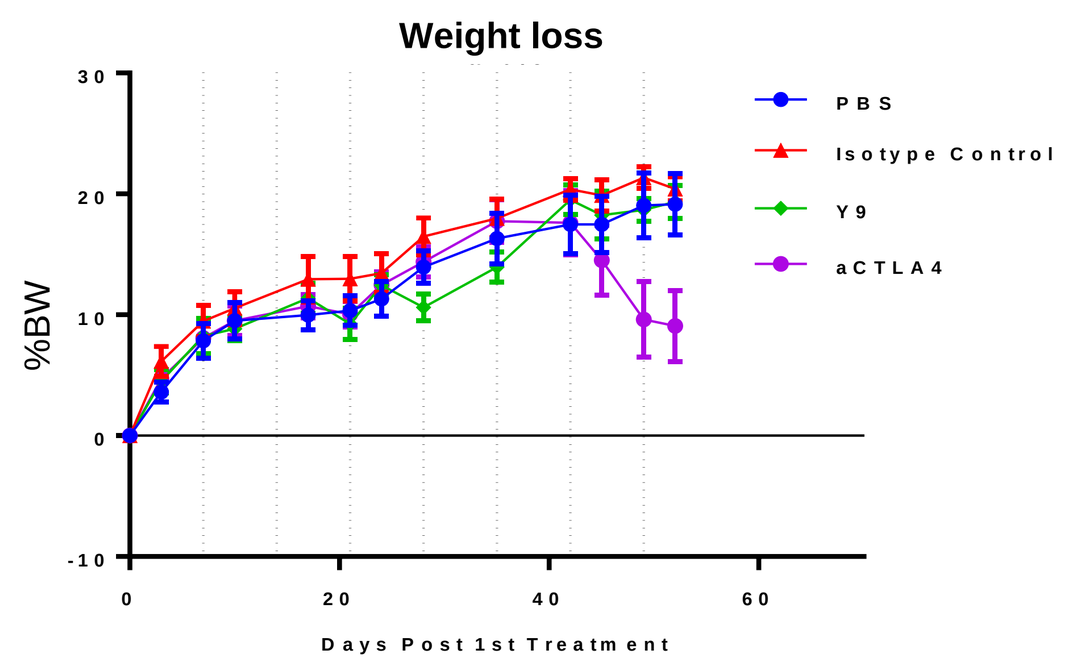
Weight loss
PBS
Isotype Control
Y9
aCTLA4
%BW
Days Post 1st Treatment

- 1mg dose, weekly, for 8 weeks

Anti-TNFR2 (Y9) group

- No obvious signs of pathology or toxicity signals detectable in blood

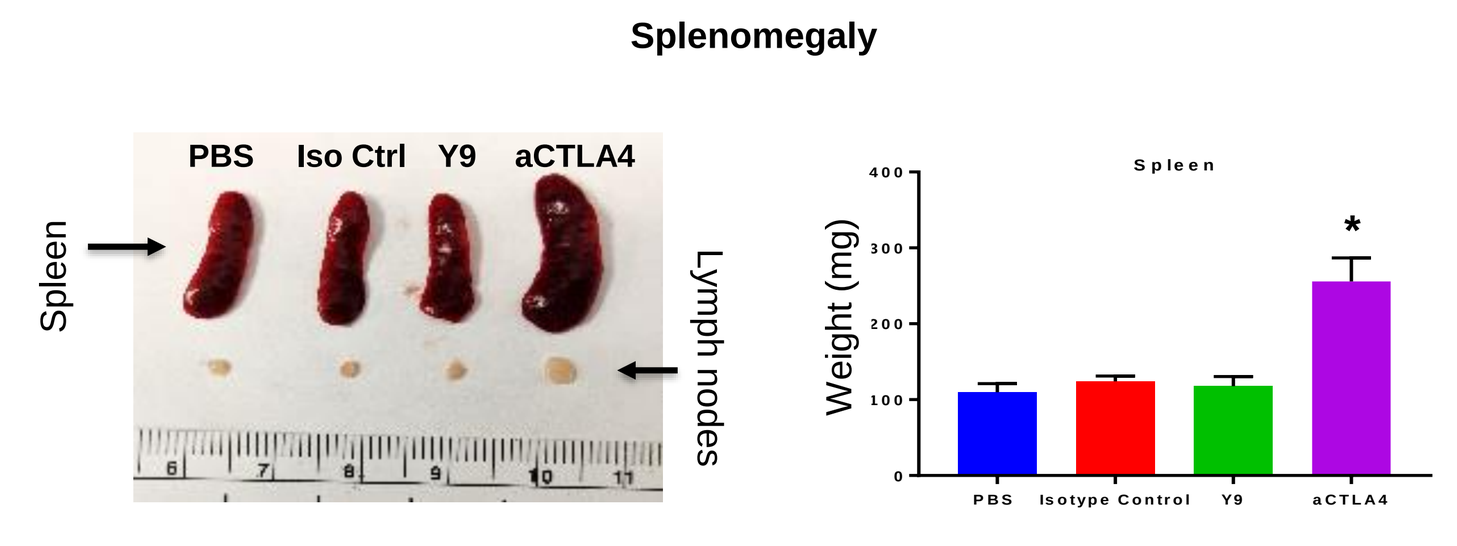
Splenomegaly
PBS
Iso Ctrl
Y9
aCTLA4
Spleen
Lymph nodes
Weight (mg)
Isotype Control

Anti-CTLA4 group

- Weight loss in entire group
- Dermatitis (legs, ears, some torso)
- Crusty, swollen eyes
- Mangy fur (mice were still active, no signs of diarrhea)
- Splenomegaly, grossly enlarged LNs
- Inflamed GI tract, enlarged intestines
- Increased liver enzymes, proliferating T cells, activated dendritic cells
- Chronic elevation of cytokines: IFNγ, IL-10, IL-2, IL-4, IL-5, IL-6, TNFα

Co-stimulatory Activity Increases Proliferation and Functionality of T cells In Vitro

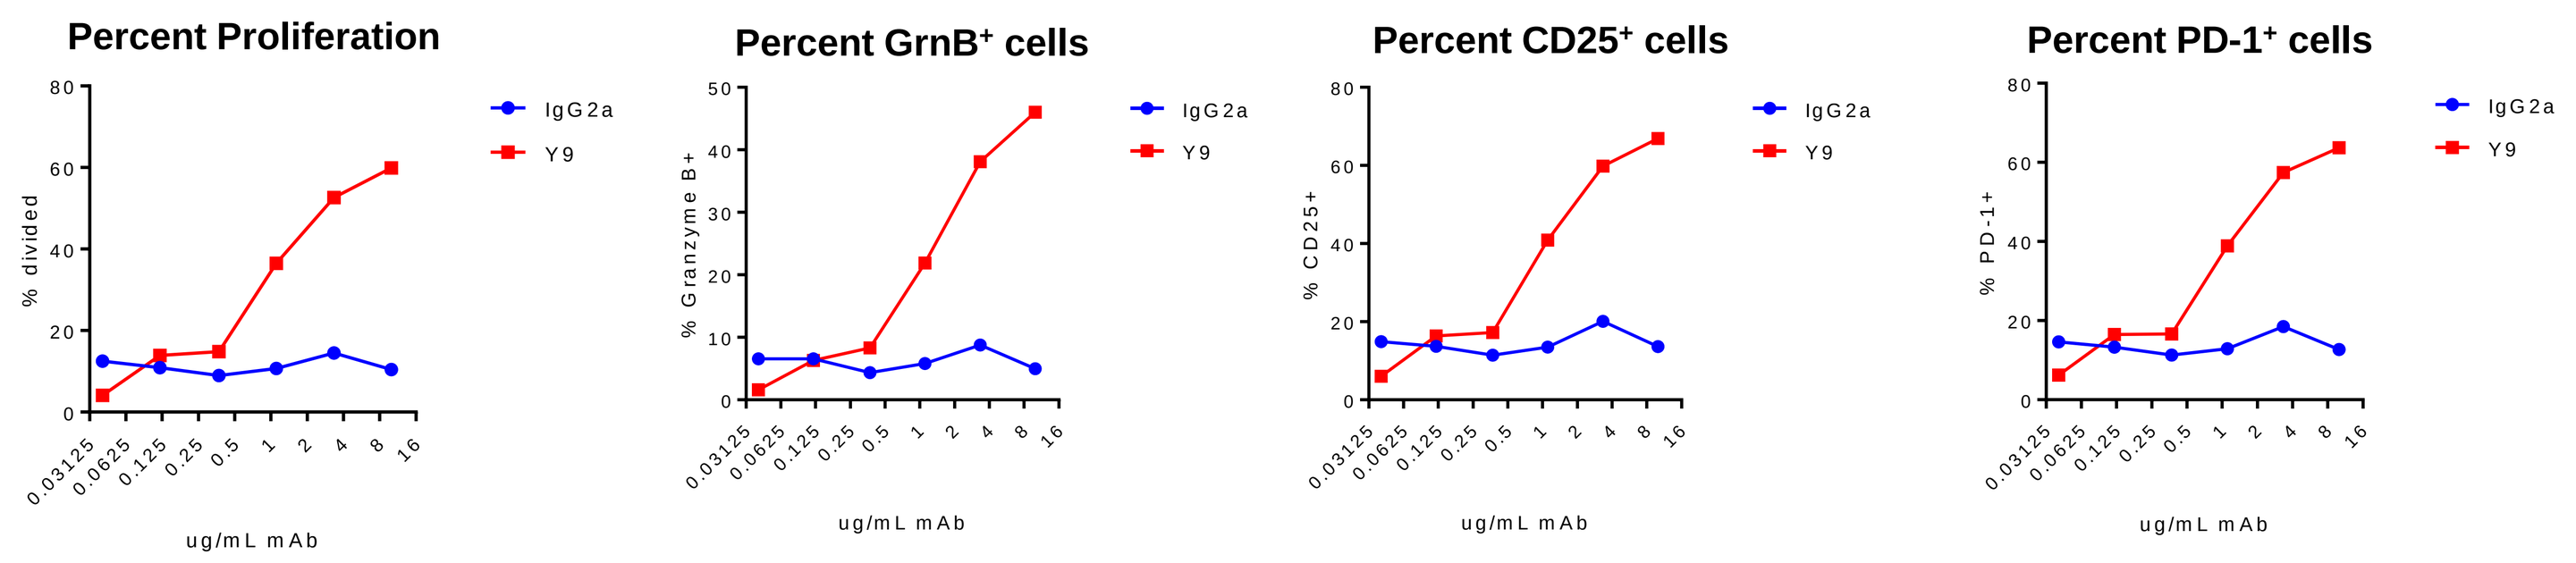
Percent Proliferation
Percent GrnB+ cells
Percent CD25+ cells
Percent PD-1+ cells
IgG2a
Y9
% divided
% Granzyme B+
% CD25+
% PD-1+
ug/mL mAb

- Isolated CD8+ T cells, activated with anti-CD3 (1.67µg/ml) and anti-CD28 (1 µg/ml)
- Dose titration of plate-bound Y9, readout after 3 days

Anti-TNFR2 Antibody Activity Requires Fcɣ Receptor Binding

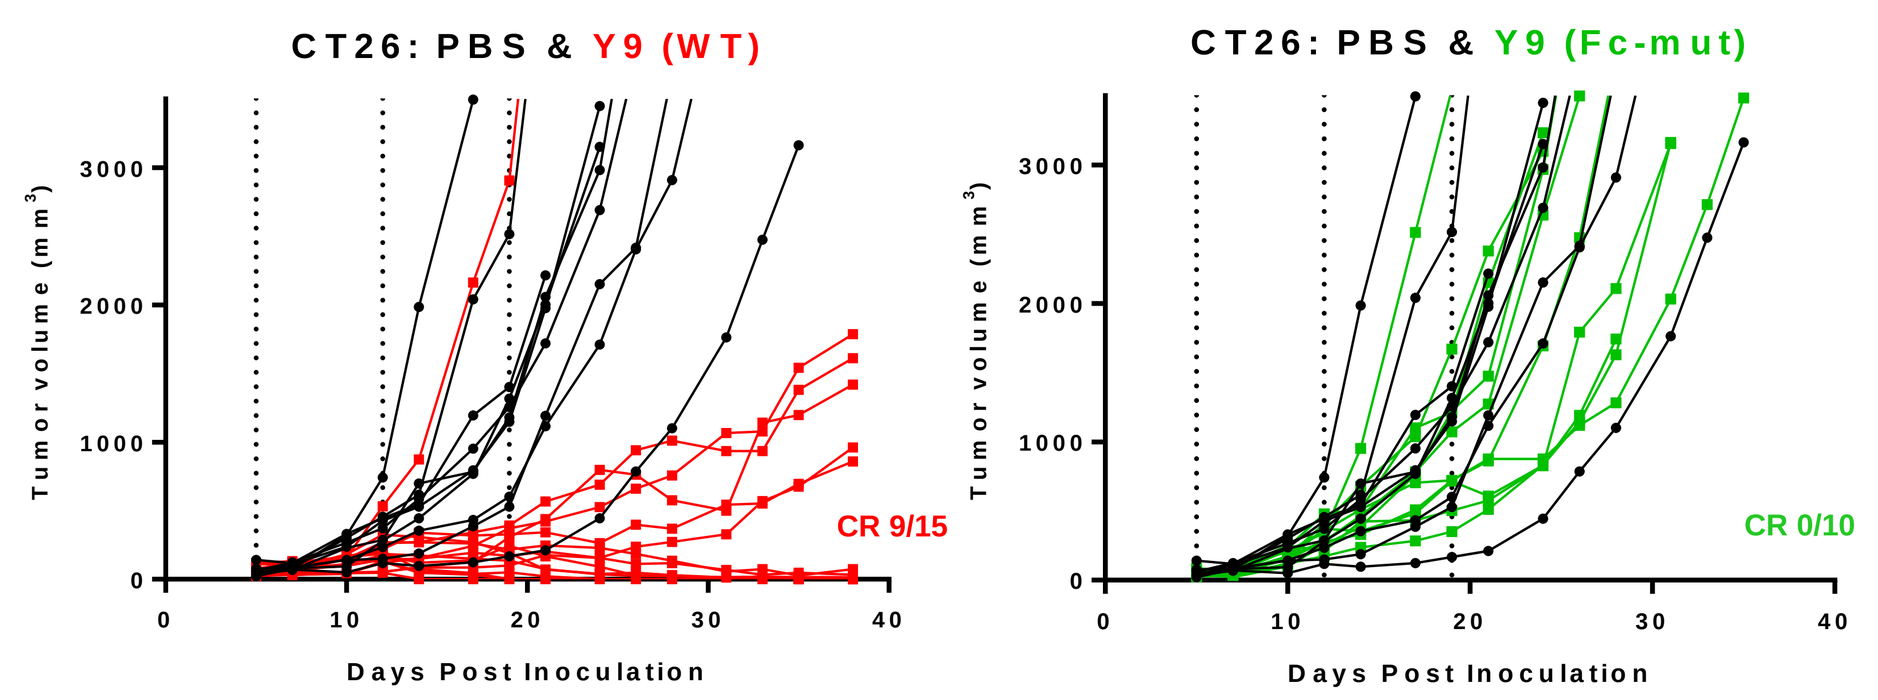
CT26: PBS & Y9 (WT)
CT26: PBS & Y9 (Fc-mut)
Tumor volume (mm3)
CR 9/15
CR 0/10
Days Post Inoculation

- Comparison of Y9 WT (mIgG2a) and Y9 Fc-mut (D265A/N297A) 3 x 300 µg dose
- Similar results available for Wehi164 and EMT6 models

Anti-TNFR2 Antibody Activity Does Not Require Expression of TNFR2 on Tumor Cells

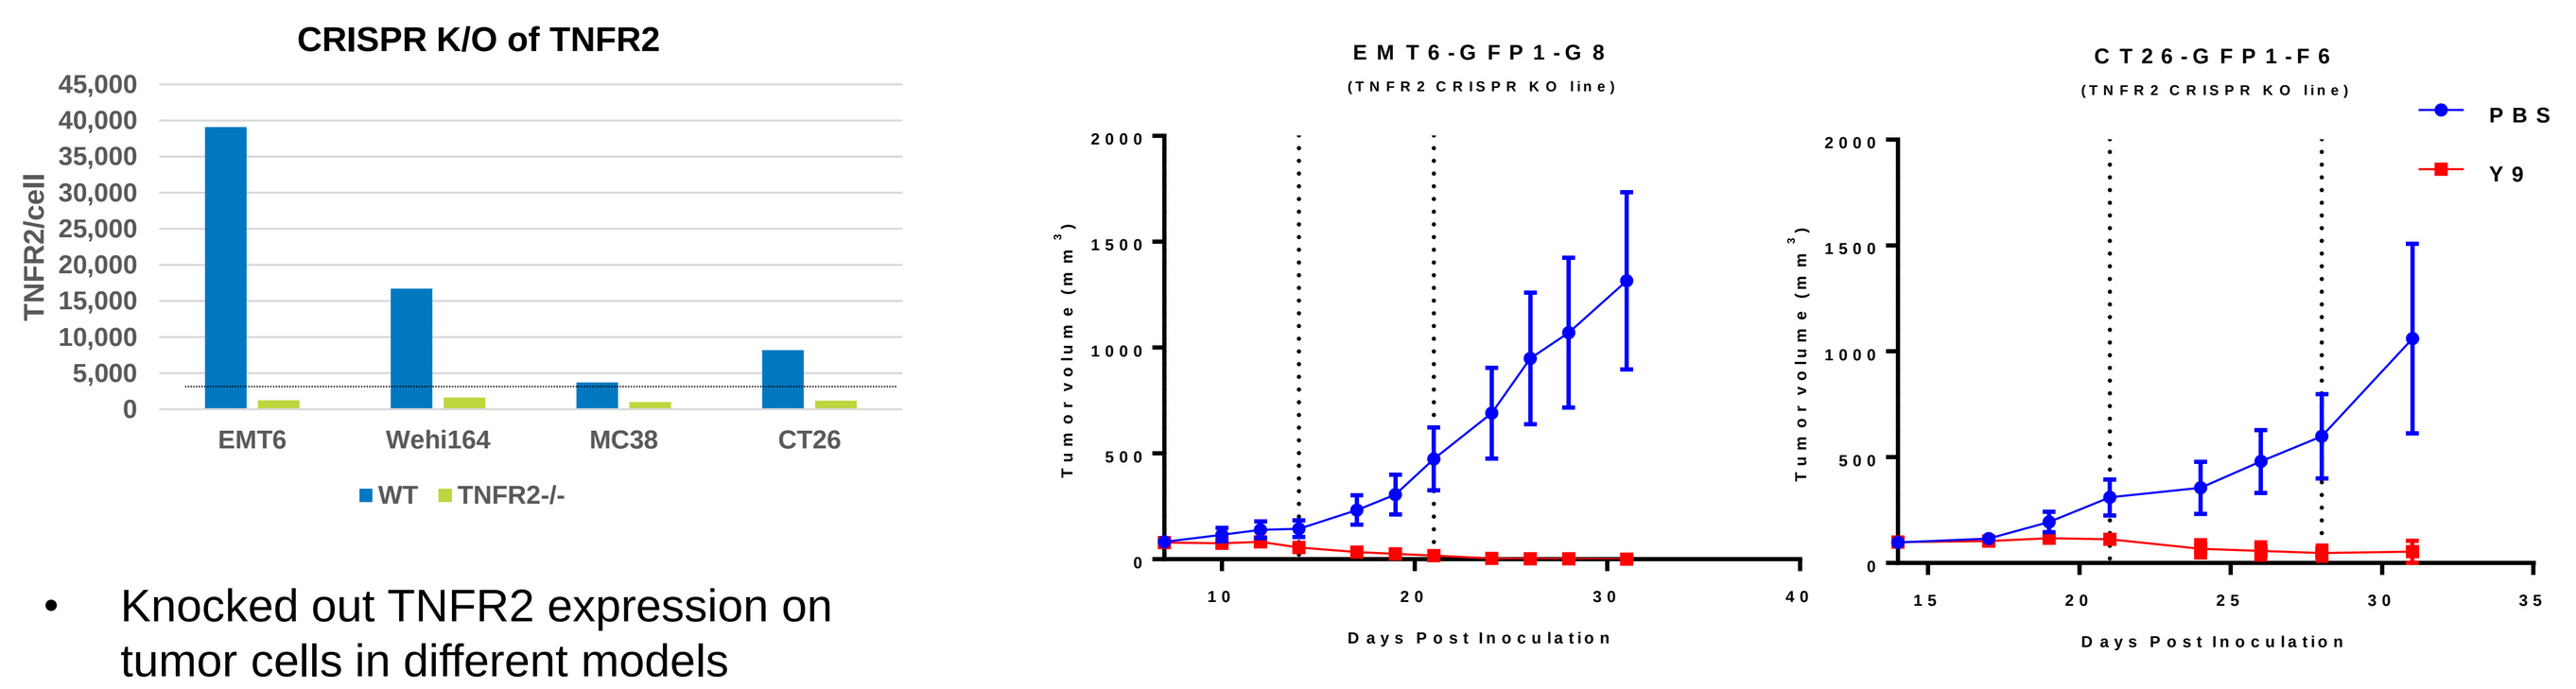
CRISPR K/O of TNFR2
TNFR2/cell
EMT6
Wehi164
MC38
CT26
WT
TNFR2-/-
EMT6-GFP1-G8
CT26-GFP1-F6
(TNFR2 CRISPR KO line)
PBS
Y9
Tumor volume (mm3)
Days Post Inoculation

- Knocked out TNFR2 expression on tumor cells in different models
- 3 x 300 µg dose; 10 mice/group (EMT6) and 5 mice/group (CT26)
- Y9 remains active in the absence of TNFR2 expression on tumor cells
- No direct contribution of TNFR2 on tumor cell to mechanism of action

CD8+ T Cells and NK Cells are Important for Anti-TNFR2 Antibody Activity

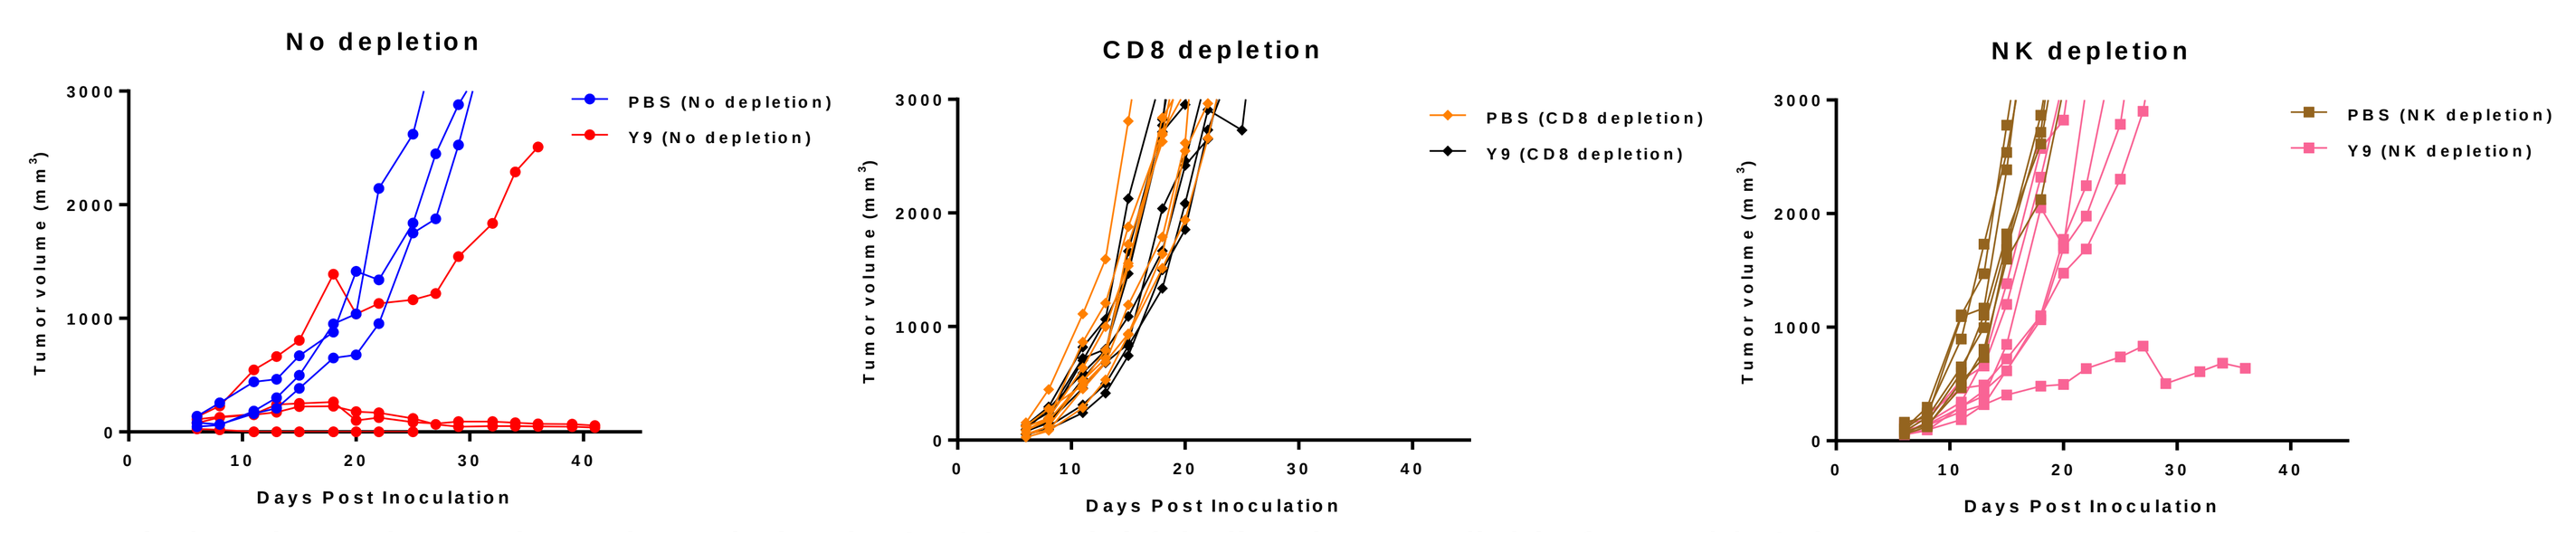
No depletion
CD8 depletion
NK depletion
PBS (No depletion)
Y9 (No depletion)
PBS (CD8 depletion)
Y9 (CD8 depletion)
PBS (NK depletion)
Y9 (NK depletion)
Tumor volume (mm3)
Days Post Inoculation

- Depletion three days prior to inoculation in CT26 model (similar data available for Wehi164)
- 1 x 1 mg of Y9 on day 7
- Data from CD4 depletion was inconclusive

Anti-TNFR2 Antibody Downregulates TNFR2 on T Cells Syngeneic Tumor Mouse Models *in vivo*

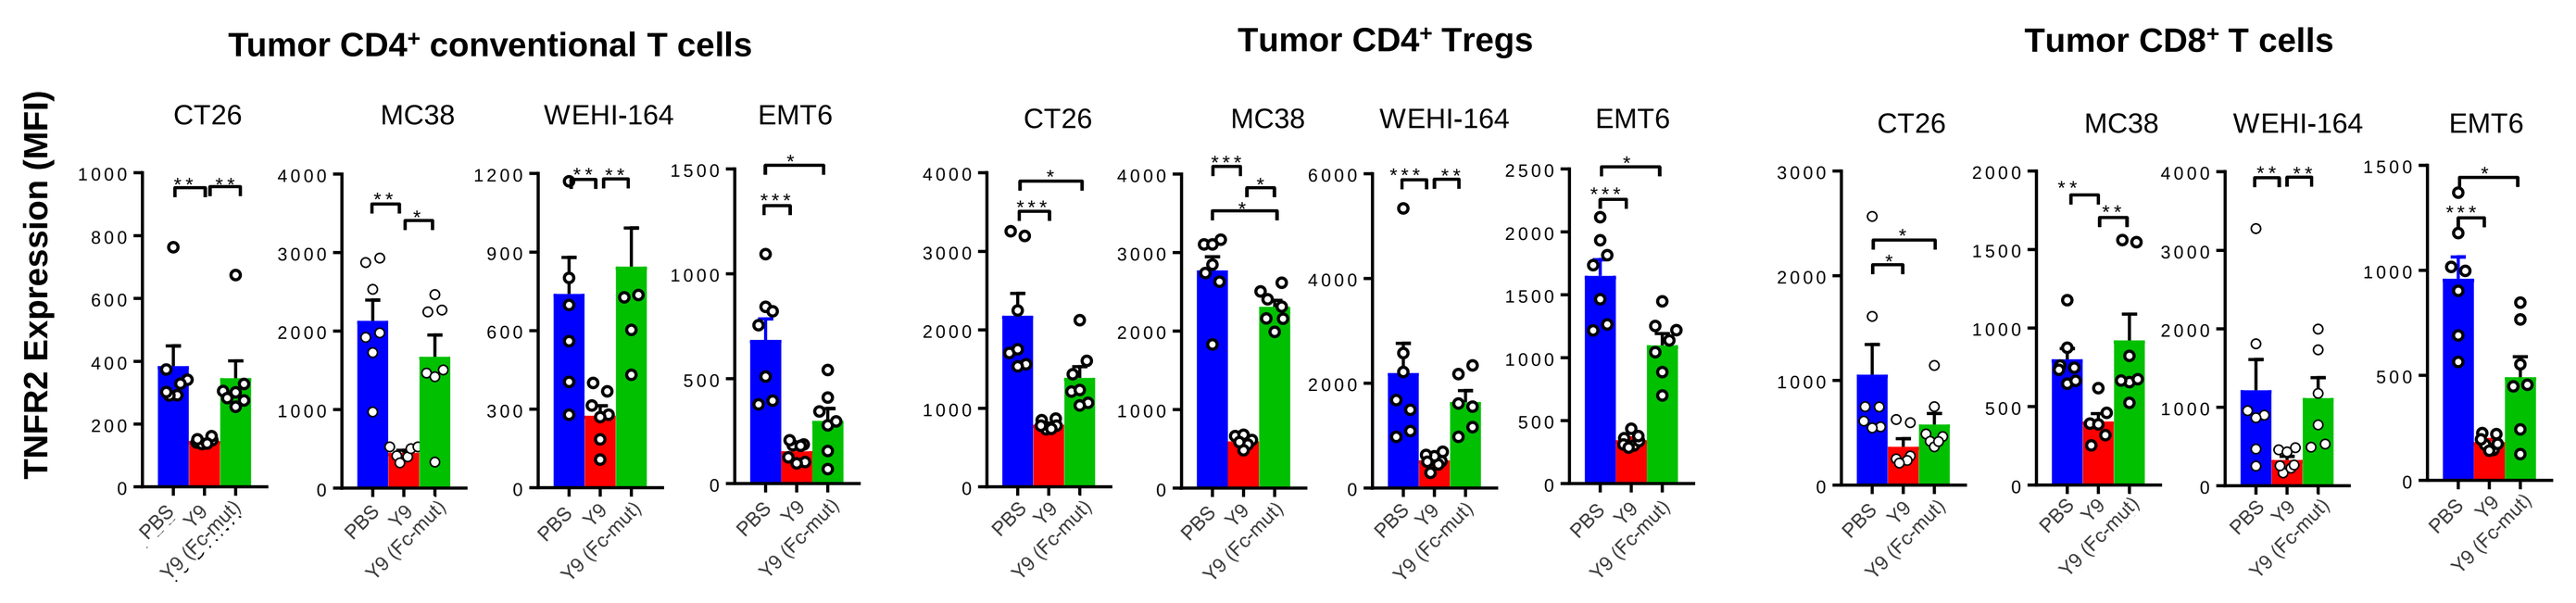
Tumor CD4+ conventional T cells
Tumor CD4+ Tregs
Tumor CD8+ T cells
CT26
MC38
WEHI-164
EMT6
TNFR2 Expression (MFI)
PBS
Y9
Y9 (Fc-mut)

- 24-36h after 300 µg of Y9
- Similar effect on tumor cells and myeloid cells
- Dependence on FcɣR-binding is potentially direct evidence for agonistic function

PBS
Y9 (WT mIgG2a)
Y9 Fc-mut (mIgG2a D265A/N297A)

Treg Depletion is Not Consistently Observed Across Responder Models

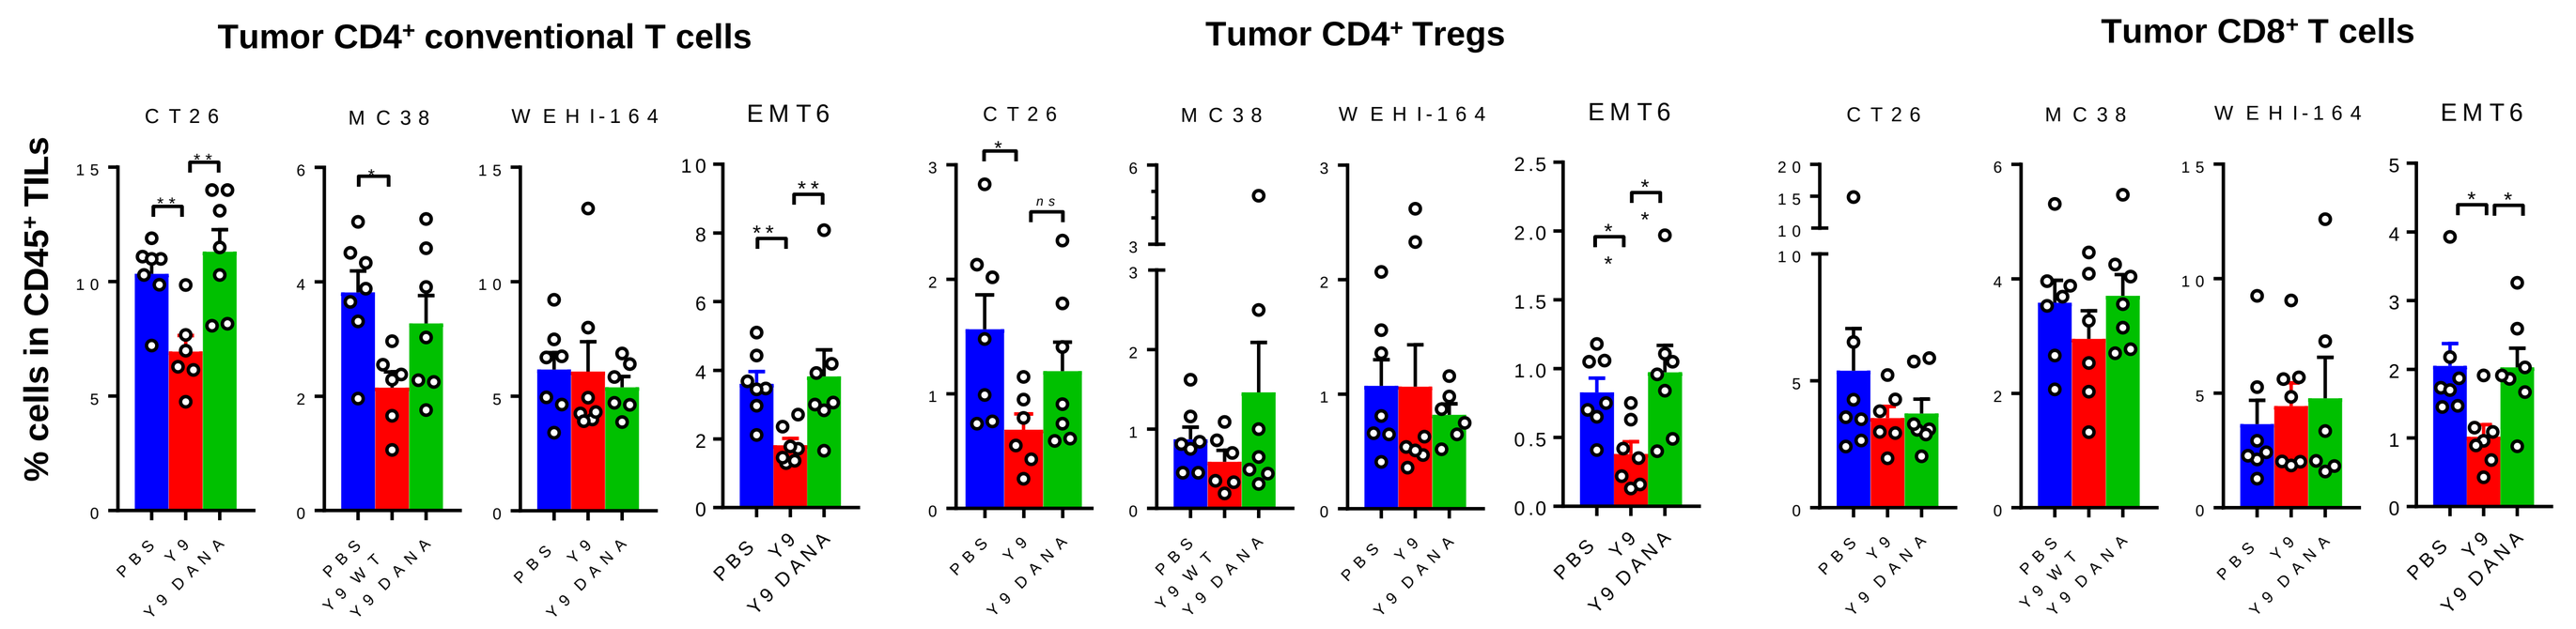
Tumor CD4+ conventional T cells
Tumor CD4+ Tregs
Tumor CD8+ T cells
CT26
MC38
WEHI-164
EMT6
% cells in CD45+ TILs
PBS
Y9
Y9 WT
Y9 DANA

- 24-36h after 300 µg of Y9

PBS
Y9 (mIgG2a)
Y9 DANA (mIgG2a D265A/N297A)

Significant CD8/Treg Ratio Change is Not Observed

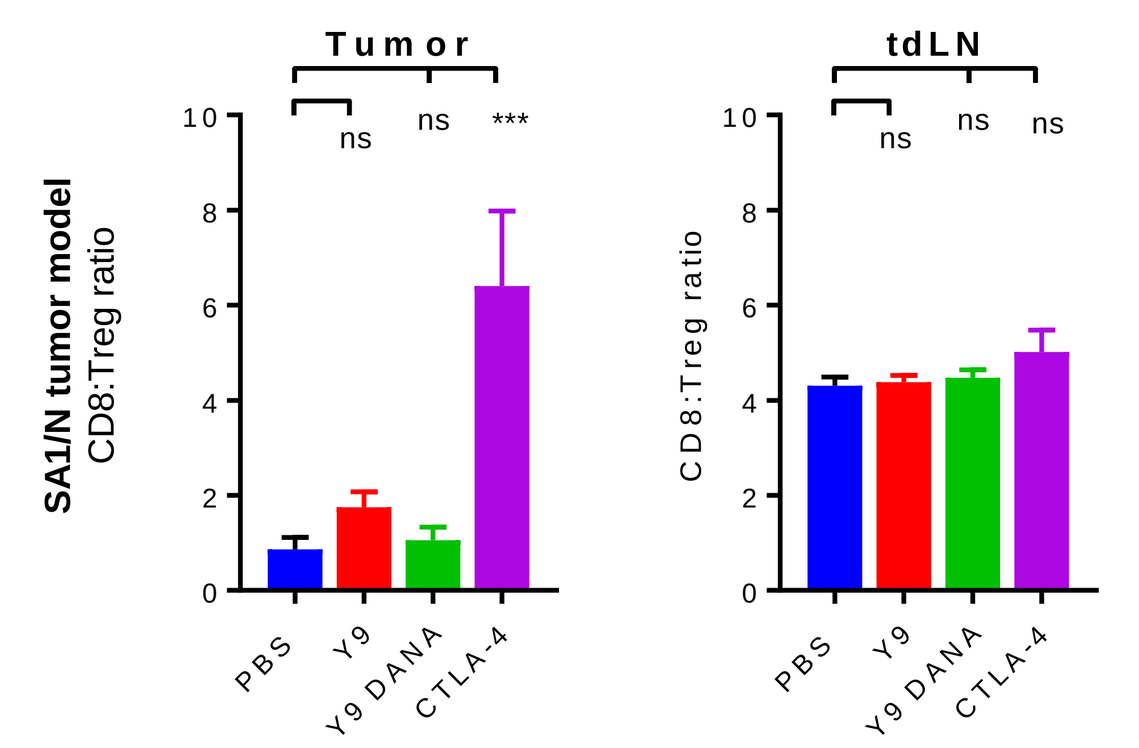
Tumor
tdLN
SA1/N tumor model
CD8:Treg ratio
ns

PBS
Y9
Y9 DANA
CTLA-4

- 24h after 300 µg dose

Anti-TNFR2 Antibody Treatment Resulted in CTLA-4 Downregulation

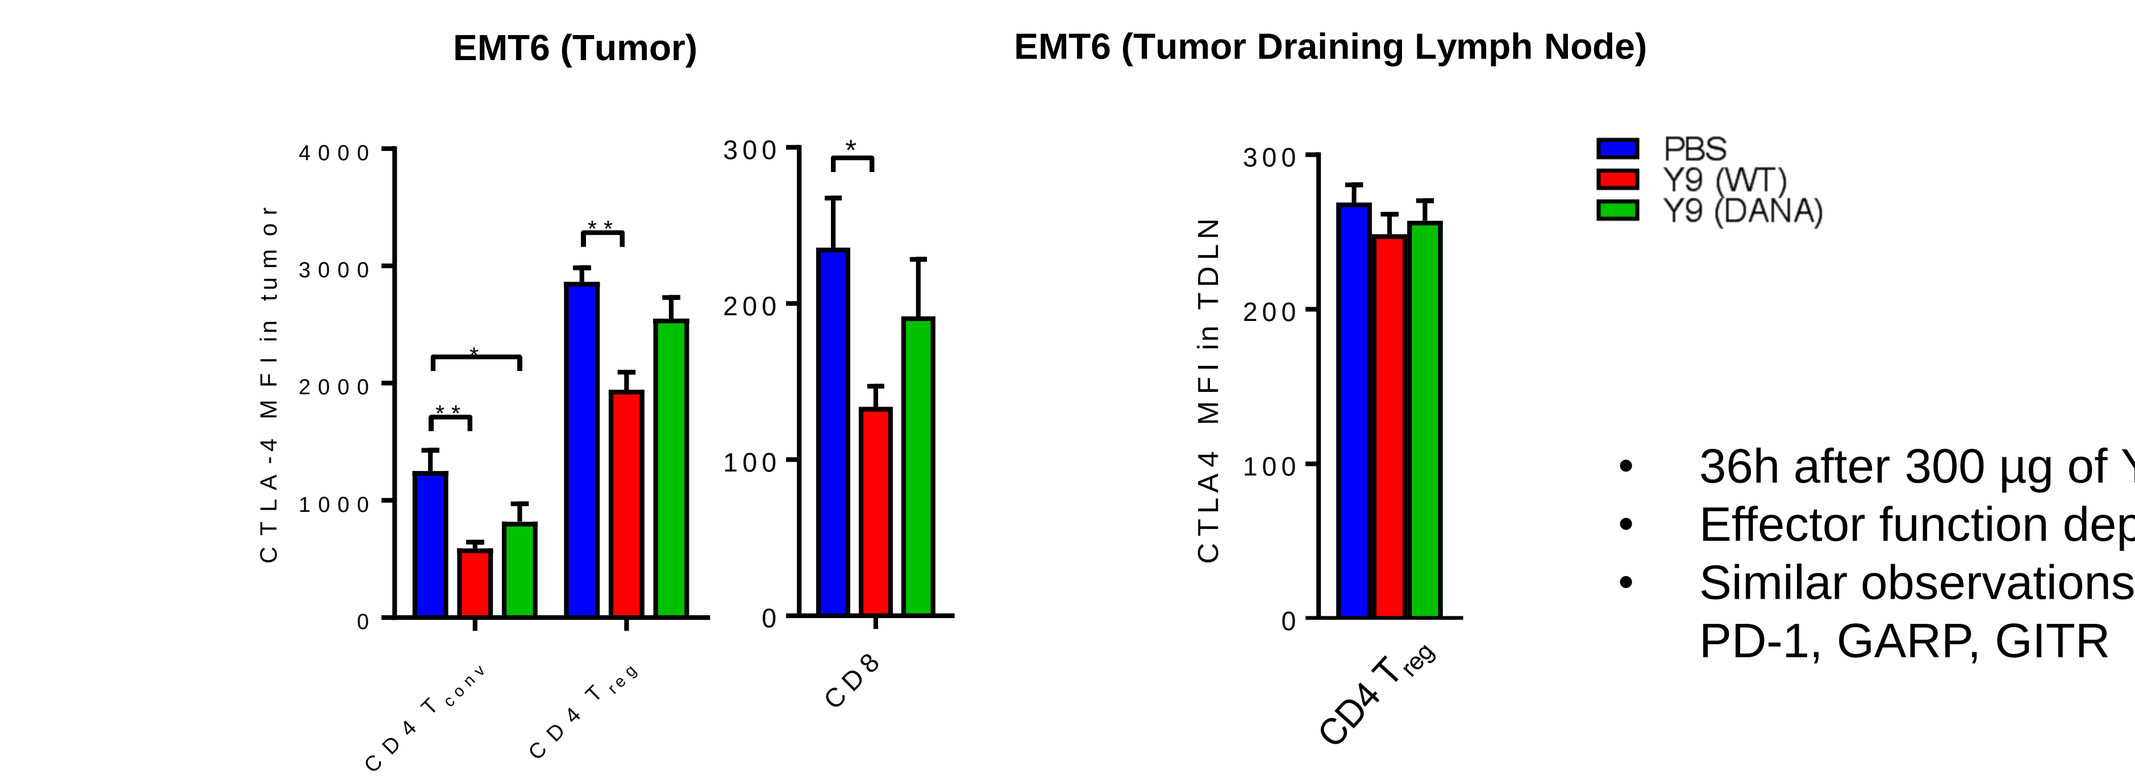
EMT6 (Tumor)
EMT6 (Tumor Draining Lymph Node)
CTLA-4 MFI in tumor
CTLA4 MFI in TDLN
CD4 Tconv
CD4 Treg
CD8
PBS
Y9 (WT)
Y9 (DANA)

- 36h after 300 µg of Y9
- Effector function dependent
- Similar observations for other markers, e.g. PD-1, GARP, GITR

Mechanism of Action Summary

Dominant Mechanism:

- Activity requires T cells, NK cells, and FcɣR binding
- Co-stimulatory activity via TNFR2 signaling
 - ✓ Increased proliferation and functionality of CD8+ T cells *in vitro*
 - ✓ FcɣR-dependent increase in TNFR2 receptor downregulation *in vivo*
 - ✓ Downregulation of T cell-linked immunosuppressive markers as secondary treatment effect

Other Mechanisms Under Investigation:

- Direct depletion of immune cell subsets (including potentially Tregs) via ADCC or ADCP
 - ✗ No consistent treatment induced changes in the frequency of T cell subsets in the tumor and periphery
 - **In Progress:** Determine importance of activating *vs.* inhibitory Fcɣ receptors using $FcɣR^{-/-}$ mice and comparison of mIgG2a and mIgG1
- Direct phenotypic effect on Tregs
 - **In Progress:** Determine importance of TNFR2 on Tregs *in vitro*, and in studies in Treg-deficient DEREG mice

Conclusion

Treatment with our murine surrogate anti-TNFR2 antibody led to robust responses in multiple syngeneic mouse tumor models both alone and in combination with checkpoint inhibition. The effect was specific to the tumor microenvironment, as no effect on Tregs in the periphery was observed. We have identified MM-401, a therapeutic antibody that binds human TNFR2 with low nanomolar affinity at the same epitope corresponding to Y9 for the murine form of TNFR2. MM-401 is being developed to potentially become a treatment option for patients that are refractory to, or relapsing on, currently approved immune checkpoint inhibitors.

All data unpublished and on file at Merrimack Pharmaceuticals, Inc.

A PASSION FOR OUTTHINKING CANCER

© 2018 Merrimack Pharmaceuticals, Inc. All rights reserved.

merrimack