Abstract: AACR- 3270

Mechanism of action of a novel agonist TNFR2-antibody that induces co-stimulation of T cells and promotes robust anti-tumor immunity

R. Fulton*, A. Camblin*, J. Sampson, J. Richards, C. Wong, A. Koshkaryev, L. Luus , Y. Jiao, L. Xu, V. Paragas, M. Razlog, M. Muda, E.M. Tam, D.C. Drummond, A. Raue

Merrimack Pharmaceuticals, Inc., Cambridge MA, USA

* Contributed equally

Summary

TNFR2 is a member of the TNF receptor superfamily that is upregulated upon T cell activation and is highly expressed by tumor-infiltrating effector and regulatory T cells (T_{regs}). We investigated TNFR2 levels on T cells in syngeneic mouse tumor models and in secondary lymphoid tissues by flow cytometry. Non-regulatory T cells in the spleen and lymph nodes expressed little TNFR2 whereas T_{regs} constitutively expressed intermediate levels. In contrast, tumor-infiltrating effector T cells expressed high levels of TNFR2 with T_{regs} expressing the highest levels. To investigate TNFR2 as a therapeutic target, we generated a novel monoclonal antibody specific to murine TNFR2 and investigated its mechanism of action. Antibodies against murine TNFR2 were generated by screening a human antibody-display library or by rabbit immunization. Antibodies were assessed for affinity, ability to compete with TNF and for developability. A select number of antibodies were expressed as murine IgG2a and evaluated for activity in multiple syngeneic mouse tumor models. The mechanism of action of the most active clone, Y9, was investigated further. *In vitro,* Y9 stimulation of purified T cells from healthy mice caused increased proliferation and effector function, indicating that Y9 has agonist properties and can provide co-stimulation. *In vivo*, Y9 treatment of mice with established tumors resulted in complete tumor clearance across a variety of models. Using CRISPR knockout cell lines, we showed that Y9 activity did not depend on TNFR2 expression on tumor cells. However, Y9 required T cells as it showed no activity in nude mice. The activity of Y9 on immune cells was further confirmed by its decreased activity in mice depleted of NK or CD8$^+$ T cells. Unlike the proposed T_{reg}-depletion mechanism for other co-stimulatory therapeutic antibodies, depletion of T_{regs} is not the primary mechanism of action of Y9 treatment. Instead, Y9 provides potent co-stimulation to anti-tumor CD8$^+$ T cells that enhances their capacity to produce effector cytokines. Y9 activity depended on FcγR binding as demonstrated by the lack of activity of an antibody variant with mutations preventing FcγR binding. We showed further that FcγR binding facilitated enhanced agonist activity by comparing activity of Y9 variants with different Fc isotypes and in FcγR knockout mice. We present a novel anti-TNFR2 antibody that exhibited pronounced anti-tumor *in vivo* activity in our mouse models with co-stimulation of tumor-specific T cells as its dominant mechanism of action. A corresponding human anti-TNFR2 antibody (MM-401) has been identified and is being developed as a potential novel treatment option for cancer patients.

Why target TNFR2?

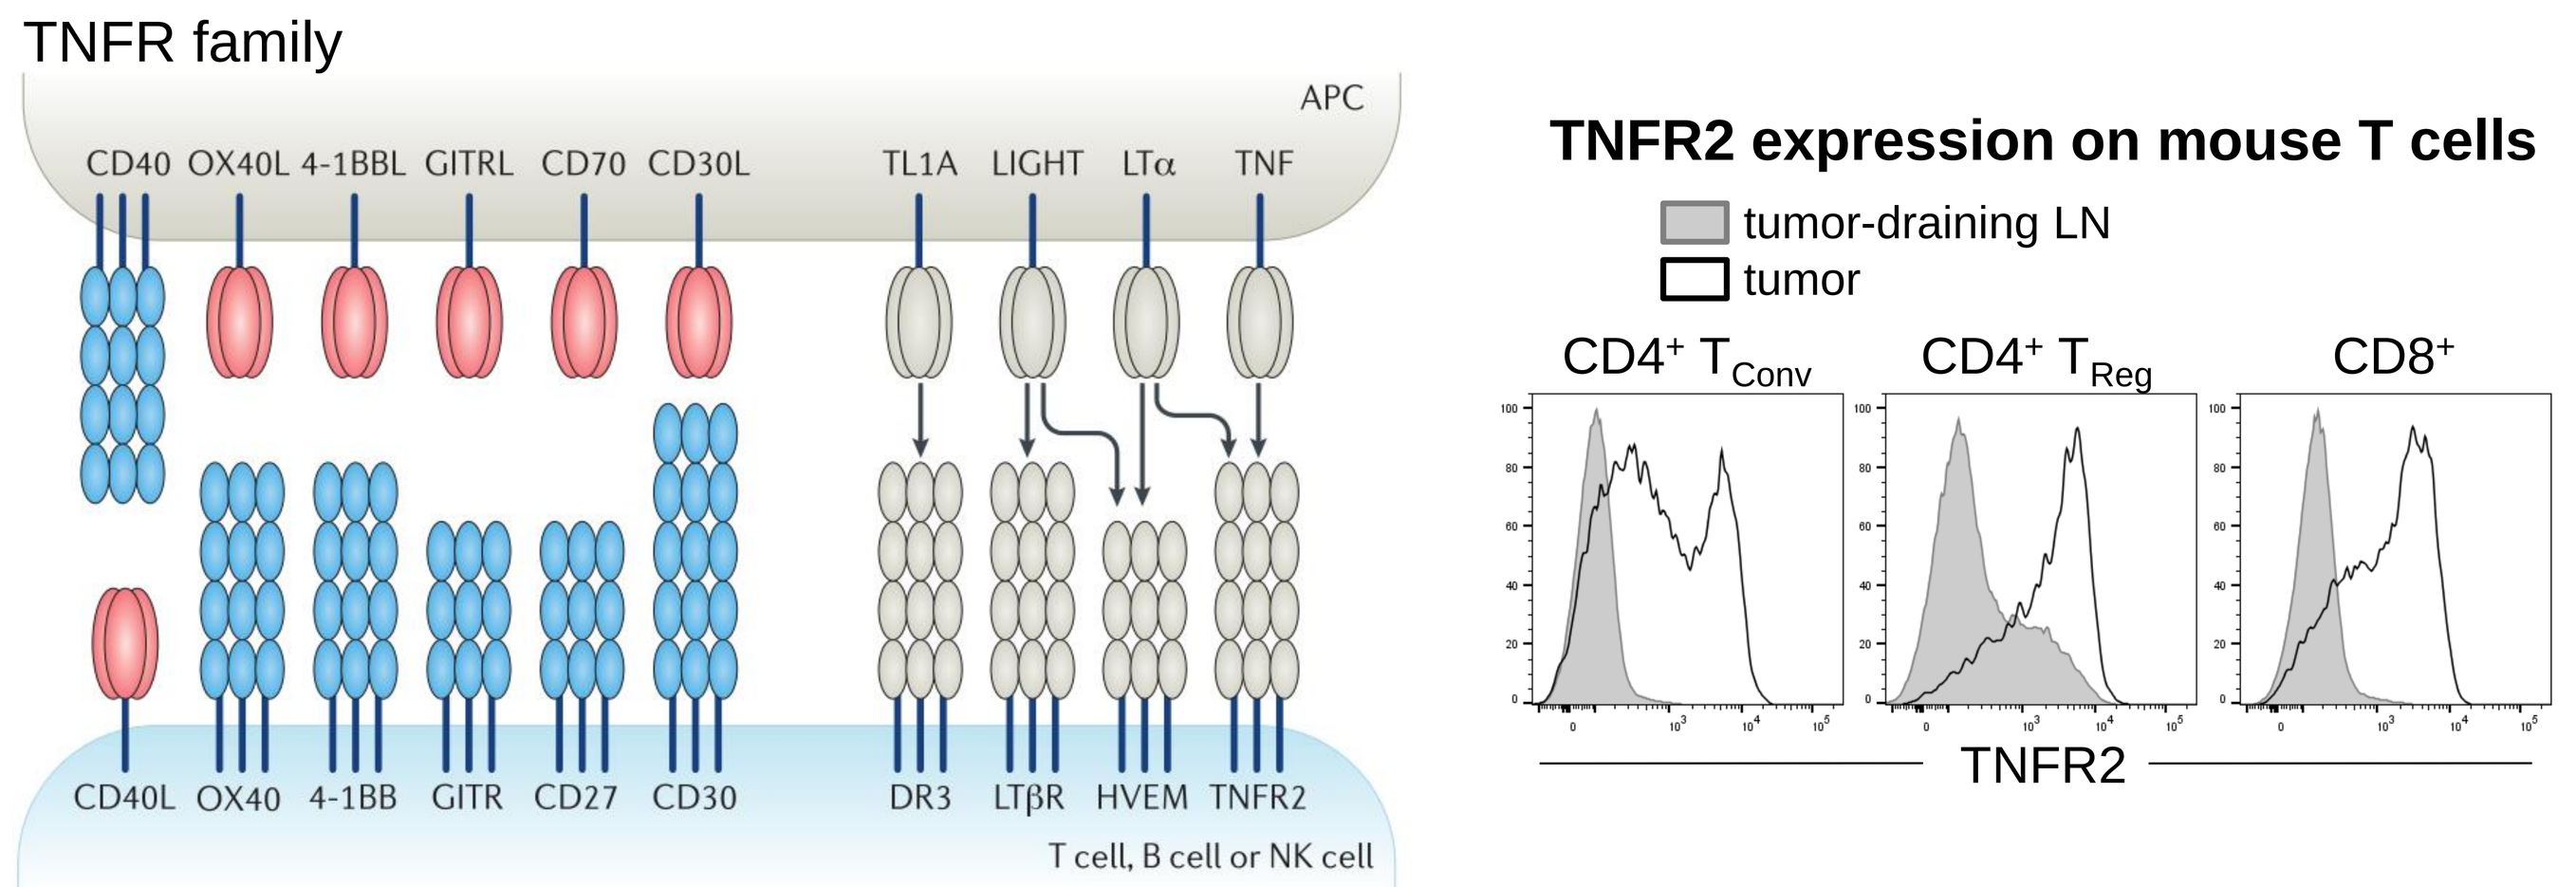

TNFR family
APC
CD40 OX40L 4-1BBL GITRL CD70 CD30L
TL1A LIGHT LTα TNF
CD40L OX40 4-1BB GITR CD27 CD30
DR3 LTβR HVEM TNFR2
T cell, B cell or NK cell
TNFR2 expression on mouse T cells
tumor-draining LN
tumor
CD4+ TConv
CD4+ TReg
CD8+
TNFR2

Mayes, P.A et al. 2018. *Nat. Rev. Drug Discov.* Jul;17(7):509-527

- Transmembrane (not soluble) TNF is the primary signaling ligand for TNFR2
- Unlike the broad tissue expression of TNFR1, TNFR2 expression is primarily restricted to immune cells
- TNFR2 on T cells is activation-associated
 - Highly expressed on activated effector T cells, CD4 T_{regs}. Low in periphery, high in tumor
 - T-cell expression patterns similar between mouse and human
- Pre-clinical evidence that TNFR2 provides co-stimulation to effector T cells and influences inhibitory state of T_{regs}

Characterization of murine surrogate anti-TNFR2 antibodies

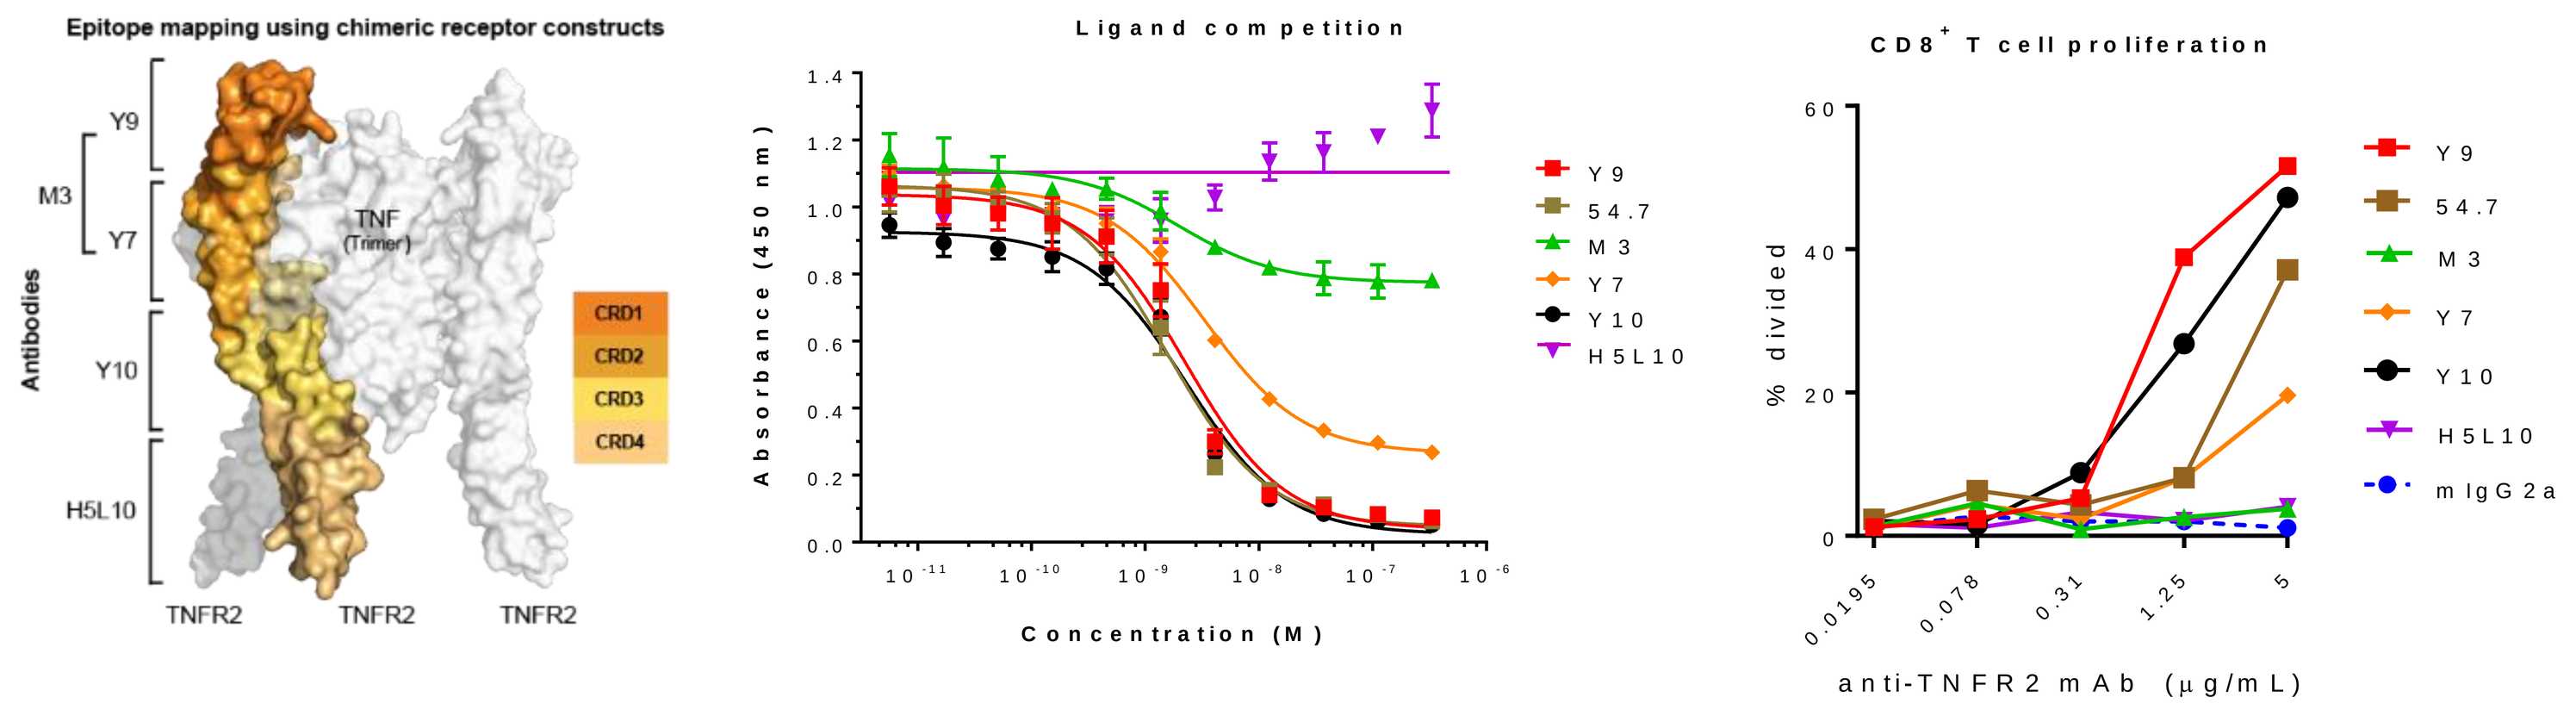

Epitope mapping using chimeric receptor constructs
Antibodies
Y9
M3
Y7
Y10
H5L10
TNF (Trimer)
CRD1
CRD2
CRD3
CRD4
TNFR2 TNFR2 TNFR2
Ligand competition
Absorbance (450 nm)
Concentration (M)
Y9
54.7
M3
Y7
Y10
H5L10
CD8+ T cell proliferation
% divided
anti-TNFR2 mAb (μg/mL)
mIgG2a

- Based on efficacy studies comparing anti-TNFR2 candidate antibodies, clone Y9 was selected as our lead mouse surrogate antibody.
- Y9 (mIgG2a) binds murine TNFR2 with an affinity of 0.5 nM as measured by biolayer interferometry (BLI)
- Y9 is a potent murine anti-TNFR2 antibody and binds well defined critical epitope
- Y9 is a TNF competitor, but TNF competition is not required for activity

Co-stimulatory activity increases proliferation and functionality of murine T cells *in vitro*

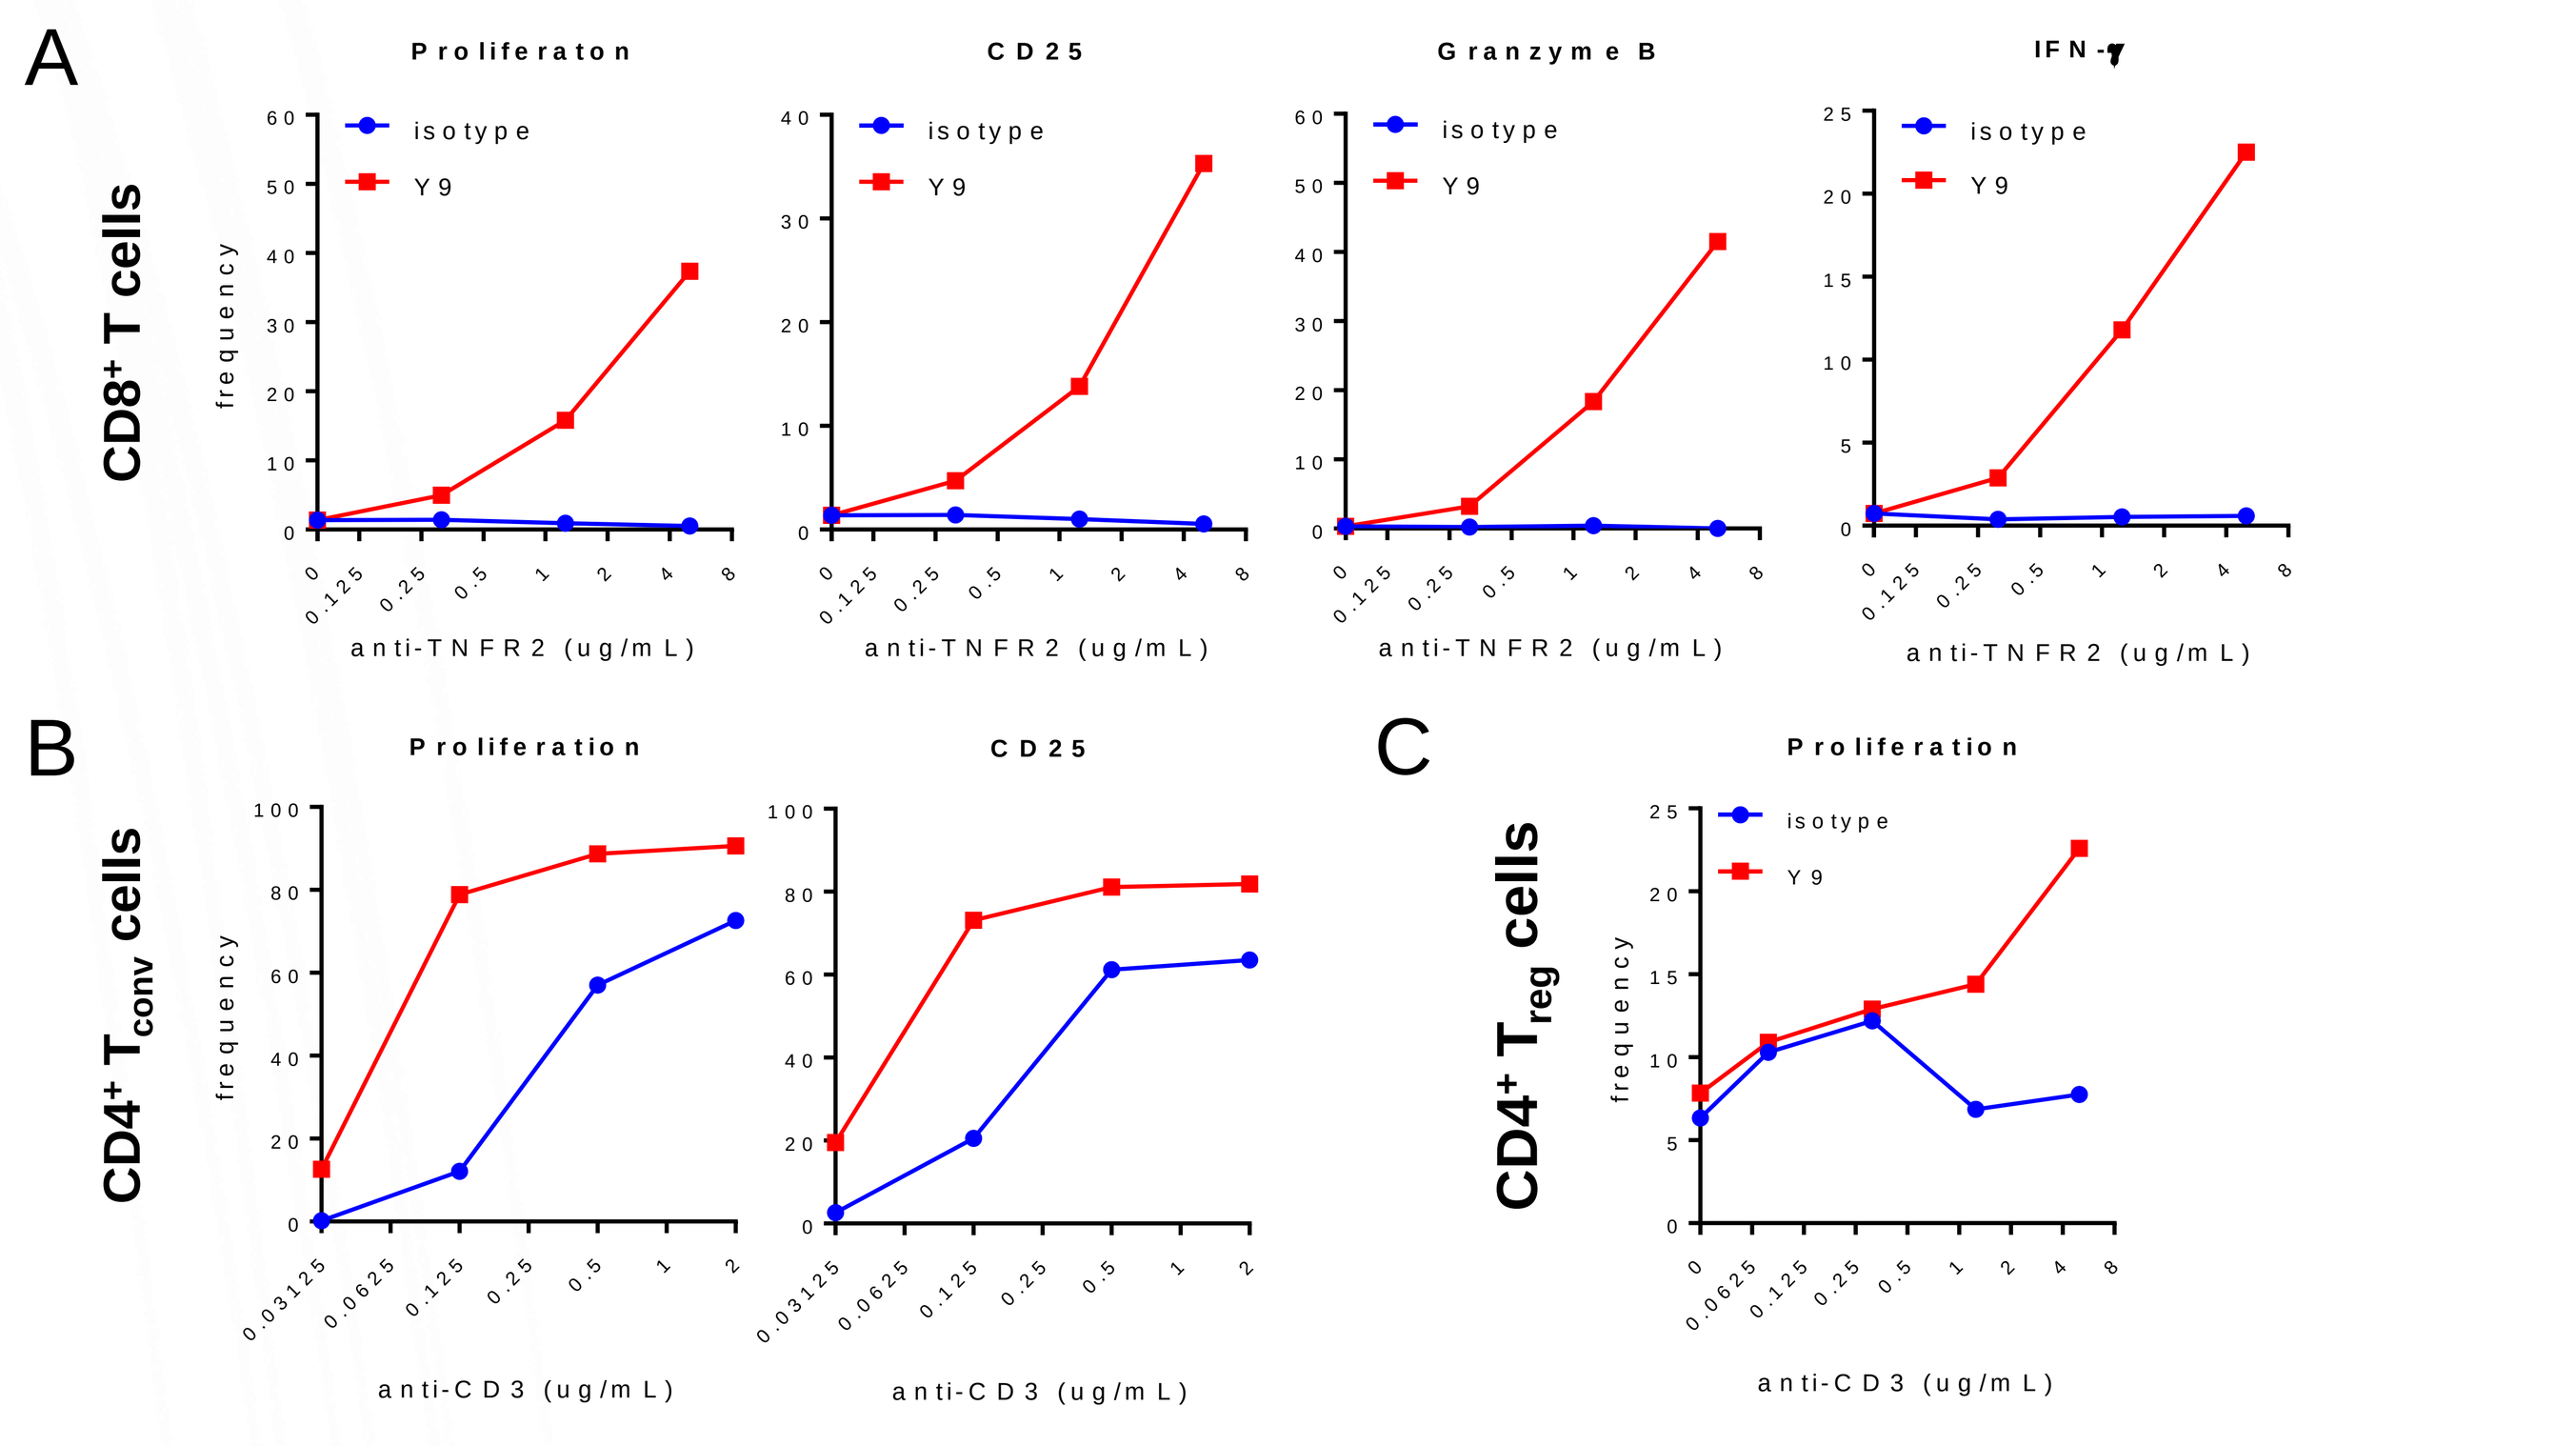

A
CD8+ T cells
Proliferaton
CD25
Granzyme B
IFN-γ
isotype
Y9
frequency
anti-TNFR2 (ug/mL)
B
CD4+ Tconv cells
Proliferation
CD25
anti-CD3 (ug/mL)
C
CD4+ Treg cells
Proliferation
anti-CD3 (ug/mL)

Total CD8$^+$ T cells **(A)** or CD25^{neg} T_{conv} **(B)** and CD25$^+$ T_{reg} **(C)** CD4$^+$ T cells were purified from naïve BALB/c mice by negative selection. Cells were labelled with CellTrace Violet to monitor cell division and stimulated *in vitro* for 72 hrs. Cytokine production by CD8$^+$ T cells was assessed by adding brefeldin A to the culture for the final 4 hrs. **A)** CD8$^+$ T cells were stimulated with 0.2 μg/mL plate bound αCD3 + 1 μg/mL soluble αCD28 + various concentrations of plate-bound Y9. **(B and C)** CD4$^+$ T cells were stimulated with 5 μg/mL plate bound Y9 and various concentrations of plate bound αCD3 and 1 μg/mL soluble αCD28.

Anti-TNFR2 and anti-PD-1 antibody combination leads to superior survival in syngeneic mouse tumor models

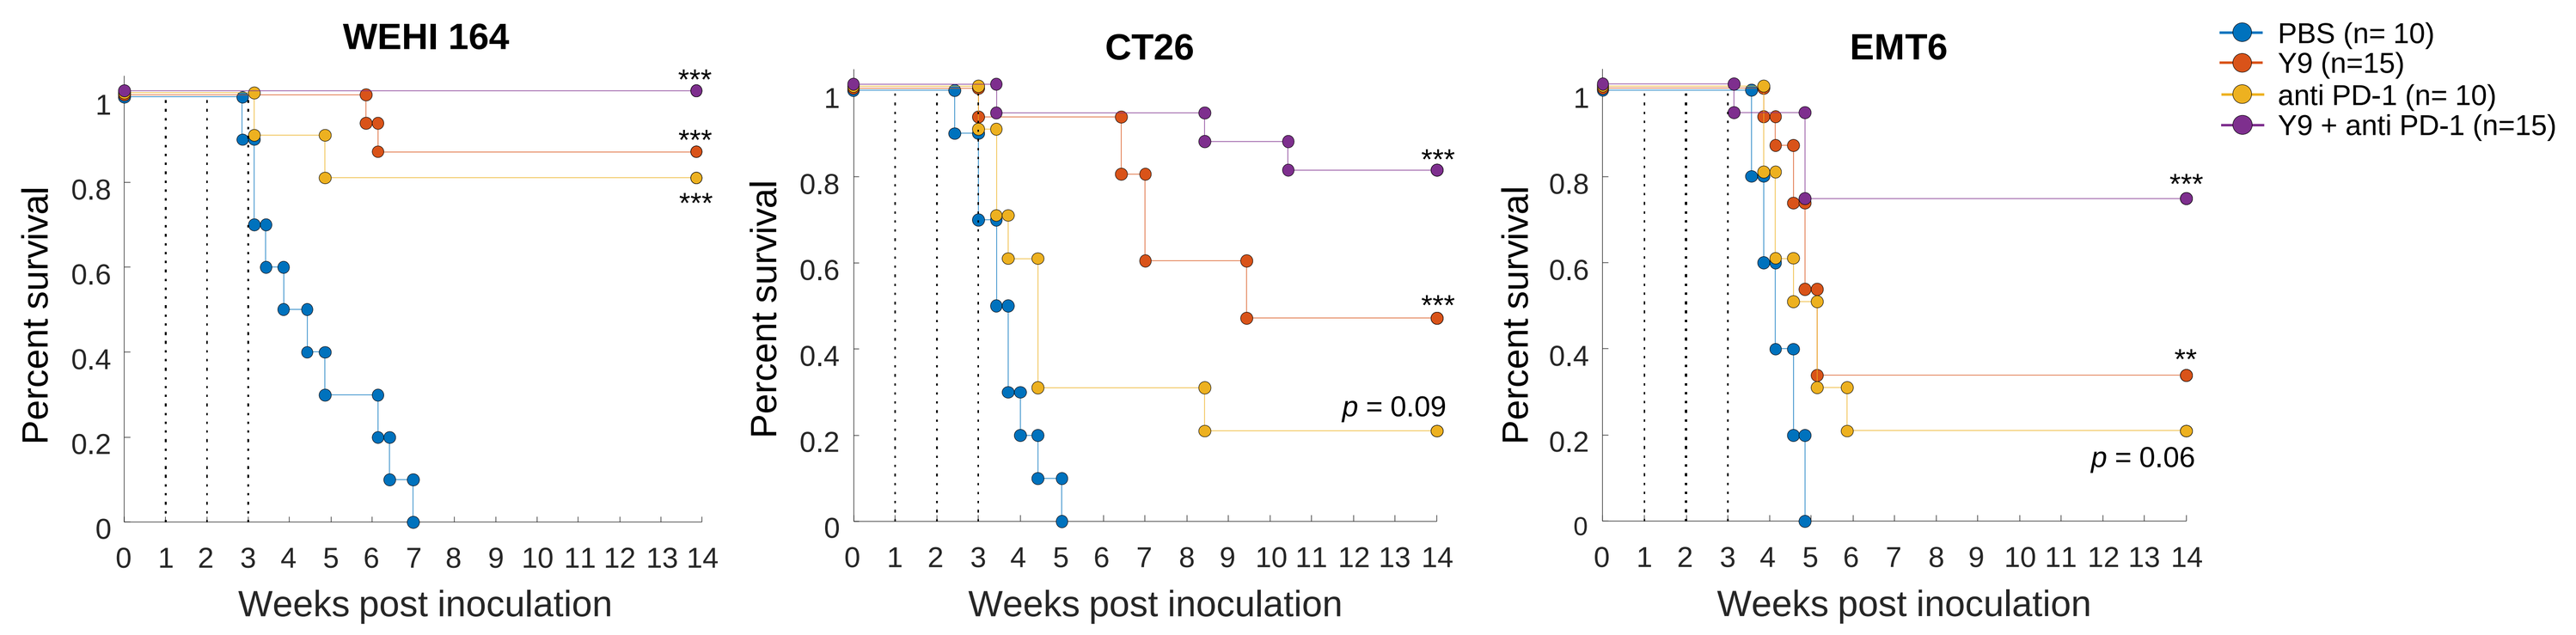

WEHI 164
CT26
EMT6
Percent survival
Weeks post inoculation
p = 0.09
p = 0.06
PBS (n= 10)
Y9 (n=15)
anti PD-1 (n= 10)
Y9 + anti PD-1 (n=15)

Additional responder models with CR
- MC38 (C57BL/6)
- MBT-2 (C3H)
- Sa1/N (A/J)

Responder model w/o CR
- A20 (BALB/c)

Non-responder models
- 4T1 (BALB/c)
- B16F10 (C57BL/6)
- LLC1 (BALB/c)

Survival curves for treatment with Y9 alone and in combination with anti-PD-1 in multiple BALB/c syngeneic s.c. tumor models. Mice with established tumors (75-100 mm^3) were given 3 x 300 μg doses for all mAbs (vertical dashed lines). The αPD-1 clone was J43 with a mouse IgG2a backbone. Endpoint defined as tumor volume of >2000 mm^3. Similar synergy was observed for Y9 + anti-PD-L1 combination therapy. CR = complete response where tumors have regressed below 60 mm^3. The statistical significance was calculated relative to PBS control.

Previously treated mice show immune memory to tumor re-challenge

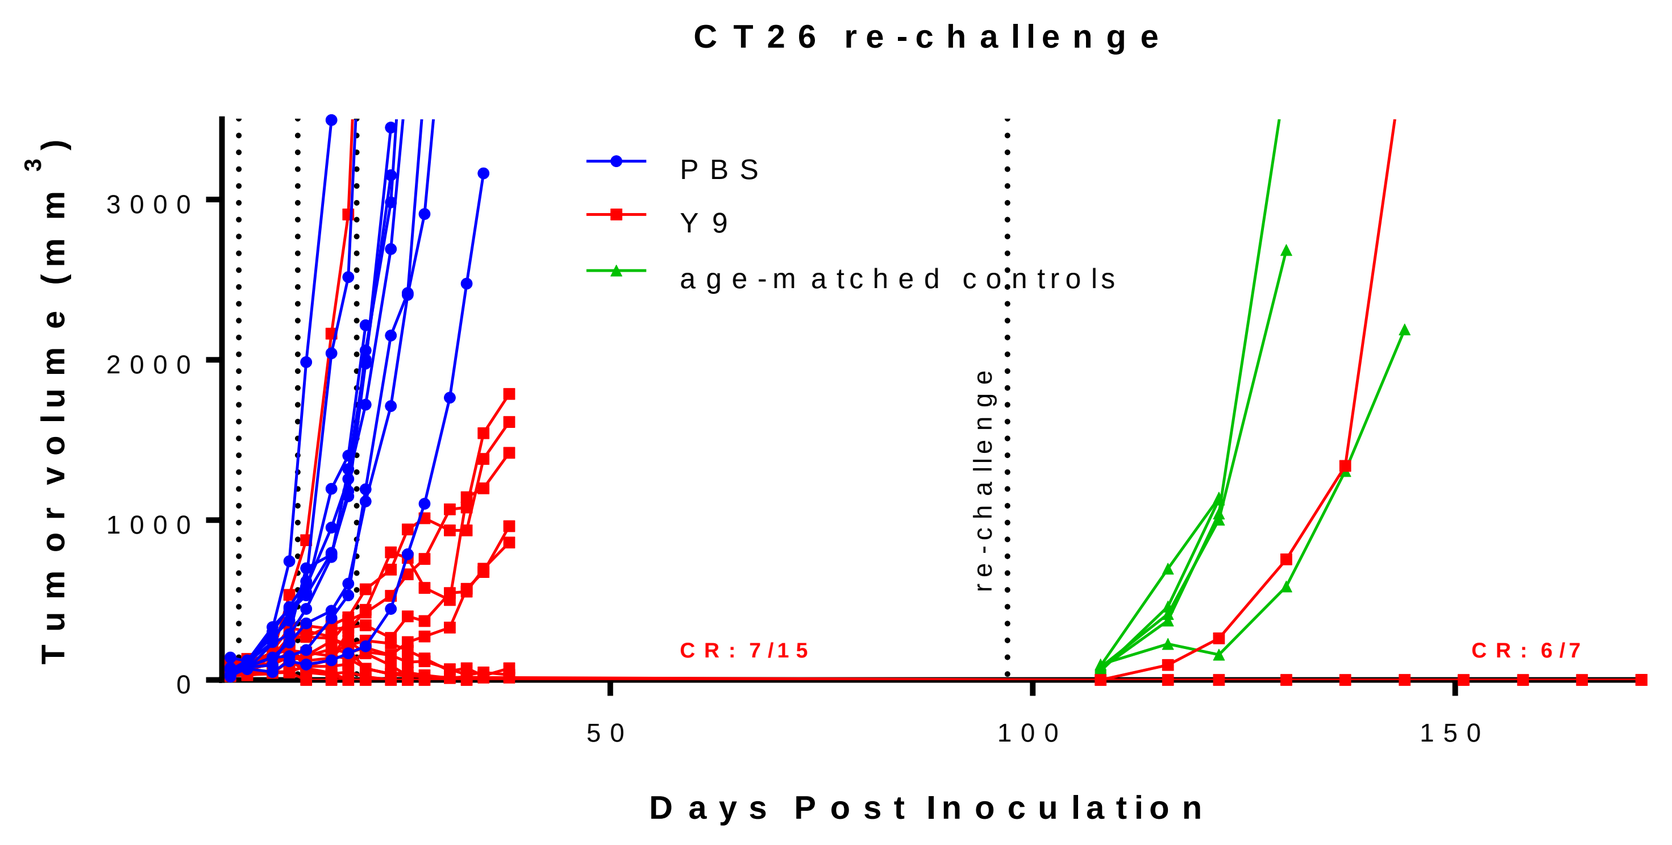

CT26 re-challenge
PBS
Y9
age-matched controls
re-challenge
Tumor volume (mm3)
CR: 7/15
CR: 6/7
Days Post Inoculation

BALB/c mice with established CT26 tumors (75-100 mm^3) were given 3 x 300 μg doses of Y9 (vertical dashed lines) or PBS. Surviving mice were re-challenged on the opposite flank with CT26 on day 97. CR = complete response where tumors have regressed below 60 mm^3. Similar results were observed for WEHI 164 and EMT6.

Anti-TNFR2 antibody activity requires Fcɣ receptor binding

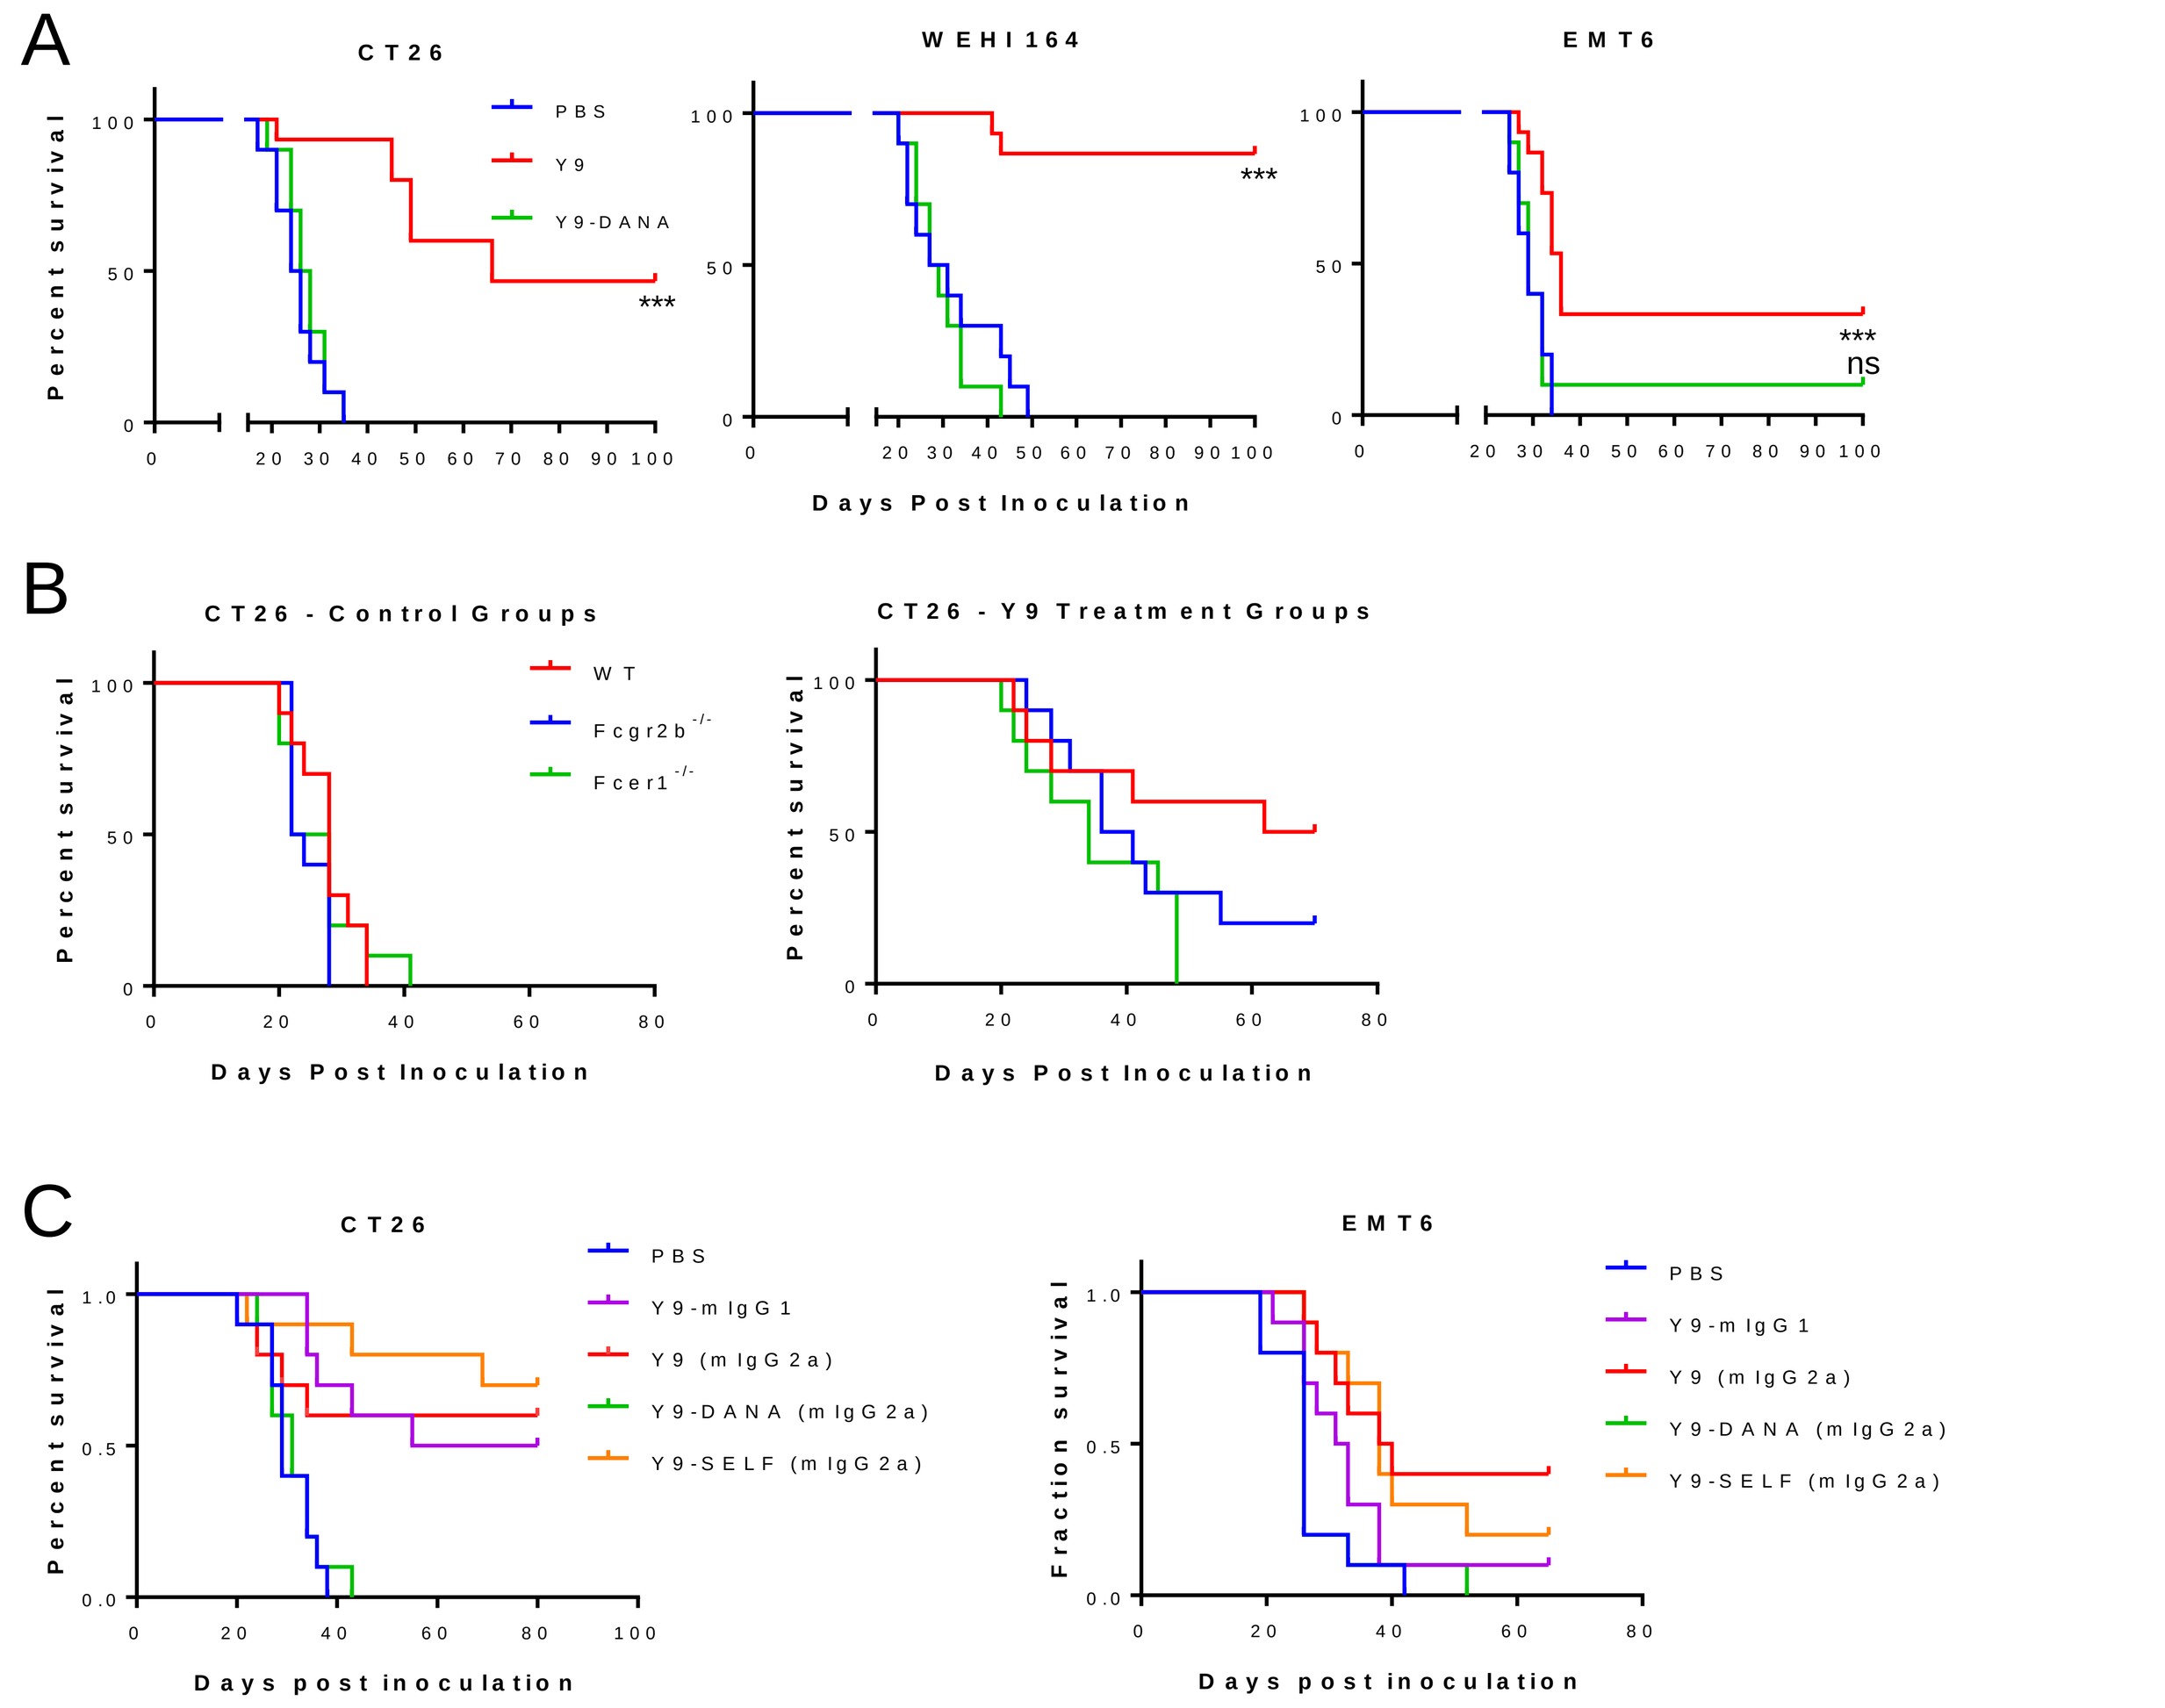

A
CT26
WEHI164
EMT6
PBS
Y9
Y9-DANA
Percent survival
Days Post Inoculation
ns
B
CT26 - Control Groups
CT26 - Y9 Treatment Groups
WT
Fcgr2b-/-
Fcer1-/-
C
CT26
EMT6
PBS
Y9-mIgG1
Y9 (mIgG2a)
Y9-DANA (mIgG2a)
Y9-SELF (mIgG2a)
Fraction survival
Days post inoculation

BALB/c mice with established s.c. tumors (75-100 mm^3) were given 3 x 300 μg doses for all mAbs and tumor volume was tracked. **(A)** Comparison of Y9 WT (mIgG2a) and Y9 Fc-mutant (D265A/N297A) that lacks binding to FcγR. **(B)** Wild-type or mice lacking the inhibitory FcγRIIb (*Fcgr2b*$^{-/-}$) or activating FcγR (*Fcer1*$^{-/-}$) were treated with control PBS or Y9. Similar outcomes were observed in the EMT6 tumor model. **(C)** BALB/c mice with either CT26 or EMT6 tumors were treated with control PBS, Y9 (mouse IgG2a), Y9 switched to a mouse IgG1 isotype, Y9-DANA, or mouse IgG2a Y9-SELF (S267E + L328F mutations) with increased affinity for mouse FcγRIIb.

CD8$^+$ T cells and NK cells are important for anti-TNFR2 antibody activity

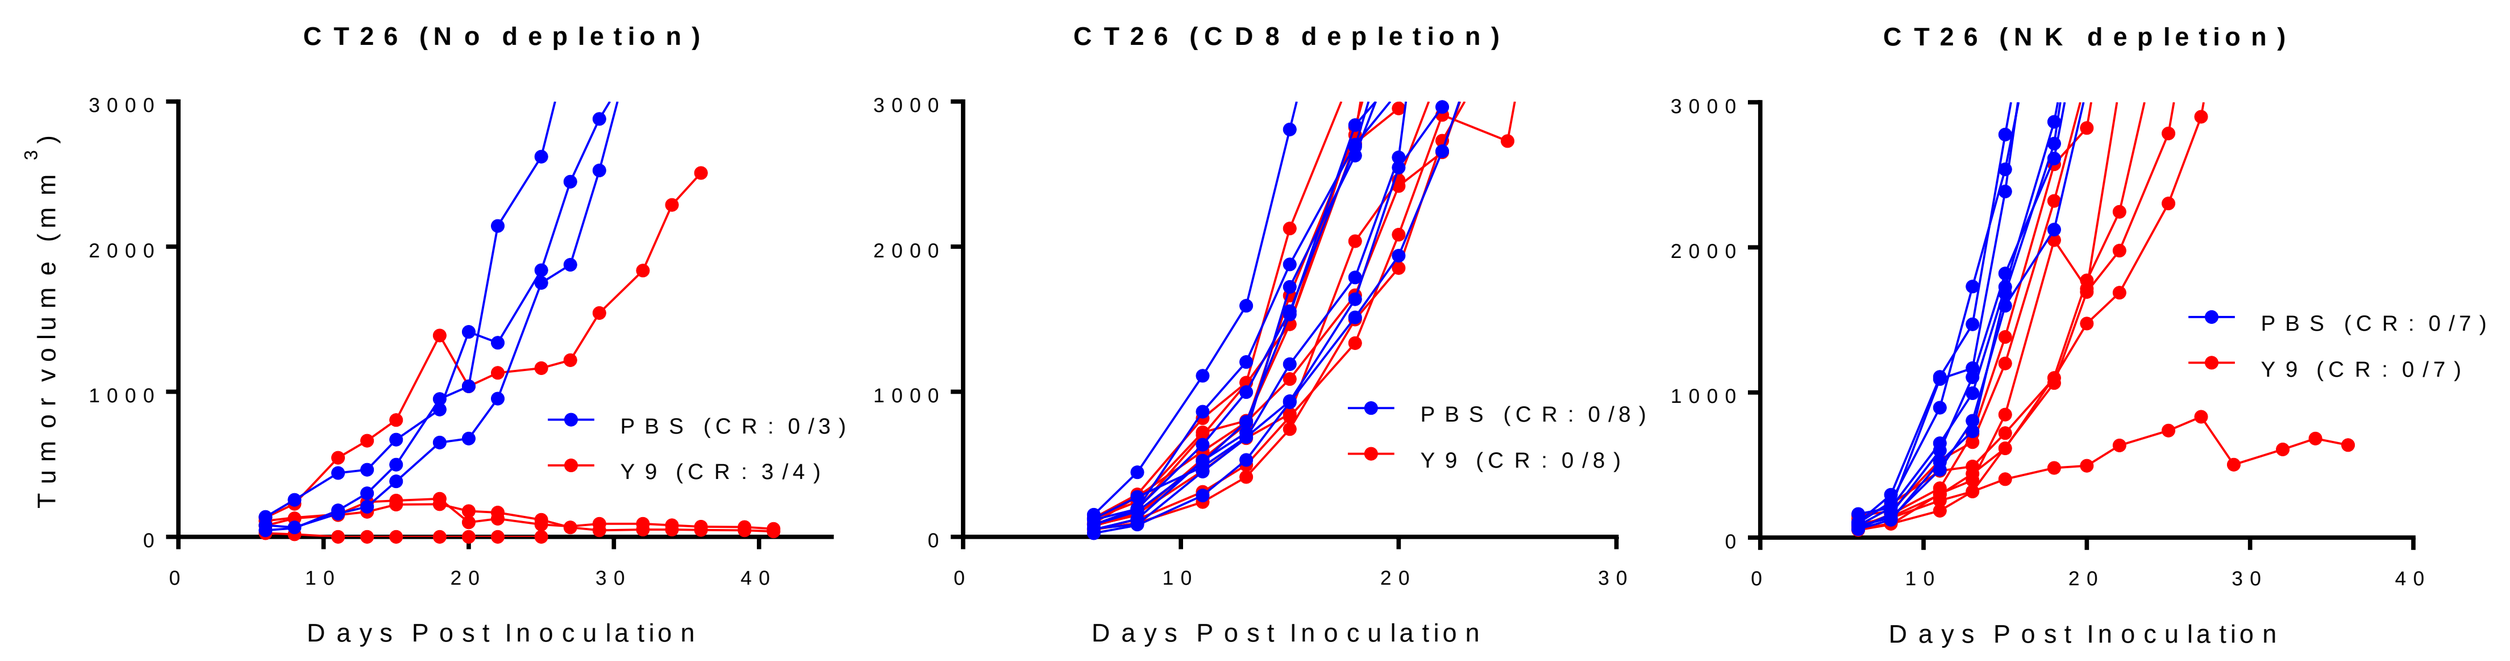

CT26 (No depletion)
CT26 (CD8 depletion)
CT26 (NK depletion)
Tumor volume (mm3)
Days Post Inoculation
PBS (CR: 0/3)
Y9 (CR: 3/4)
PBS (CR: 0/8)
Y9 (CR: 0/8)
PBS (CR: 0/7)
Y9 (CR: 0/7)

BALB/c mice were given an injection of depleting antibody on day -3 and day 4 relative to CT26 tumor cell inoculation: 300 μg αCD4 (clone GK1.5), 300 μg αCD8 (clone 53-6.7), or 50 μL polyclonal αAsialo GM-1 to NK cells. When CT26 tumors were established (~85 mm^3) mice were given a single injection of control PBS or Y9. CR = complete response where tumors have regressed below 60 mm^3.

T_{reg} depletion is not consistently observed across responder models

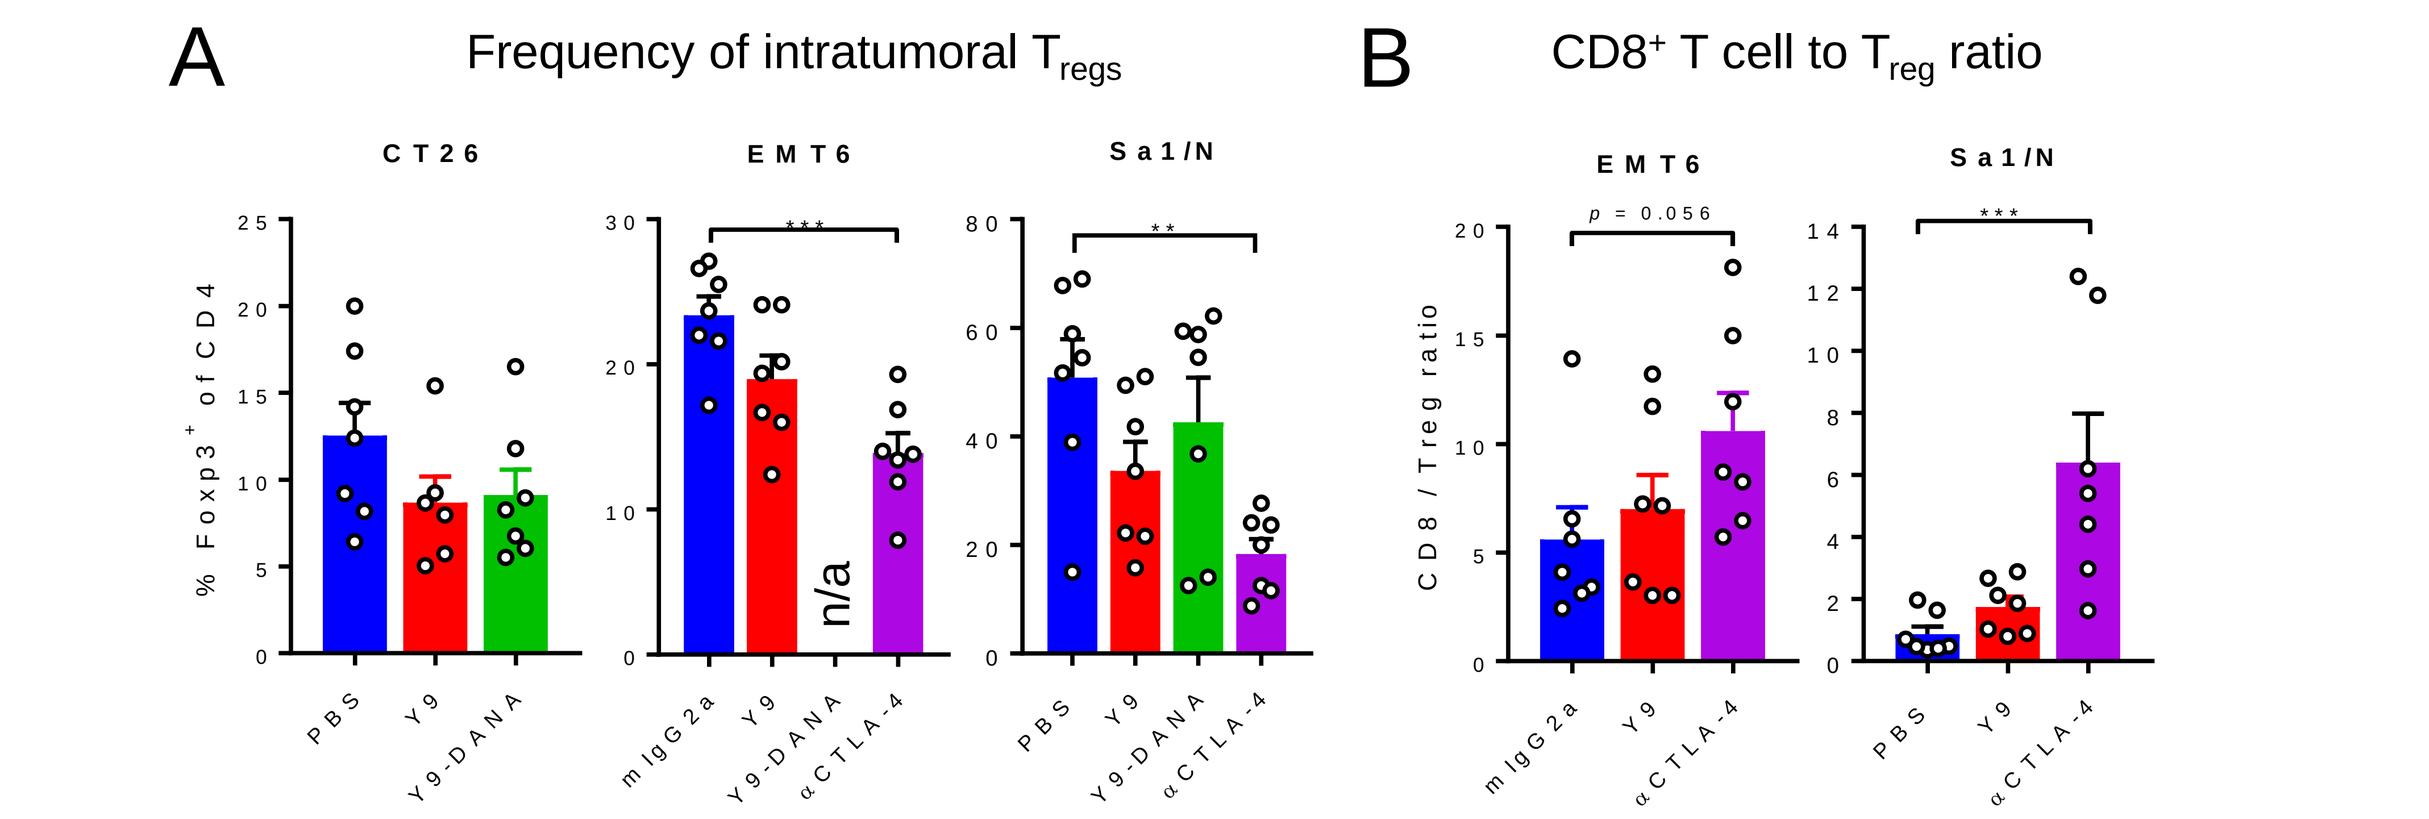

A
Frequency of intratumoral Tregs
CT26
EMT6
Sa1/N
% Foxp3+ of CD4
n/a
PBS
Y9
Y9-DANA
mIgG2a
αCTLA-4
B
CD8+ T cell to Treg ratio
EMT6
Sa1/N
p = 0.056
CD8 / Treg ratio

BALB/c mice with established s.c. tumors were treated with control PBS or 300 μg of Y9, Y9-DANA, or αCTLA-4 (clone 9D9 with mouse IgG2a isotype). 24-36 hrs later, tumors were harvested, dissociated, and T cell subsets were analyzed by flow cytometry. **(A)** Frequency of T_{regs} within the CD4$^+$ T cell pool. **(B)** CD8/T_{reg} ratio. Anti-CTLA-4 mAb was used as a positive control for T_{reg} depletion.

Anti-TNFR2 co-stimulation enhances the magnitude and effector function of CD8$^+$ T cells

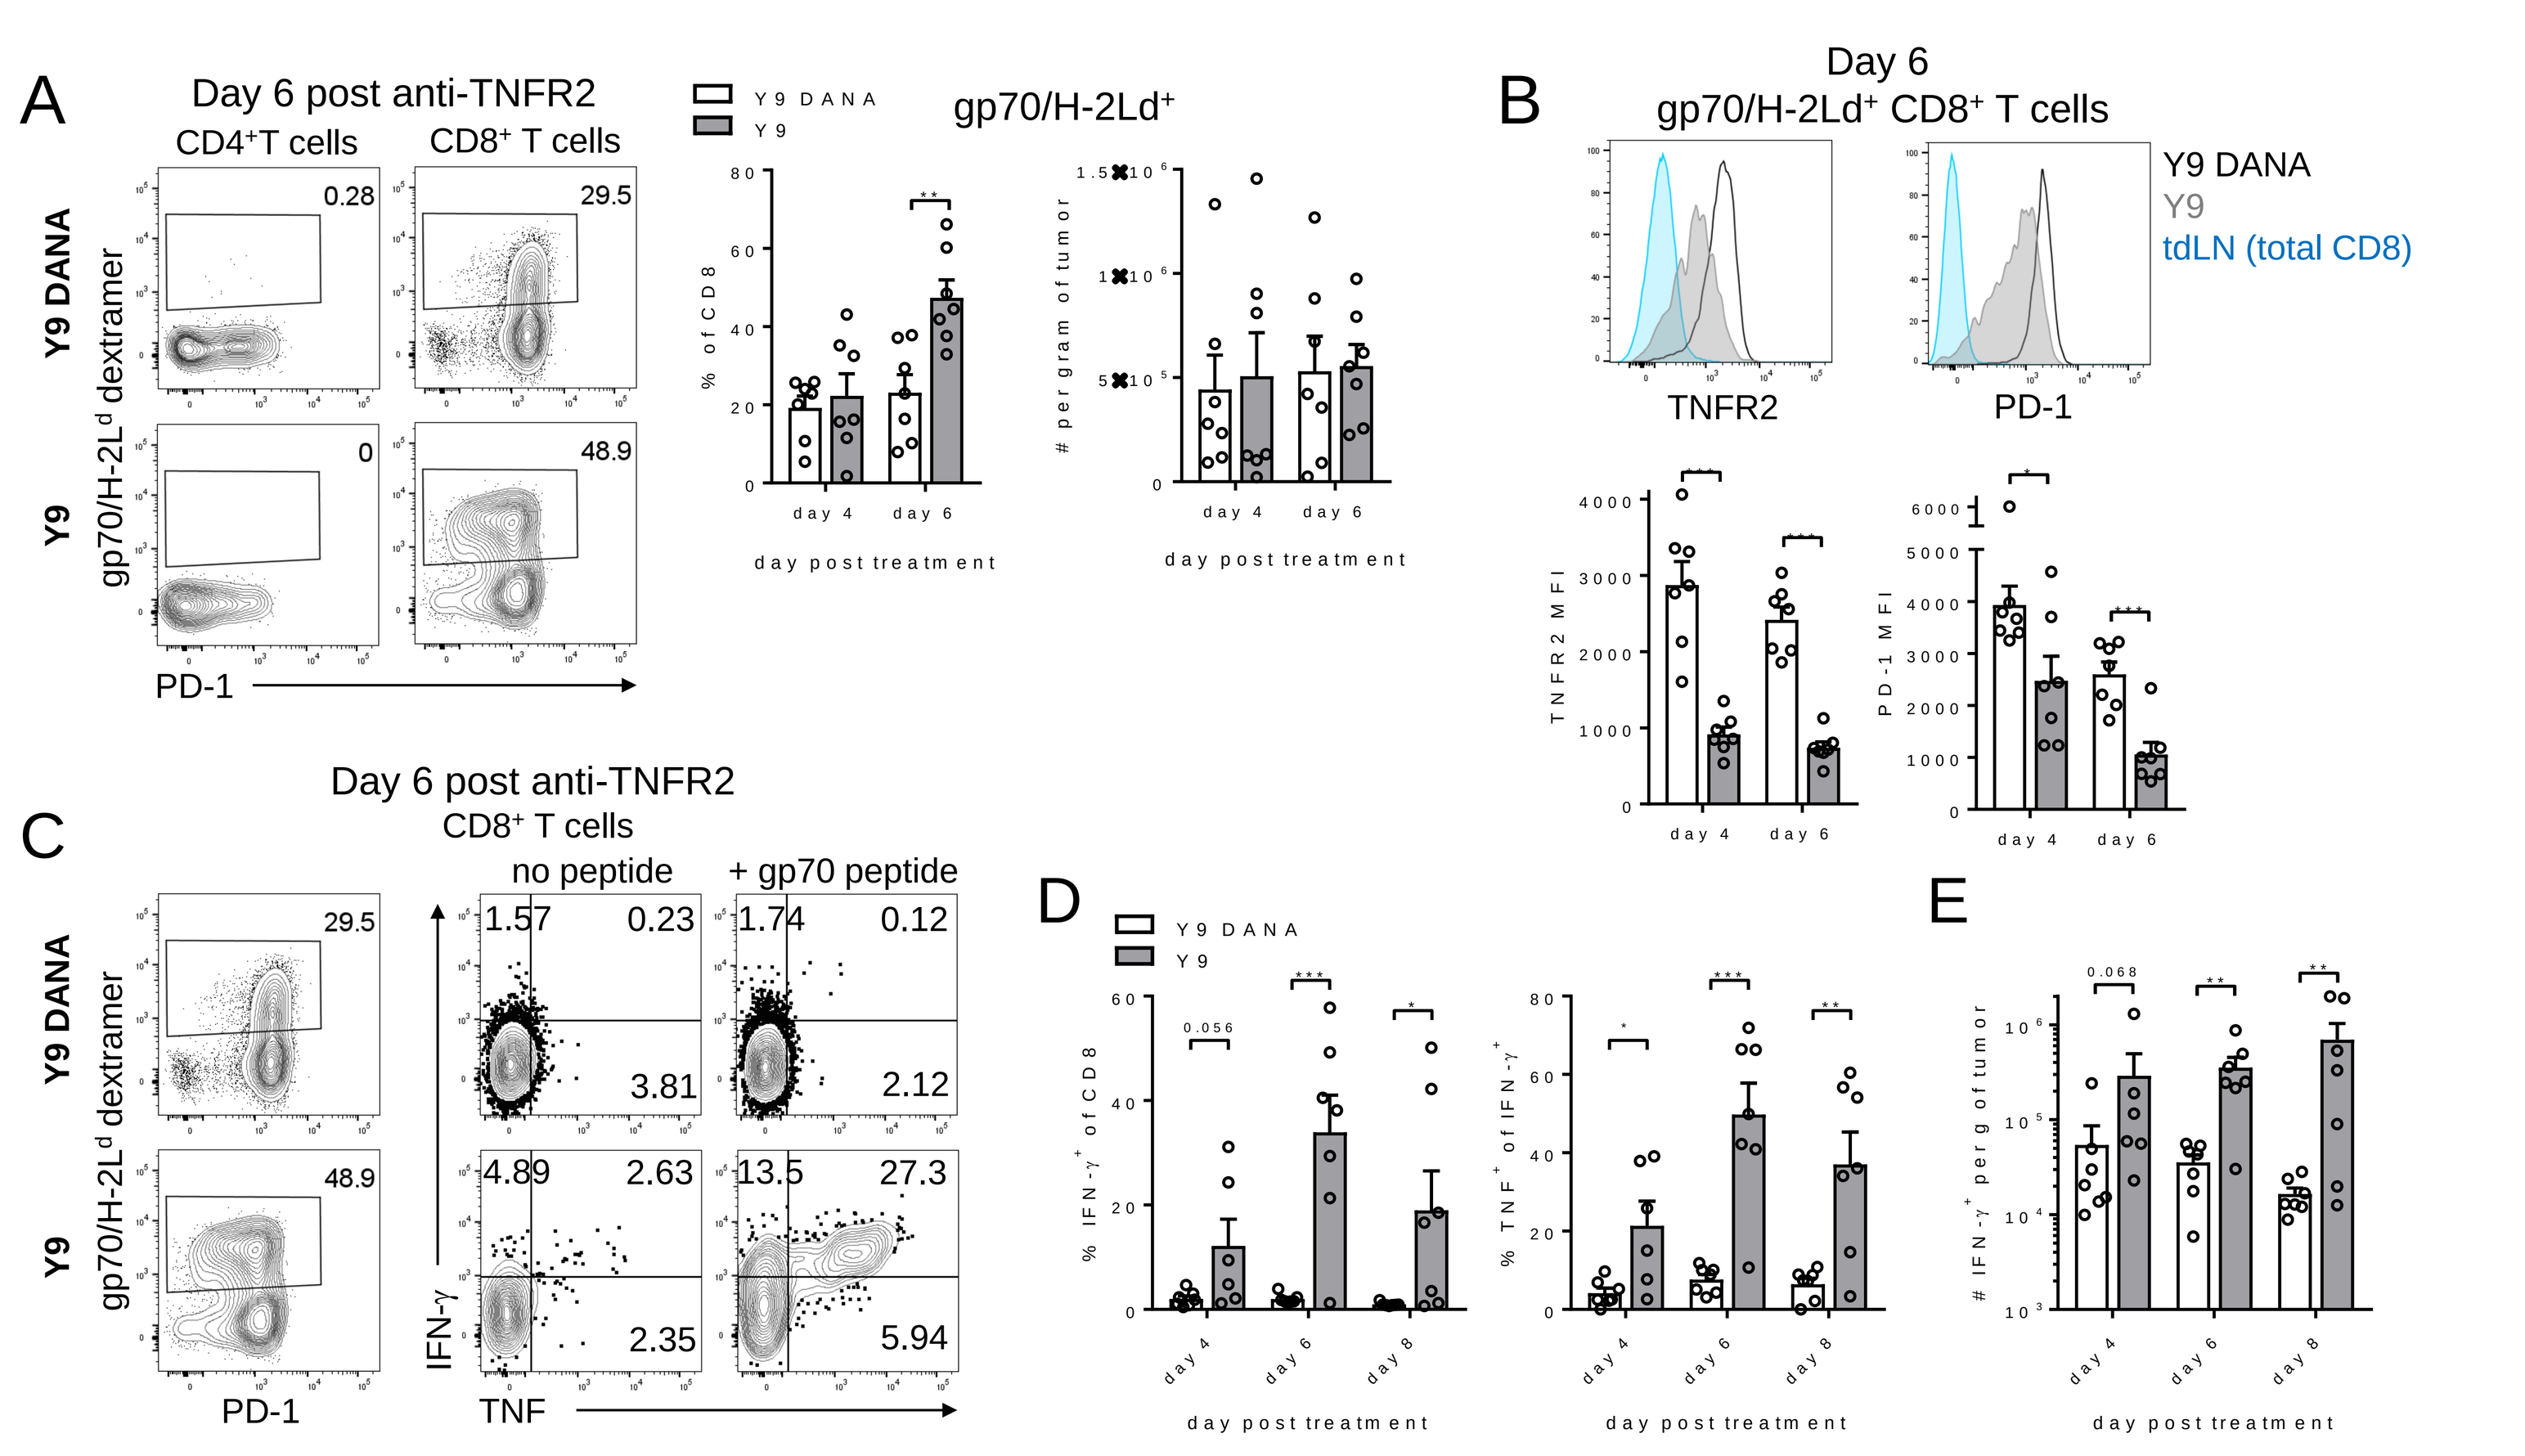

A
Day 6 post anti-TNFR2
CD4+T cells
CD8+ T cells
Y9 DANA
Y9
gp70/H-2Ld dextramer
PD-1
0.28
29.5
0
48.9
gp70/H-2Ld+
% of CD8
per gram of tumor
day 4
day 6
day post treatment
B
Day 6
gp70/H-2Ld+ CD8+ T cells
Y9 DANA
Y9
tdLN (total CD8)
TNFR2
PD-1
TNFR2 MFI
PD-1 MFI
C
Day 6 post anti-TNFR2
CD8+ T cells
no peptide
+ gp70 peptide
IFN-γ
TNF
D
E
% IFN-γ+ of CD8
% TNF+ of IFN-γ+
IFN-γ+ per g of tumor
day 8

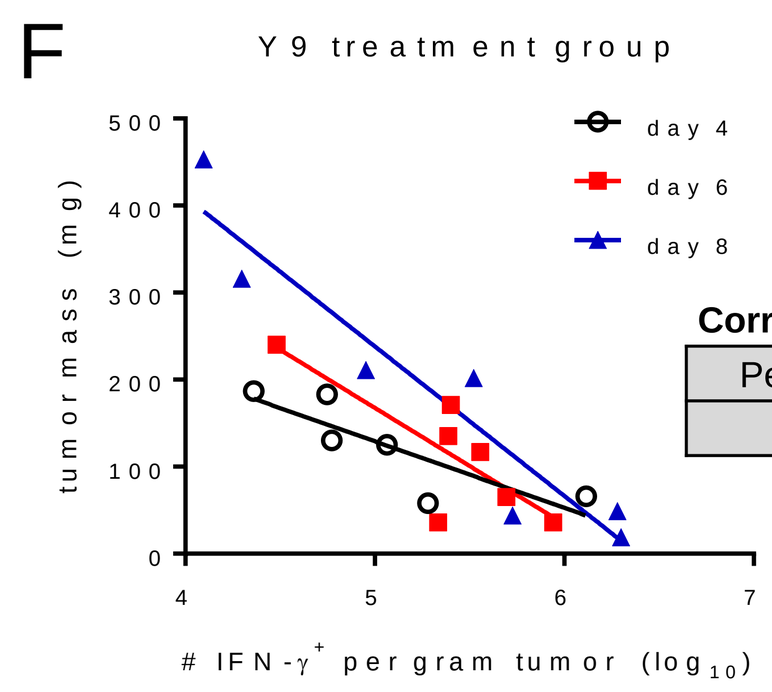

F
Y9 treatment group
day 4
day 6
day 8
tumor mass (mg)
IFN-γ+ per gram tumor (log10)

Correlation	day 4	day 6	day 8
Pearson r	-0.8387	-0.8079	-0.9496
P value	0.0369	0.0279	0.0011

BALB/c mice bearing established CT26 s.c. tumors (~150 mm^3) were equally distributed into treatment groups and treated 1 x 300 μg Y9 or Y9 Fc-mutant (Y9 DANA). At various times, tumors were harvested, dissociated, and the AH1 gp70-specific CD8$^+$ T cell response was characterized using gp70/H-2L^d dextramer **(A and B)** or cells were stimulated *ex vivo* with gp70 peptide + Golgi inhibitor for 5 h **(C-E)**. Following stimulation, cells were stained for intracellular IFN-γ and TNF. **(F)** Pearson correlation between tumor mass and the # of IFN-γ^+ gp70-specific CD8$^+$ T cells per gram of tumor.

Mechanism of action summary

Dominant Mechanism:

- **Co-stimulatory activity on T cells**
 - ✓ Potent in vitro stimulation of CD8$^+$ and CD4$^+$ T cells
 - ✓ Increases magnitude and effector function of tumor infiltrating CD8$^+$ T cells
 - ✓ TNFR2 receptor downregulation
 - ✓ Dependency on inhibitory and activating Fcɣ receptors
 - ✓ Comparable activity of mIgG2a, mIgG1, and variants with enhanced binding to inhibitory Fcɣ receptors
 - ✓ Fast downregulation of immunosuppressive markers on T cells
- **Direct depletion of immune cell subsets (including potentially Tregs) via ADCC, etc.**
 - ✗ No consistent reduction in the frequency of T cell subsets in the tumor and periphery
 - ✗ No loss of efficacy after CD4 T cell depletion (including Tregs) (data not shown)

Other Mechanisms Under Investigation:

- **Direct phenotypic effect on Tregs**

 In Progress: Determine importance of TNFR2 on Tregs in vitro

 In Progress: Studies in Treg-specific TNFR2 knockout mice (using DEREG mice)

Conclusions and Cross-references

Agonistic anti-TNFR2 antibodies show broad anti-tumor activity in syngeneic mouse models and a favorable toxicity profile compared to anti-CTLA-4 in a long-term exposure study in mice (see Richards *et al*. below). A human anti-TNFR2 antibody (MM-401) with low nanomolar affinity and binding to the same epitope as the murine surrogate antibody (Y9) has been developed. MM-401 is being developed as a potential novel treatment option for cancer patients. See Sampson *et al*. below for characterization of MM-401.

Richards *et al.* MM-401, a novel anti-TNFR2 antibody that induces T cell co-stimulation, robust anti-tumor activity and immune memory. AACR 2019, Abstract #4846.

Sampson *et al.* A novel human TNFR2-antibody (MM-401) modulates T cell responses in anti-cancer immunity. AACR 2019, Abstract #555.

All data unpublished and on file at Merrimack Pharmaceuticals, Inc.

A PASSION FOR OUTTHINKING CANCER

© 2019 Merrimack Pharmaceuticals, Inc. All rights reserved.